Targeting Notch/Delta-like ligand 4 (DLL4) and vascular endothelial growth factor (VEGF) pathways by an anti-DLL4/Anti-VEGF bi-specific monoclonal antibody inhibits tumor growth and reduces cancer stem cell frequency in solid tumors

Wan-Ching Yen, Aaron Sato, Chris Bond, Jennifer Cain, Janak Raval, Fumiko Axelrod, Cecile Chartier, Shirley Ma, Marcus Fischer, Rene Meisner, Jalpa Shah, Austin Gurney, John Lewicki, and Timothy Hoey. OncoMed Pharmaceuticals, Inc. Redwood City, CA

ABSTRACT

Both Notch/Delta-like ligand 4 (DLL4) and vascular endothelial growth factor (VEGF) pathways play a critical role in angiogenesis and tumor growth. Due to differential regulatory effects of VEGF and DLL4 on the vasculature, blockage of DLL4 or VEGF inhibits tumor growth by distinct mechanisms: anti-DLL4 treatment induces an abnormal increase of poorly perfused blood vessels, which results in a nonproductive angiogenesis unable to support tumor growth, whereas the anti-VEGF therapy significantly decreases vasculature reducing the blood supply to tumors. We have recently developed a high binding affinity bi-specific monoclonal antibody that targets both human DLL4 and human VEGF. *In vitro*, this antibody bound with low nanomolar affinity to hVEGF and hDLL4, and reduced HUVEC proliferation induced by VEGF. The bi-specific antibody demonstrated significant *in vivo* anti-tumor efficacy in various solid tumors, delayed tumor recurrence following termination of chemotherapy, and decreased the frequency of tumor initiating cells. Analysis of tumor vasculature after treatment with anti-DLL4/VEGF revealed inhibition of vascular gene expression and endothelial cell proliferation, indicating that the anti-VEGF effect on the vasculature is dominant over the anti-DLL4 effect. Notably, at doses where both anti-DLL4 and anti-VEGF alone produces suboptimal anti-tumor effect, dual targeting resulted in an enhanced anti-tumor growth inhibition. These results indicate that our bi-specific anti-DLL4/VEGF is active in colon, ovarian and gastric xenograft models and may be useful for treatment of a variety of solid tumors.

BACKGROUND

- Both DLL4 and VEGF play important roles in tumor angiogenesis
- Blockade of DLL4 or VEGF inhibits angiogenesis through distinct mechanisms
 - –DLL4 blockade inhibits functional lumen development
 - –VEGF blockade inhibits endothelial proliferation
- DLL4-Notch signaling is part of a negative feedback loop in the angiogenic process
- We previously demonstrated that targeting DLL4/Notch signaling in the tumors reduces tumorigenic potential of cancer stem cells in patient-derived xenograft tumor models (Hoey et al. Cell Stem Cell 5:168-177, 2009)
 - – DLL4 blockade leads to upregulation of VEGF, and unregulated endothelial hyperproliferation
- Anti-DLL4/VEGF retains the anti-cancer stem cells impact of anti-DLL4 and has potentially increased anti-angiogenic activity relative to current anti-VEGF therapeutics
- OncoMed's anti-DLL4/anti-VEGF bi-specific design:

Bi-specific
anti-DLL4 + anti-VEGF

- **Dual inhibitor of both DLL4 and VEGF**
- **Proprietary bi-specific antibody technology**
 - **Two distinct heavy chains recognizing DLL4 and VEGF respectively**
 - **Mutations at CH3 domain of the heavy chains that drives heterodimer formation**
 - **Common light chain**
 - **Humanized IgG2**

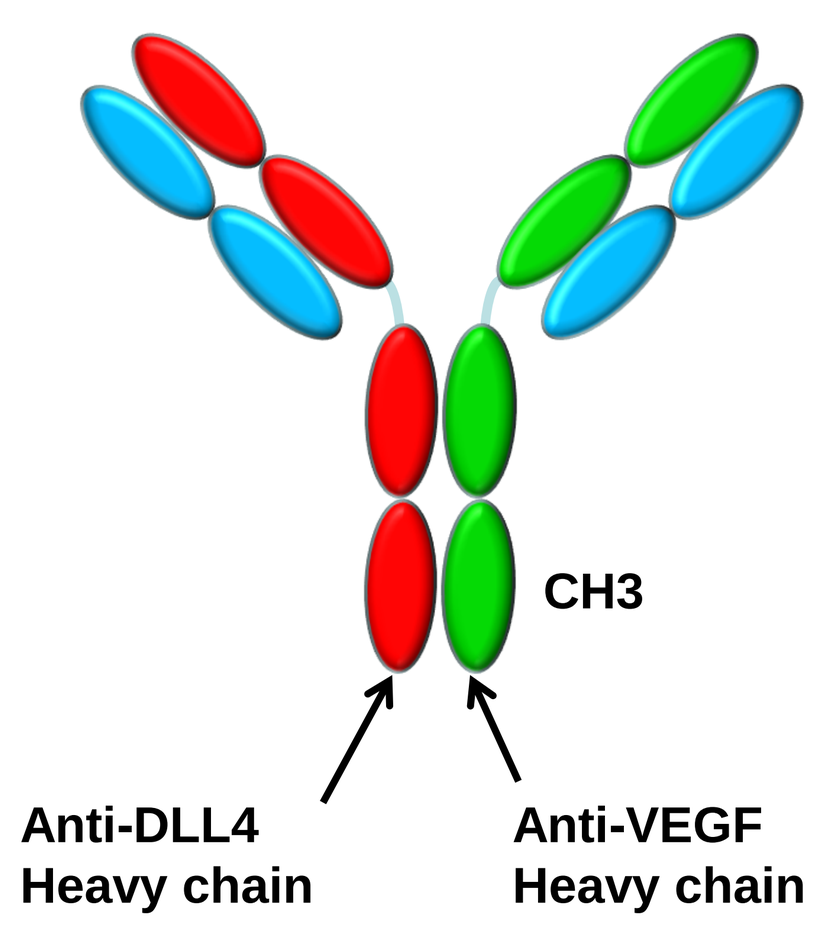
CH3
Anti-DLL4
Heavy chain
Anti-VEGF
Heavy chain

Figure 1 Schematic representation of bispecific anti-VEGF/DLL4 molecule. The heterodimeric heavy chain is comprised of a human IgG2 with point mutations in the CH3 domain that drive heterodimer formation. Each heavy chain is paired with a common light chain that supports binding of both the DLL4 and VEGF antigens.

MATERIALS and METHODS

***In vitro* binding assay**: Binding affinities for the anti-VEGF and anti-DLL4 arms of the bispecific molecule were determined by surface Plasmon resonance using a Biacore 2000. Each antigen was coupled to the SPR chip by either streptavidin-biotin (VEGF) or direct coupling to a CM5 chip with NHS chemistry. Association and dissociation curves were determined for each antigen at multiple concentrations and the affinity was measured by globally fitting the data using a 1-1 binding model.
***In vitro* HUVEC proliferation assay**: Human endothelial cells were harvested from *in vitro* cultures and distributed in the number of 5,000 into a 96-well plate, then placed into starvation [low serum] medium overnight before being incubated with the mixture of rhVEGF (10 ng/ul) and multiple concentrations of test antibodies from the highest 20 ug/ml to the lowest 0.0512 ng/ml. After seven days, the cells were incubated with Alamar Blue at 37°C before reading the plate with Molecular Device Micro-plate Reader SpectraMax M5.

***In vivo* mouse xenograft efficacy studies**: NOD/SCID mice were implanted subcutaneously with patient-derived tumor xenografts of human ovarian tumor OMP-OV40, colon tumor OMP-C8 or gastric tumors STM1. Tumors were treated with control mAb, anti-DLL4, anti-VEGF or the bi-specific antibody when the tumors reached the size of 150 mm^3.
Immunoflourescence analysis: Histologic analysis used frozen sections. Slides were imaged with an Olympus BX51 microscope using IPLabs v4.0.
RT-PCR analysis: For gene analysis, RNA was isolated from tumor tissues followed by c-DNA synthesis. The resulting c-DNA was analyzed by real-time PCR.
***In vivo* tumorigenicity studies:** NOD/SCID mice bearing subcutaneous OMP-C8 tumors were treated with anti-VEGF, anti-DLL4 or the bi-specific antibody in combination with irinotecan for 4 weeks. Following termination of chemotherapy, tumors were harvested from 4-5 mice of each treatment group and dissociated into single cells, which were depleted of mouse cells, and re-implanted in a new batch of mice; tumor incidence and size were recorded until 80 days after implantation of tumor cells. The remaining mice continued to be treated with antibodies single agents for additional several weeks.

RESULTS

***In Vitro* Binding**

IgG	hVEGF (nM)	mVEGF (nM)	hDLL4 (nM)	mDLL4 (nM)
Anti-VEGF/DLL4 Bi-specific Ab	0.36	25.5	1.3	NB

Impact on VEGF-Induced Human Endothelial Cell Proliferation

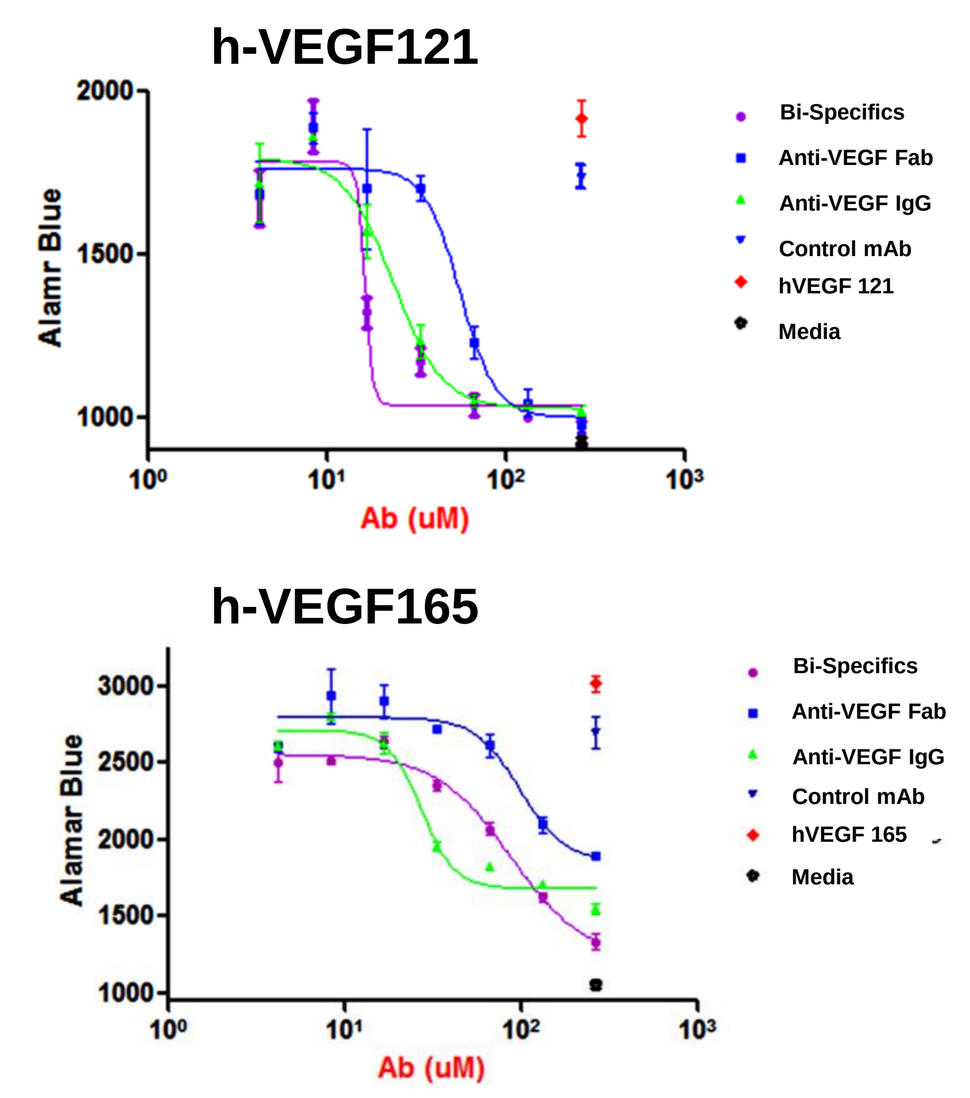
h-VEGF121
Alamr Blue
Ab (uM)
Bi-Specifics
Anti-VEGF Fab
Anti-VEGF IgG
Control mAb
hVEGF 121
Media
h-VEGF165
Alamar Blue
Ab (uM)
hVEGF 165

Table 1 (Left): The anti-hVEGF and anti-hDLL4 bispecific molecule demonstrated higher affinity to human VEGF compared to mouse VEGF and to human DLL4 while it did not bind to mouse DLL4. NB: No Binding. **Figure 2 (Right):** The bi-specific antibody reduced proliferation of human endothelial cells in the presence of hVEGF in a dose-dependent manner.

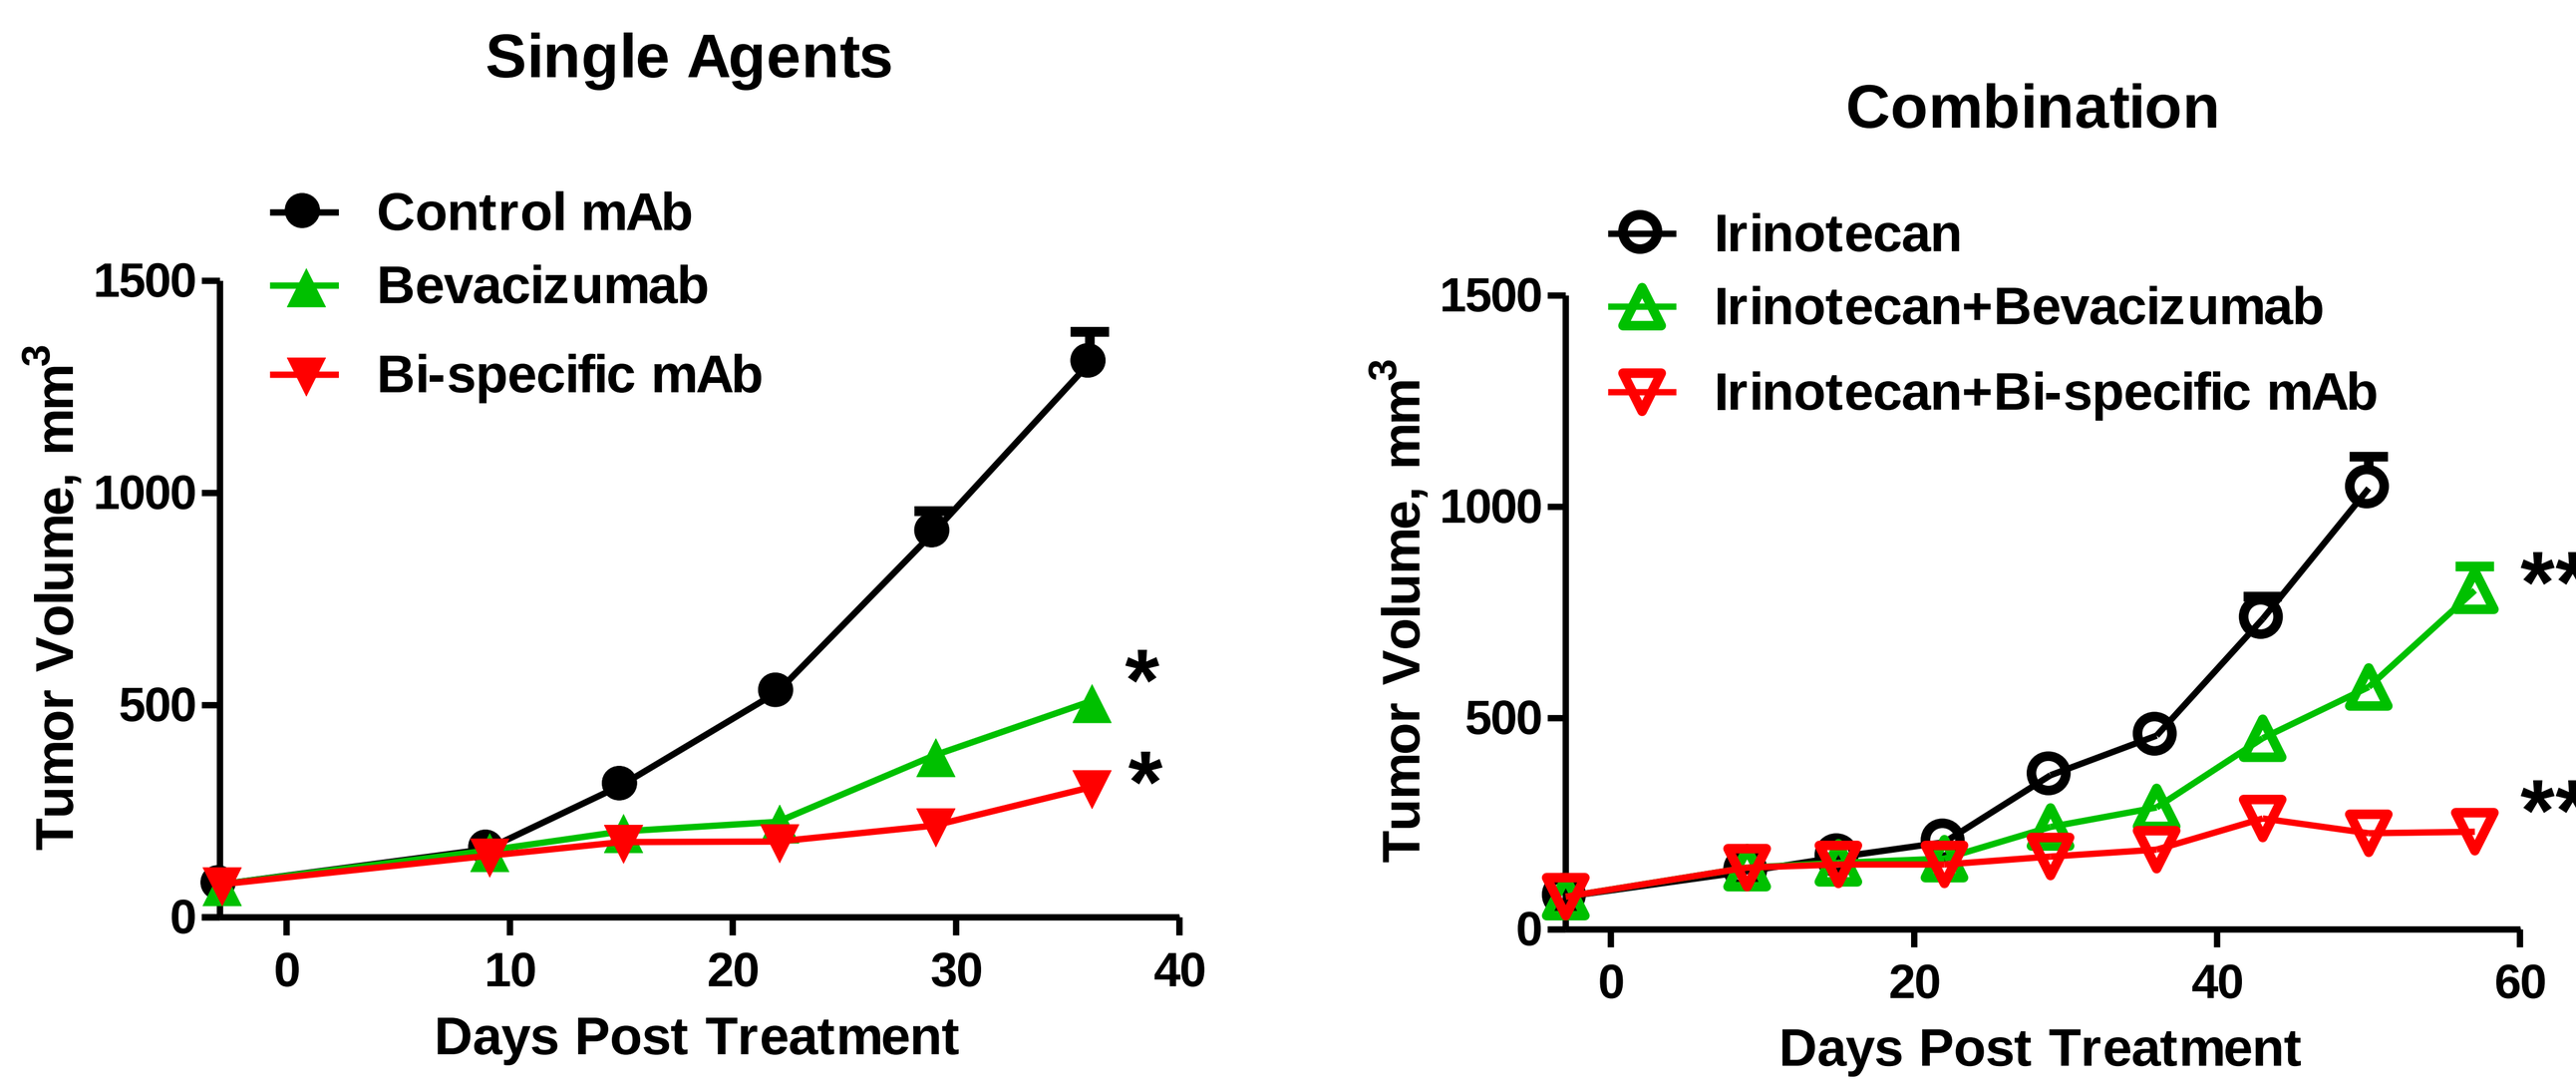
Single Agents
Control mAb
Bevacizumab
Bi-specific mAb
Tumor Volume, mm3
Days Post Treatment
Combination
Irinotecan
Irinotecan+Bevacizumab
Irinotecan+Bi-specific mAb
Tumor Volume, mm3
Days Post Treatment

Figure 3: Anti-DLL4/VEGF Bi-specific Antibody Inhibits Colon Xenograft Tumor Growth. Kras mutant colon xenograft tumors OMP-C8 were treated with either control mAb, 7.5 mg/kg irinotecan, 7.5 mg/kg bevacizumab or 15 mg/kg bi-specific mAb once a week for 4 weeks. *:p<0.05 vs. control mAb, **: p<0.05 vs. irinotecan by two-way ANOVA.

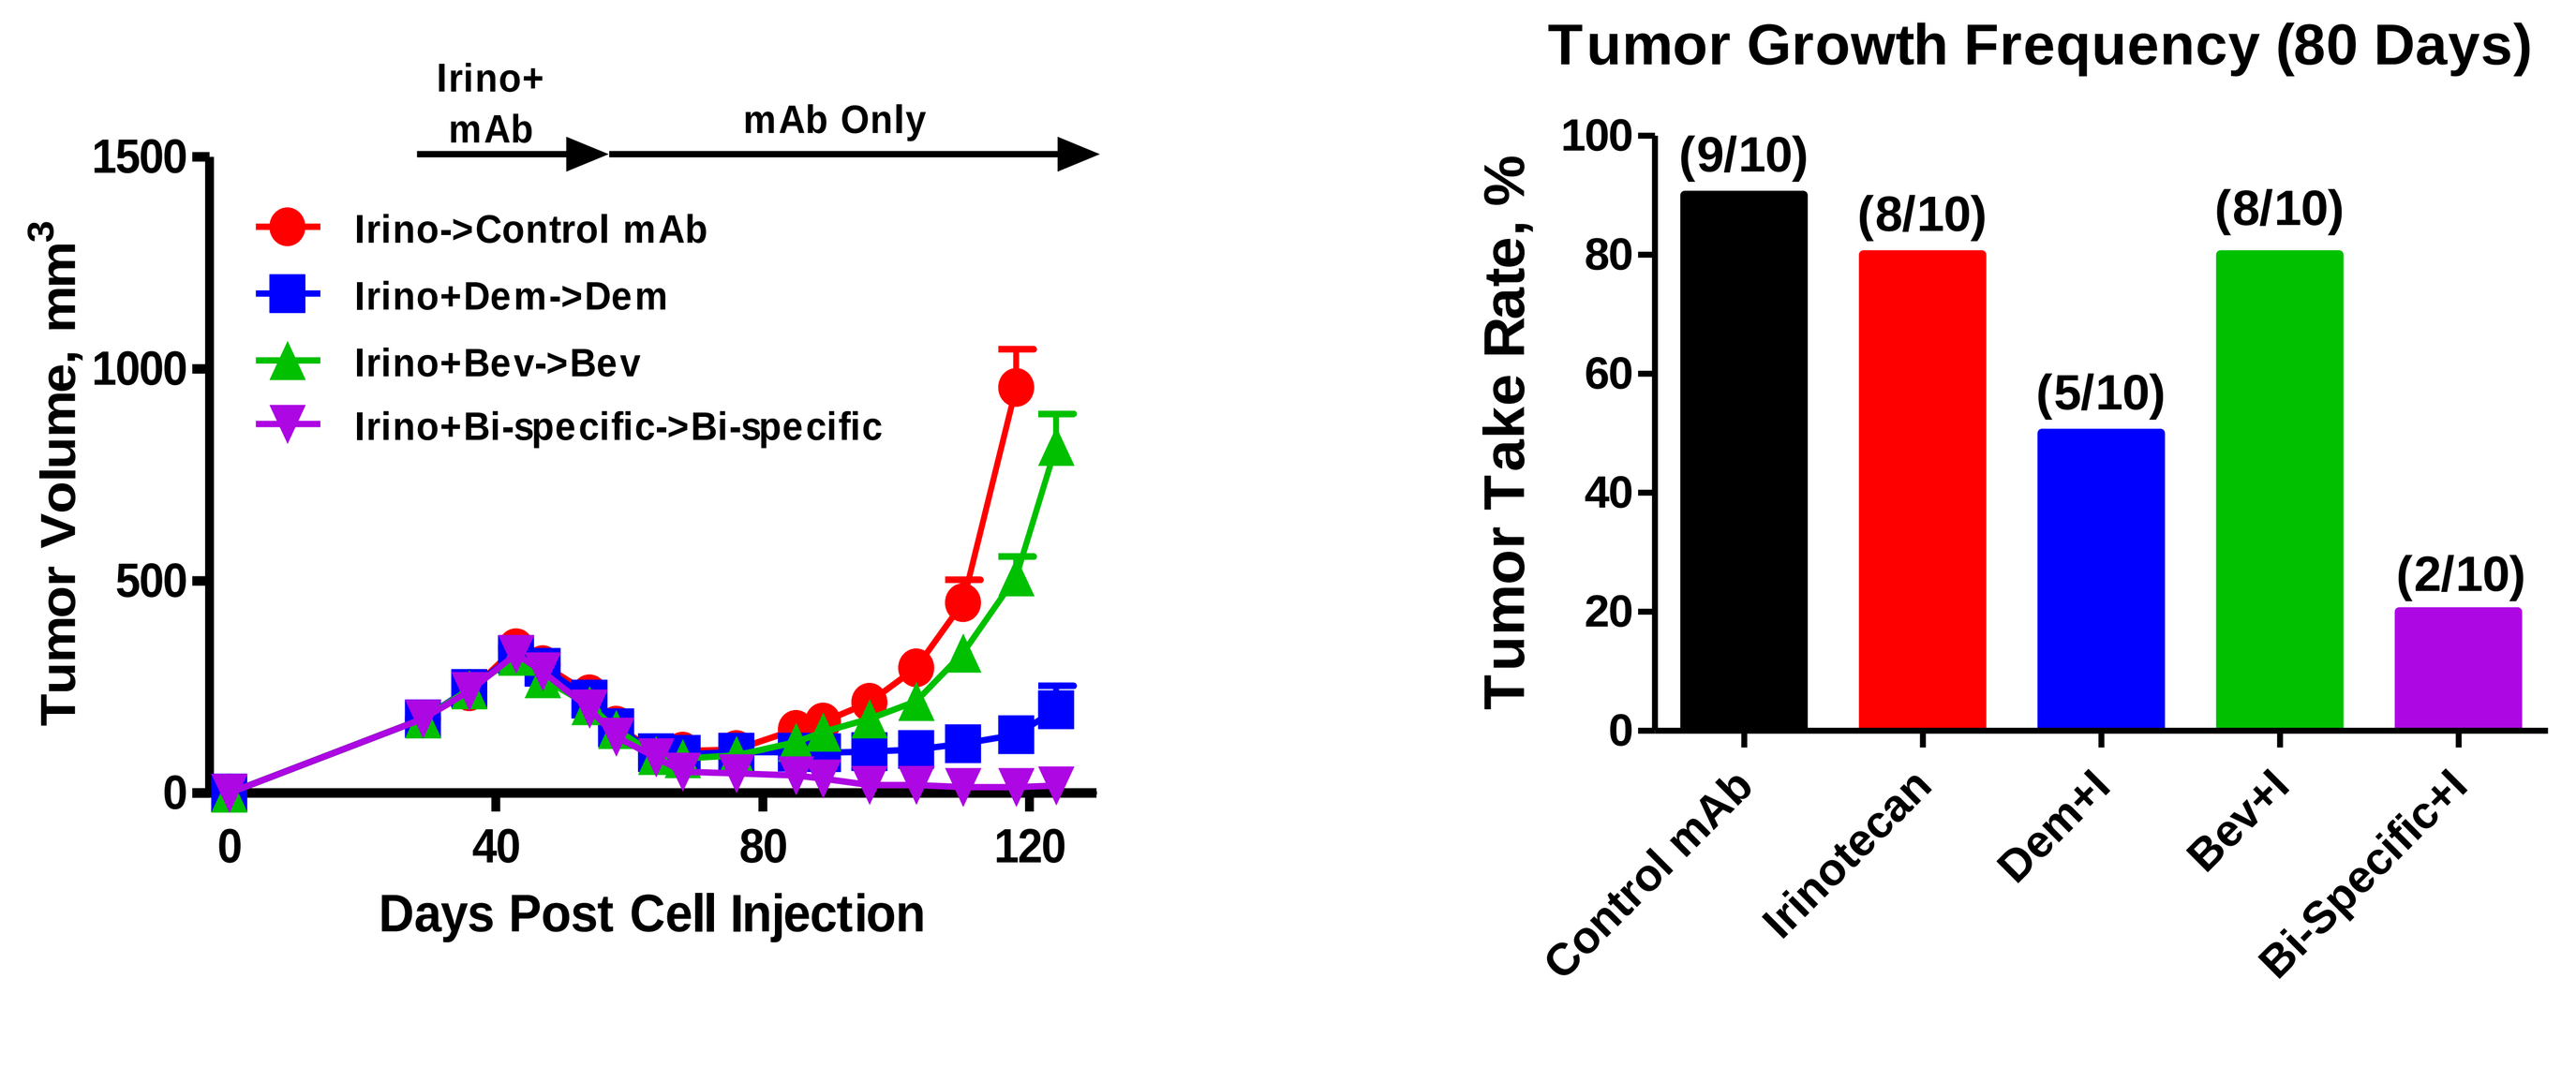
Irino+
mAb
mAb Only
Irino->Control mAb
Irino+Dem->Dem
Irino+Bev->Bev
Irino+Bi-specific->Bi-specific
Tumor Volume, mm3
Days Post Cell Injection
Tumor Growth Frequency (80 Days)
Tumor Take Rate, %
(9/10)
(8/10)
(5/10)
(8/10)
(2/10)
Control mAb
Irinotecan
Dem+I
Bev+I
Bi-Specific+I

Figure 4: Anti-DLL4/VEGF Bi-specific Antibody Delay Tumor Recurrence and Reduces Colon Cancer Stem Cell Frequency. Left: OMP-C8 colon tumors were treated with either irinotecan, bevacizumab, demcizumab or bi-specific mAb once a week for 5 weeks. Thereafter, irinotecan was terminated and followed with mAb alone. Right: Control and treated OMP-C8 colon tumors were were harvested 4 weeks post treatment, isolated into single cell suspension and serially passaged into NOD/SCID mice. Tumor take was analyzed 80 days later. *:p<0.05 vs. control mAb, **: p<0.05 vs. irinotecan by one-way ANOVA.

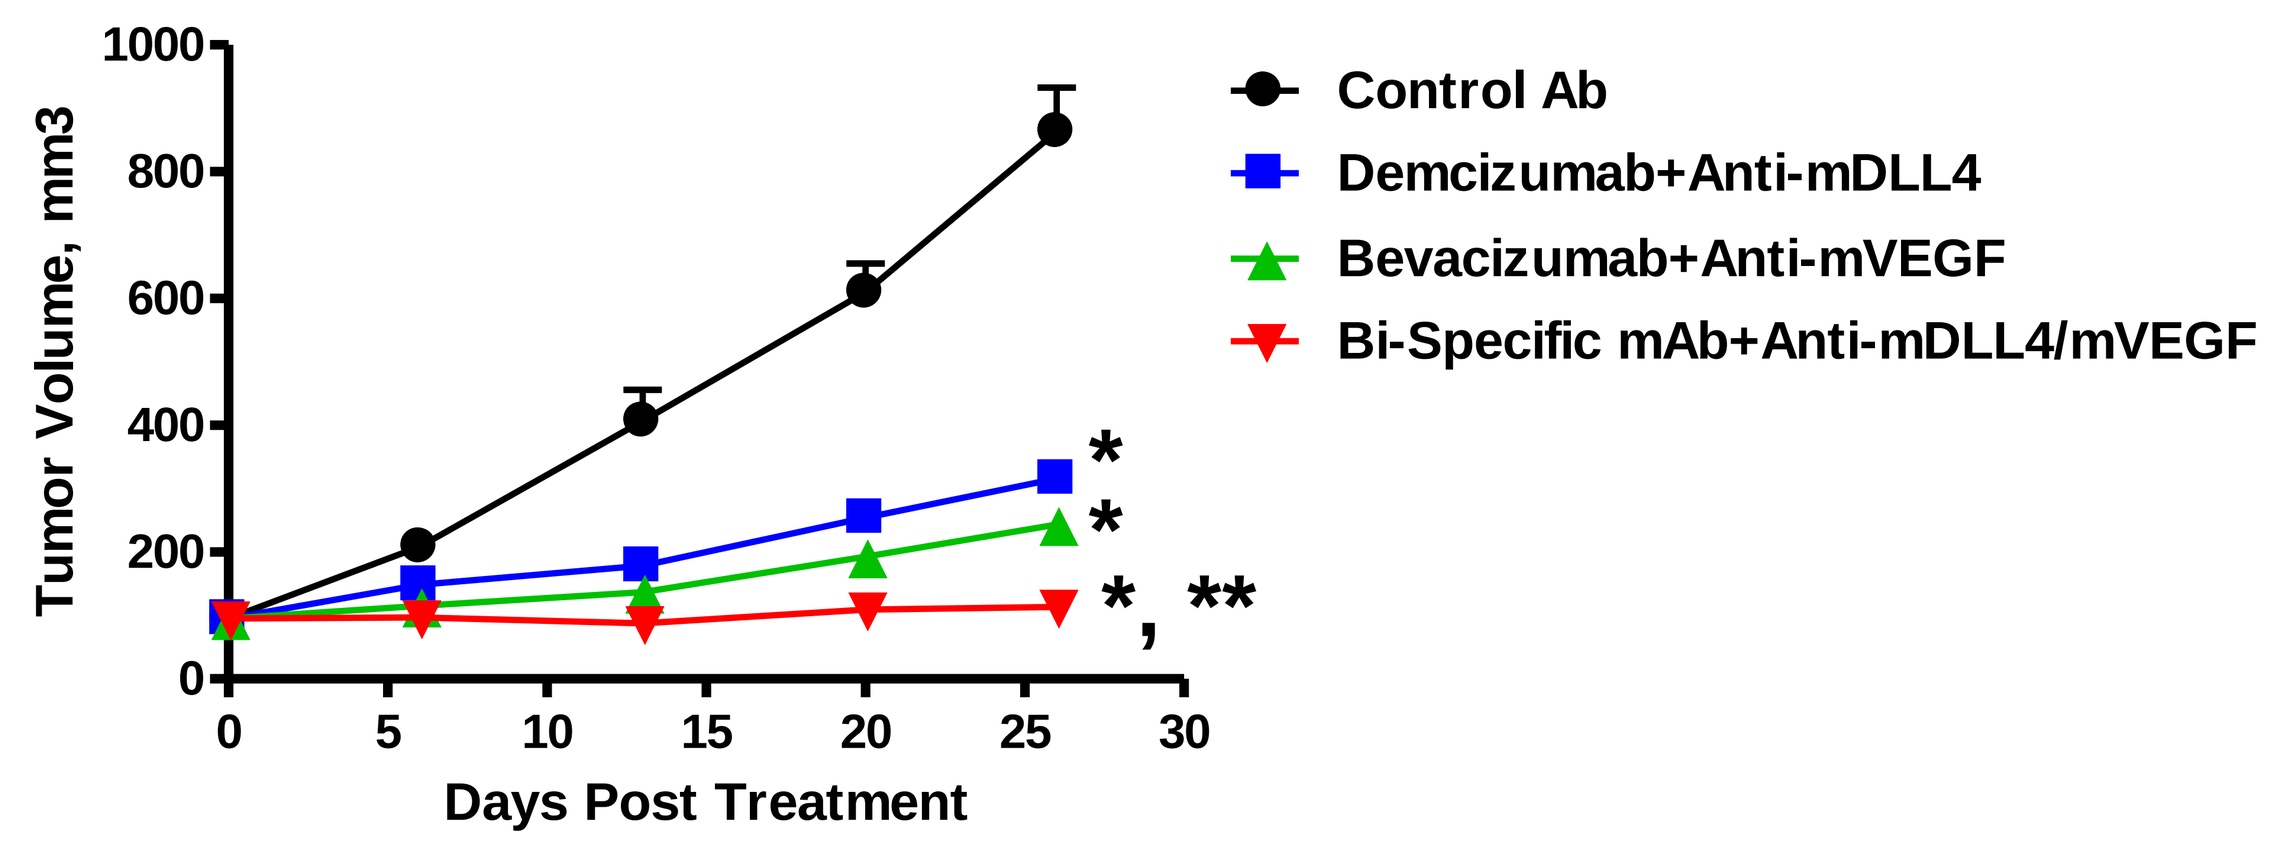
Control Ab
Demcizumab+Anti-mDLL4
Bevacizumab+Anti-mVEGF
Bi-Specific mAb+Anti-mDLL4/mVEGF
Tumor Volume, mm3
Days Post Treatment

Figure 5: Anti-Tumor Activity in Ovarian Xenograft Tumors Resulting from Simultaneous, Complete Blockade of DLL4 and VEGF. OMP-OV40 ovarian tumors were treated with either control mAb, anti-VEGF, anti-DLL4 or bi-specific mAb at 10 mg/kg once a week for 4 weeks. *:p<0.05 vs. control mAb, **: p<0.05 vs. anti-VEGF/anti-DLL4 alone by two-way ANOVA.

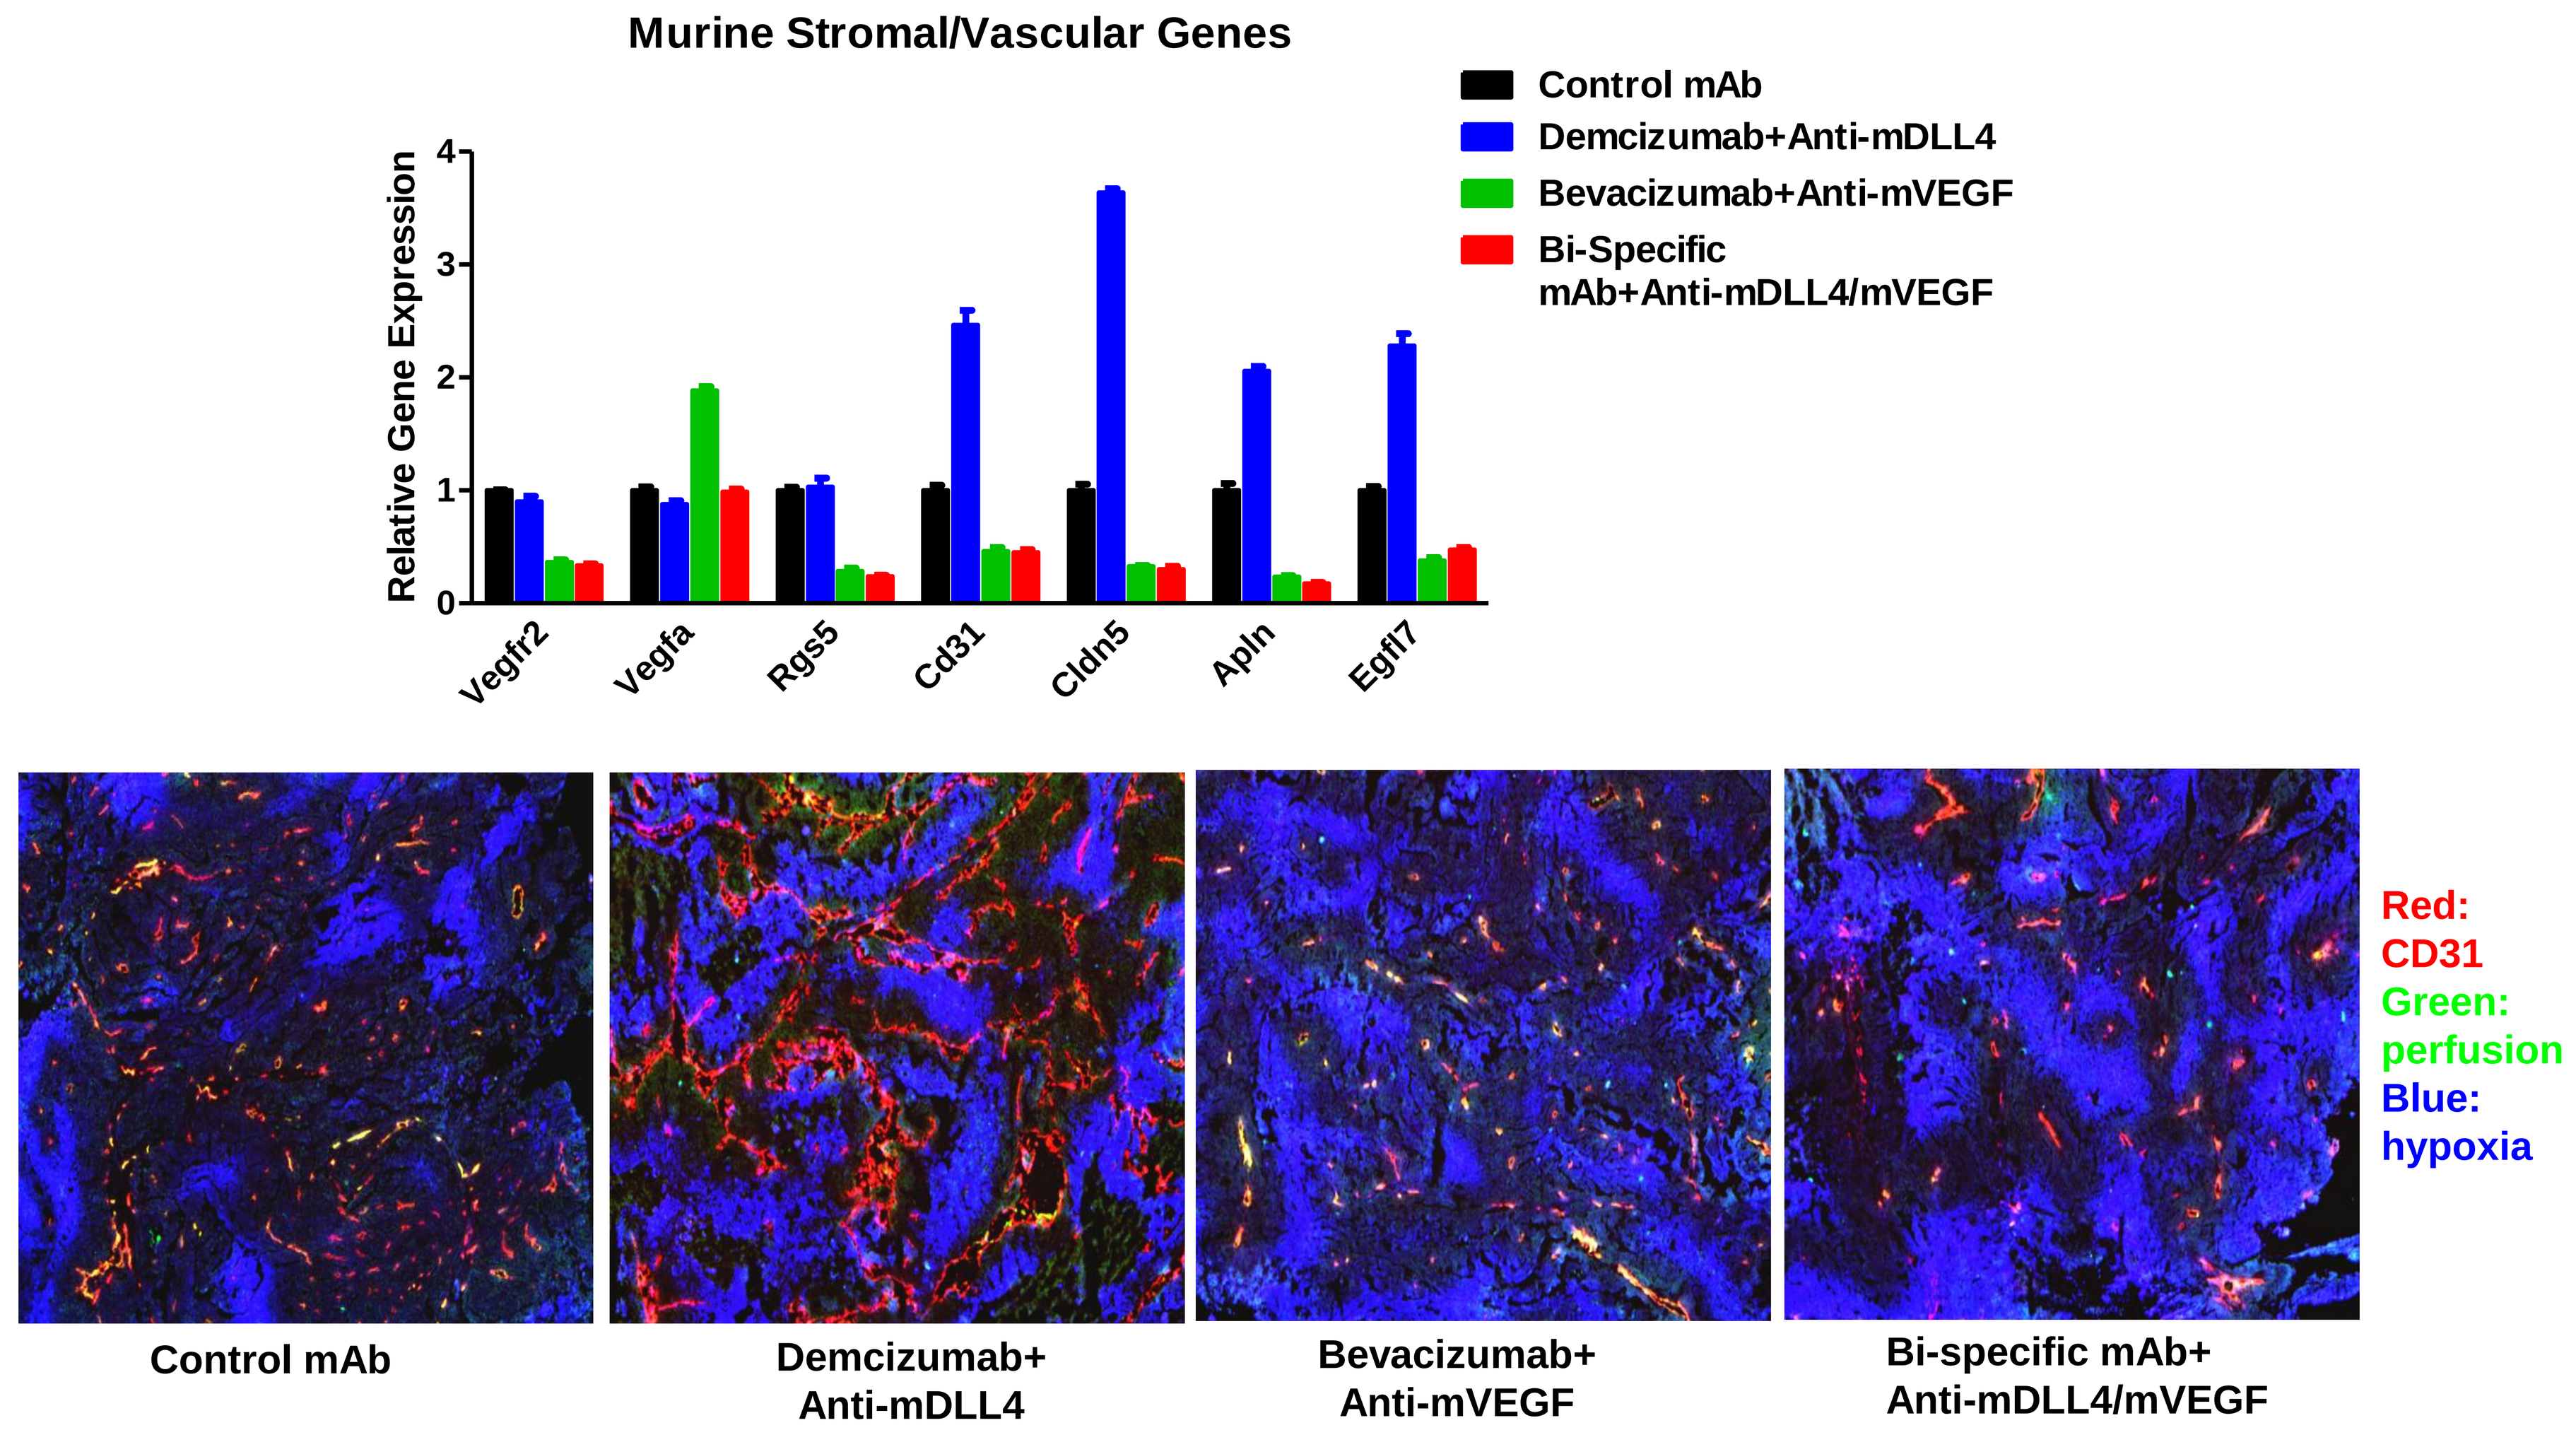
Murine Stromal/Vascular Genes
Control mAb
Demcizumab+Anti-mDLL4
Bevacizumab+Anti-mVEGF
Bi-Specific mAb+Anti-mDLL4/mVEGF
Relative Gene Expression
Vegfr2
Vegfa
Rgs5
Cd31
Cldn5
Apln
Egfl7
Red: CD31
Green: perfusion
Blue: hypoxia
Control mAb
Demcizumab+ Anti-mDLL4
Bevacizumab+ Anti-mVEGF
Bi-specific mAb+ Anti-mDLL4/mVEGF

Figure 6. Anti-VEGF Effect is Dominated Over Anti-DLL4 Effect on Angiogenic Hyperproliferation. Control and treated tumors at the conclusion of the study from Figure 5 were harvested and used for gene expression (top) and histologic analysis (Bottom). Images were taken at 20x.

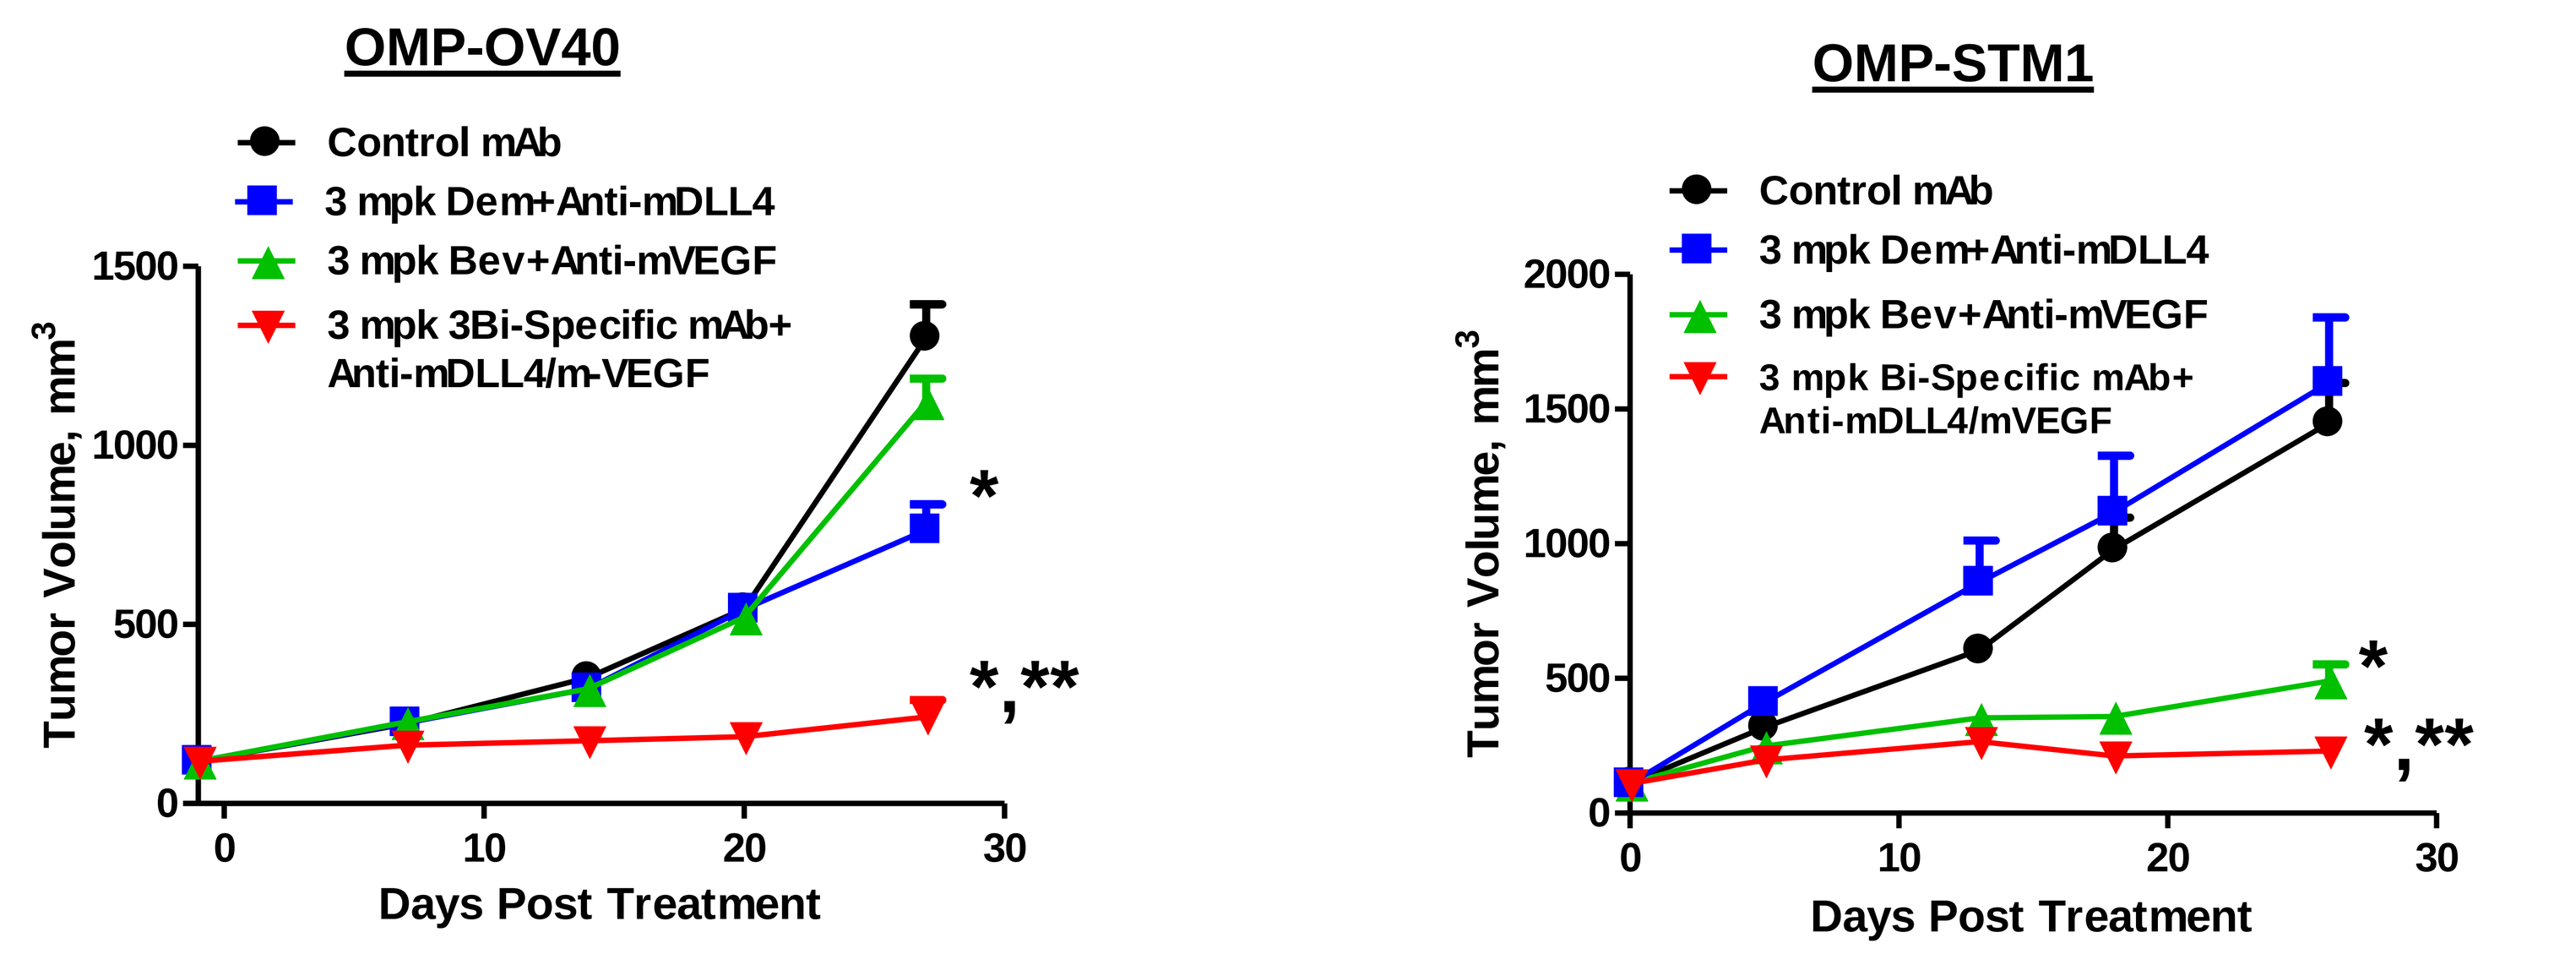
OMP-OV40
Control mAb
3 mpk Dem+Anti-mDLL4
3 mpk Bev+Anti-mVEGF
3 mpk 3Bi-Specific mAb+ Anti-mDLL4/m-VEGF
Tumor Volume, mm3
Days Post Treatment
OMP-STM1
Control mAb
3 mpk Dem+Anti-mDLL4
3 mpk Bev+Anti-mVEGF
3 mpk Bi-Specific mAb+ Anti-mDLL4/mVEGF
Tumor Volume, mm3
Days Post Treatment

Figure 7. Bi-Specific mAb Produces an Enhanced Anti-tumor Effect Compared to Either Agent Alone at Suboptimal Dose. Ovarian tumor OMP-OV40 and gastric tumor OMP-STM1 were treated with control mAb, 3 mg/kg of anti-hmDLL4, anti-hmVEGF or the bi-specific mAbs once a week for 4 weeks. *:p<0.05 vs. control mAb; **: p<0.05 vs. anti-VEGF/anti-DLL4 alone by two-way ANOVA.

CONCLUSIONS

- Anti-DLL4/Anti-VEGF bi-specific mAb has significant activity in xenograft tumor models
- Simultaneous inhibition of DLL4 and VEGF produces anti-tumor effect superior to anti-DLL4 or anti-VEGF alone
- Simultaneous inhibition of DLL4 and VEGF induces significant down-regulation of vasculature-related genes and decreases vasculature density, suggesting a dominant anti-VEGF-mediated angiogenic effect over the anti-DLL4 effect on endothelial cell hyperproliferation
- The bi-specific antibody has superior effect over anti-DLL4 alone on delaying tumor recurrence and reducing cancer stem cell frequency in tumors
- We have developed a clinical candidate and working toward an IND filing in 2014