A Phase 1b Study of the Anti-Cancer Stem Cell Agent Demcizumab (DEM, anti-DLL4) and Gemcitabine (GEM) with or without Nab-Paclitaxel in Patients with Pancreatic Cancer

[1]Hidalgo M, [2]Cooray P, [3]Carrato A, [4]Jameson M [5]Parnis F, [6]Jeffery M, [7]Grimison P, [8]Stagg R, [8]Holmgren E, [8]Kapoun AM, [8]Dupont J, [8]Yen WC, [9]Tebbutt N

[1]START, Madrid, Spain; [2]Box Hill Hospital, Box Hill, Australia; [3]Ramon y Cajal Hospital, Madrid; Spain; [4]Waikato Hospital, Hamilton, New Zealand; [5]Adelaide Cancer Centre, Adelaide, Australia; [6]Christchurch Hospital, Christchurch, New Zealand; [7]Sydney Cancer Centre, Sydney, Australia; [8]OncoMed, Redwood City, CA; and [9]Austin Hospital, Heidelberg, Australia.

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without written permission from ASCO and the authors of this poster.

Background

There is accumulating evidence that the cell types within tumors are heterogeneous and that a subset of the cells retain the property to self-renew and give rise to more differentiated progeny. These cells, called **Cancer Stem Cells (CSCs)** or tumor initiating cells drive tumor growth and metastasis and are more resistant to chemotherapy and radiotherapy than the remaining tumor cells. The ability to characterize the CSCs through surface markers and functional limiting tumor dilution assays, using minimally passaged human tumors, has enabled the identification of novel agents that specifically target the CSC population. One pathway which appears critical for the CSCs is the Notch pathway. The pathway is comprised of 4 Notch receptors (1-4) and 5 ligands, Jagged (1-2) and delta-like ligand (DLL1, 3 and 4). The DLL4 ligand contributes to CSC self-renewal and vascular development. **Demcizumab (DEM)** is a humanized IgG2 antibody that binds to DLL4. In minimally passaged human tumor xenografts, DEM was observed to have activity against a variety of tumors including colorectal cancer, breast cancer, lung cancer, pancreatic cancer, melanoma and ovarian cancer. The impact of treatment on the frequency of tumorigenicity was assessed using a limiting dilution assay. In several models, using different chemotherapeutic agents, while the chemotherapy alone decreased tumor volume, the frequency of tumor initiating cells was increased in the residual tumor. In contrast, DEM alone decreased the frequency of CSCs and the greatest reduction was observed when DEM was combined with GEM and nab-paclitaxel.

Nonclinical Xenograft Data

Comparison of Anti-DLL4 Combination Effect with GEM vs. GEM + Nab-Paclitaxel in a Patient Derived Pancreatic Xenograft (OMP-PN16)

Reduction in Tumor Volume

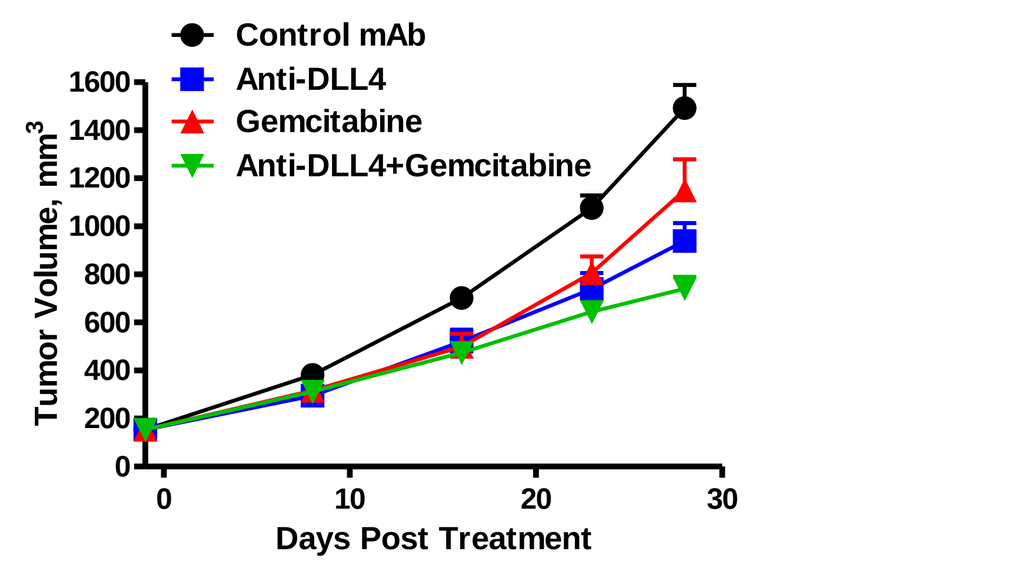
Control mAb
Anti-DLL4
Gemcitabine
Anti-DLL4+Gemcitabine
Tumor Volume, mm³
Days Post Treatment

Reduction in Tumor Volume

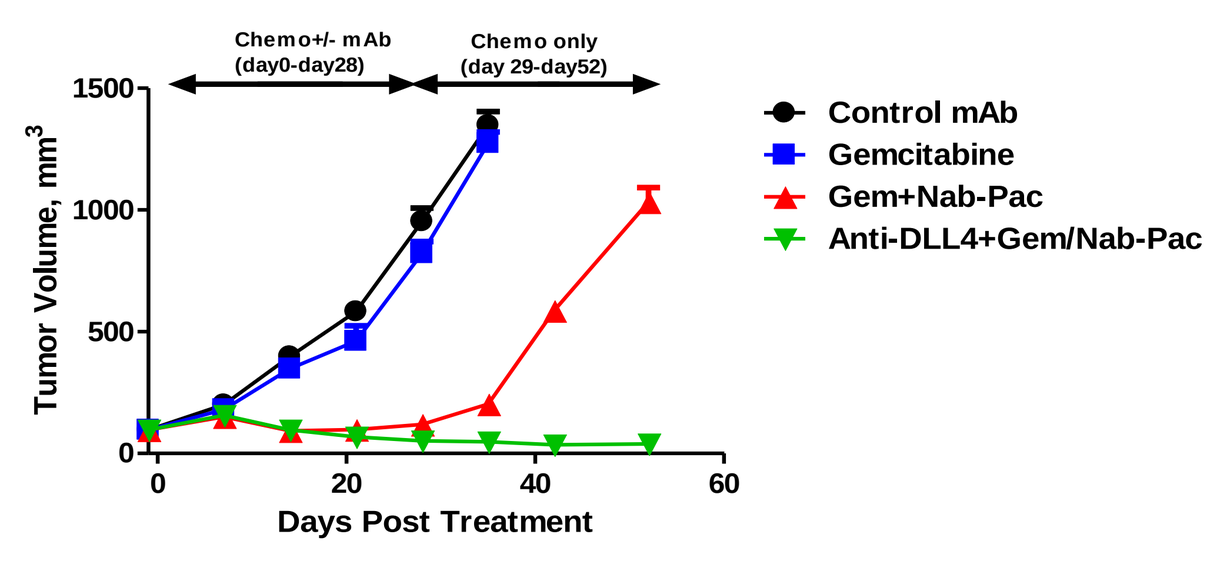
Chemo+/- mAb (day0-day28)
Chemo only (day 29-day52)
Control mAb
Gemcitabine
Gem+Nab-Pac
Anti-DLL4+Gem/Nab-Pac
Tumor Volume, mm³
Days Post Treatment

Activity of Anti-DLL4 in Combination with GEM Plus Nab-Paclitaxel in Patient Derived Pancreatic Xenograft (OMP-PN13)

Reduction in Tumor Volume

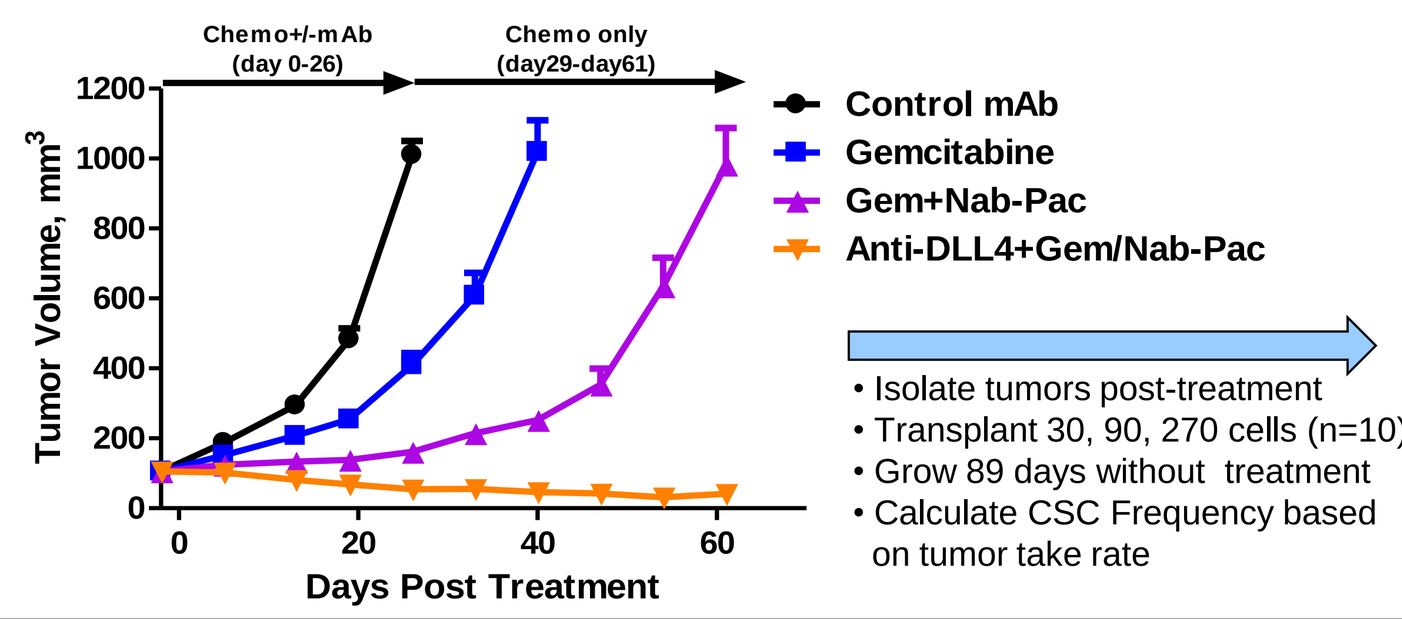
Chemo+/-mAb (day 0-26)
Chemo only (day29-day61)
Control mAb
Gemcitabine
Gem+Nab-Pac
Anti-DLL4+Gem/Nab-Pac
Tumor Volume, mm³
Days Post Treatment

- Isolate tumors post-treatment
- Transplant 30, 90, 270 cells (n=10)
- Grow 89 days without treatment
- Calculate CSC Frequency based on tumor take rate

CSC Frequency

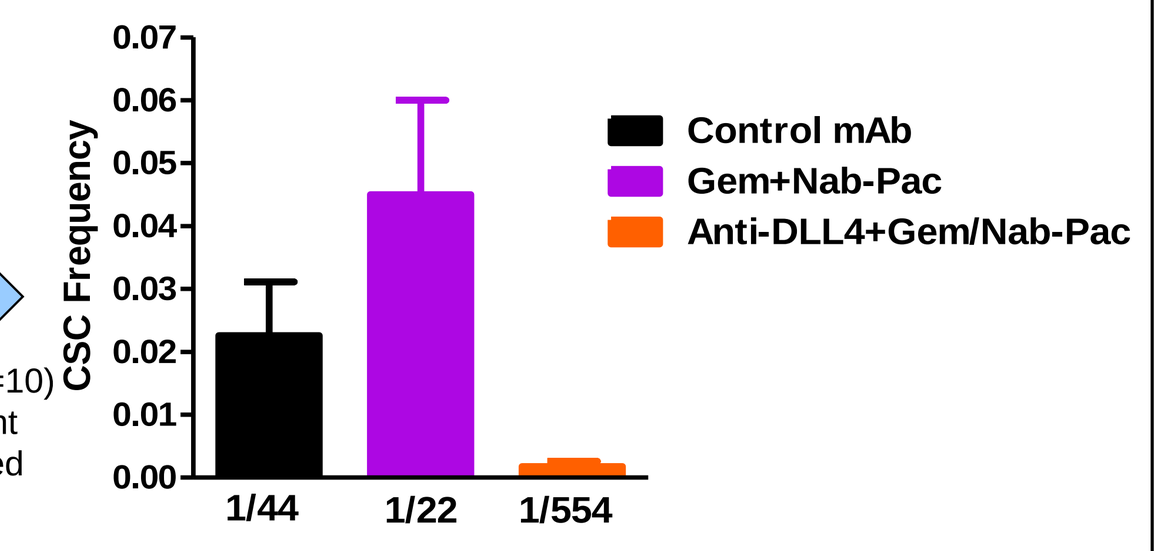
CSC Frequency
Control mAb
Gem+Nab-Pac
Anti-DLL4+Gem/Nab-Pac
1/44
1/22
1/554

Methods

This is an open-label Phase 1b dose escalation study of DEM plus GEM with or without nab-paclitaxel in pts with 1st line pancreatic cancer. The study endpoints included: 1) safety, 2) maximum tolerated dose (MTD), 3) immunogenicity, 4) pharmacokinetics (PK), 5) antitumor activity, and 6) biomarkers of Notch signaling and CSCs. Pts received DEM 2.5 mg/kg Q2W or Q4W, or 5mg/kg Q4W with GEM 1000 mg/m^2 given 7 of the 1st 8 weeks & then 3 of every 4 weeks or GEM 1000 mg/m^2 and nab-paclitaxel 125 mg/m^2 on Days 0, 7 and 14 every 28-days until disease progression. Dosing of subjects in the 1st cohort was paused due to emergence of reversible cardiotoxicity secondary to DEM in other ongoing studies with prolonged treatment of DEM. The protocol was amended to include a cardiac risk mitigation plan including cardiac monitoring with B-type natriuretic peptide (BNP) and echocardiography and the administration of cardioprotective medication (i.e, an angiotensin-converting enzyme inhibitor or carvedilol) for rising BNP. As reversible cardiopulmonary toxicity occurred in 1 patient in the 3rd cohort receiving 5 mg/kg who was dosed for more than 126 days, the subjects in the subsequent 4 cohorts received truncated dosing of DEM (i.e. 70 days of therapy) and gemcitabine with nab-paclitaxel. A DSMB reviewed the data from each dose cohort after the last subject in that cohort had been on study for 56 days to decide whether it was safe to proceed to the next dose cohort. Data through April 7, 2015 are presented.

Patient Demographics (n=56)

Dose Level – mg/kg	2.5 every 2 weeks DEM/ GEM	2.5 every 4 weeks DEM/ GEM	5 every 4 weeks DEM/ GEM	2.5* every 2 weeks DEM/ GEM/ Nab-Pac	5* every 2 weeks DEM/ GEM/ Nab-Pac	3.5* every 2 weeks DEM/ GEM/ Nab-Pac	3.5* every 2 weeks DEM/ GEM/ Nab-Pac	Total
N	8	8	8	6	8	9	9	56
Median age (years)	63.5	63.5	69	70.5	59	65	63	65
Male/Female	4/4	6/2	3/5	3/3	3/5	2/7	5/4	26/30
Locally Advanced/ Metastatic at Diagnosis	3/5	0/8	0/8	2/4	4/4	3/6	5/4	17/39
Prior Surgery	1	2	3	3	2	1	1	13
Prior Neoadjuvant/ Adjuvant Therapy	1	-	1	1	2	1	-	6
Prior Radiotherapy	-	-	-	-	1	-	-	1

* Truncated demcizumab dosing for 70 days

Related AEs >15% Pts (n=56) All Grades by Dose Level (mg/kg)

Dose Level – mg/kg	2.5 every 2 weeks DEM/ GEM	2.5 every 4 weeks DEM/ GEM	5 every 4 weeks DEM/ GEM	Total DEM/ GEM	2.5* every 2 weeks DEM/ GEM/ Nab-Pac	5* every 2 weeks DEM/ GEM/ Nab-Pac	3.5* every 2 weeks DEM/ GEM/ Nab-Pac	3.5* every 2 weeks DEM/ GEM/ Nab-Pac	Total DEM*/ GEM/ Nab-Pac	Total All Patients
N	8	8	8	**24**	6	8	9	9	32	56
Fatigue	2	3	2	**7 (29%)**	2	3	6	2	13 (41%)	20 (36%)
Nausea	3	-	3	**6 (25%)**	3	5	2	2	12 (38%)	18 (32%)
Vomiting	3	1	3	**7 (29%)**	2	3	1	-	6 (19%)	13 (23%)
Hypertension	2	2	3	**7 (29%)**	1	-	2	2	5 (16%)	12 (21%)
Diarrhea	-	1	1	**2 (8%)**	1	4	3	1	9 (28%)	11 (20%)
Decreased appetite	2	1	2	**5 (21%)**	2	2	1	-	5 (16%)	10 (18%)
BNP Increased	1	1	1	**3 (13%)**	1	2	2	2	7(22%)	10 (18%)
Peripheral Edema	1	3	1	**5 (21%)**	-	2	2	-	4 (13%)	9 (16%)

* Truncated demcizumab dosing for 70 days

Reversible Cardiopulmonary Toxicity* (Any Grade) (N=56)

Dose Level – mg/kg	2.5 every 2 weeks DEM/ GEM	2.5 every 4 weeks DEM/ GEM	5 every 4 weeks DEM/ GEM	Total DEM/ GEM	2.5* every 2 weeks DEM/ GEM/ Nab-Pac	5* every 2 weeks DEM/ GEM/ Nab-Pac	3.5* every 2 weeks DEM/ GEM/ Nab-Pac	3.5* every 2 weeks DEM/ GEM/ Nab-Pac	Total DEM/ GEM/ Nab-Pac
N	8	8	8	**24**	6	8	9	9	**32**
Pulmonary HTN (Reversible)	-	1	1**	**2**	-	-	-	-	**-**
Congestive heart failure (Reversible)	-	-	-	**-**	-	-	-	-	**-**
Right-sided heart failure (Reversible)	-	-	1**	**1**	-	-	-	-	**-**

* Truncated demcizumab dosing for 70 days
** Occurred following > 100 days of treatment and reversible following the discontinuation of demcizumab and medical management

Effect on Notch Pathway Gene Expression In Whole Blood (By DEM Dose Cohort)

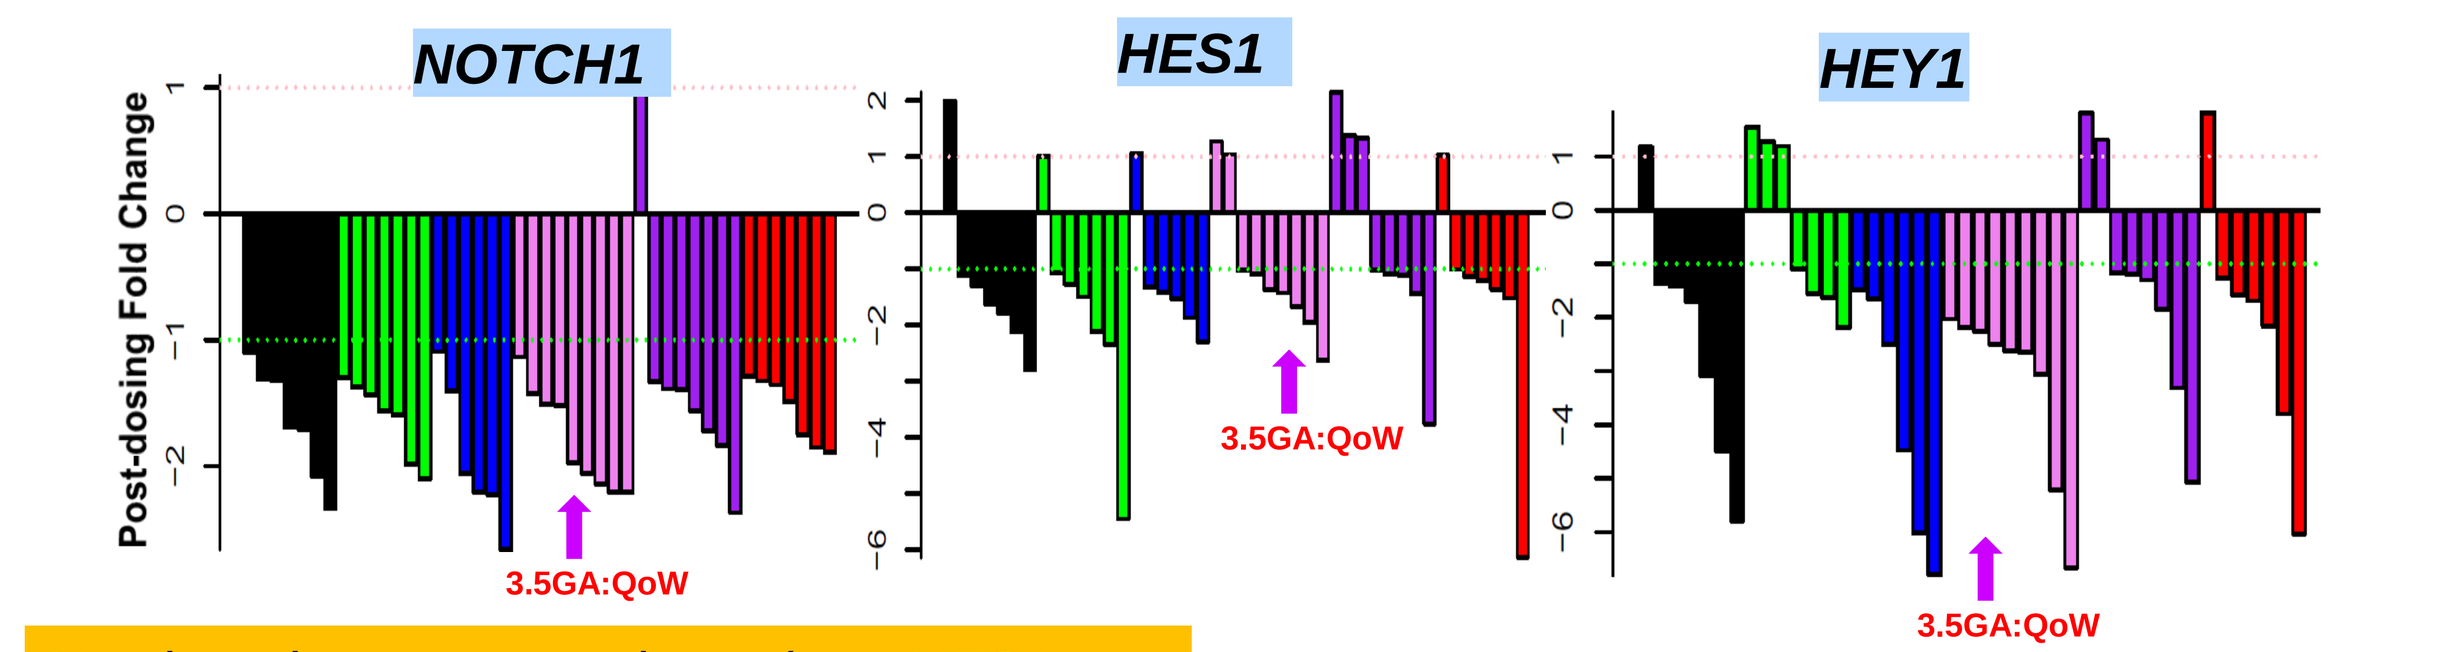
NOTCH1
HES1
HEY1
Post-dosing Fold Change
3.5GA:QoW
3.5GA:QoW
3.5GA:QoW

Notch pathway genes above (e.g. *HES1, HEY1, NOTCH1*) show clear modulation at Phase 2 dose, 3.5mg/kg GA QoW

Control genes (e.g. ACTB) are not changed

PD modulation observed up to ~150 days post last infusion in some pts

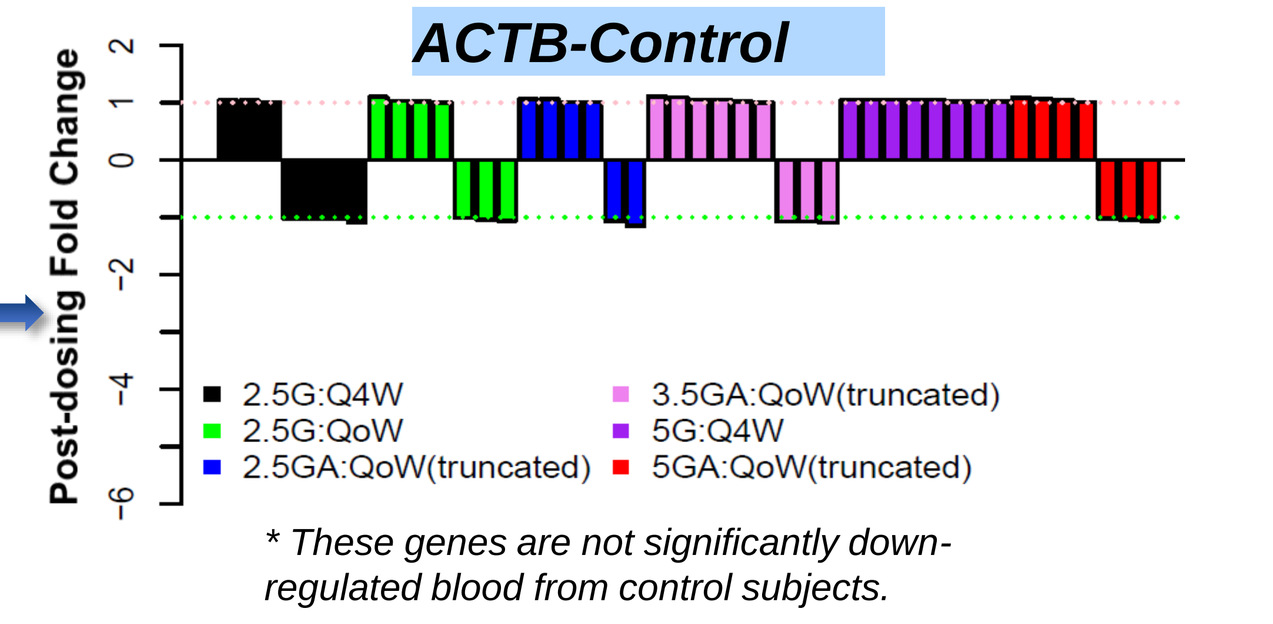
ACTB-Control
Post-dosing Fold Change
2.5G:Q4W
2.5G:QoW
2.5GA:QoW(truncated)
3.5GA:QoW(truncated)
5G:Q4W
5GA:QoW(truncated)

** These genes are not significantly down-regulated blood from control subjects.*

RECIST Best Overall Response (n=56)

Dose Level – mg/kg	2.5 every 2 weeks DEM/ GEM	2.5 every 4 weeks DEM/ GEM	5 every 4 weeks DEM/ GEM	Total DEM/GEM (Evaluable = 16)	2.5* every 2 weeks DEM/ GEM/ Nab-Pac	5* every 2 weeks DEM/ GEM/ Nab-Pac	3.5* every 2 weeks DEM/ GEM/ Nab-Pac	3.5* every 2 weeks DEM/ GEM/ Nab-Pac	Total DEM*/ GEM/ab-Pac (Evaluable = 28)
Partial Response	1	1	2	**4 (25%)**	4	3	3	4	**14 (50%)**
Stable Disease	4	2	1	**7 (44%)**	1	4	4	2	**11 (39%)**
Clinical Benefit Rate (PR + SD)	5	3	3	**11 (69%)**	5	7	7	6	**25 (89%)**
Progressive Disease	-	3	2	**5 (31%)**	1	1	1	-	**3 (11%)**
Not Evaluable	3	2	3	**8**	-	-	1	3	**4**

* Truncated demcizumab dosing for 70 days

% Change in RECIST Target Lesion Size DEM/GEM/Nab-Paclitaxel Patients

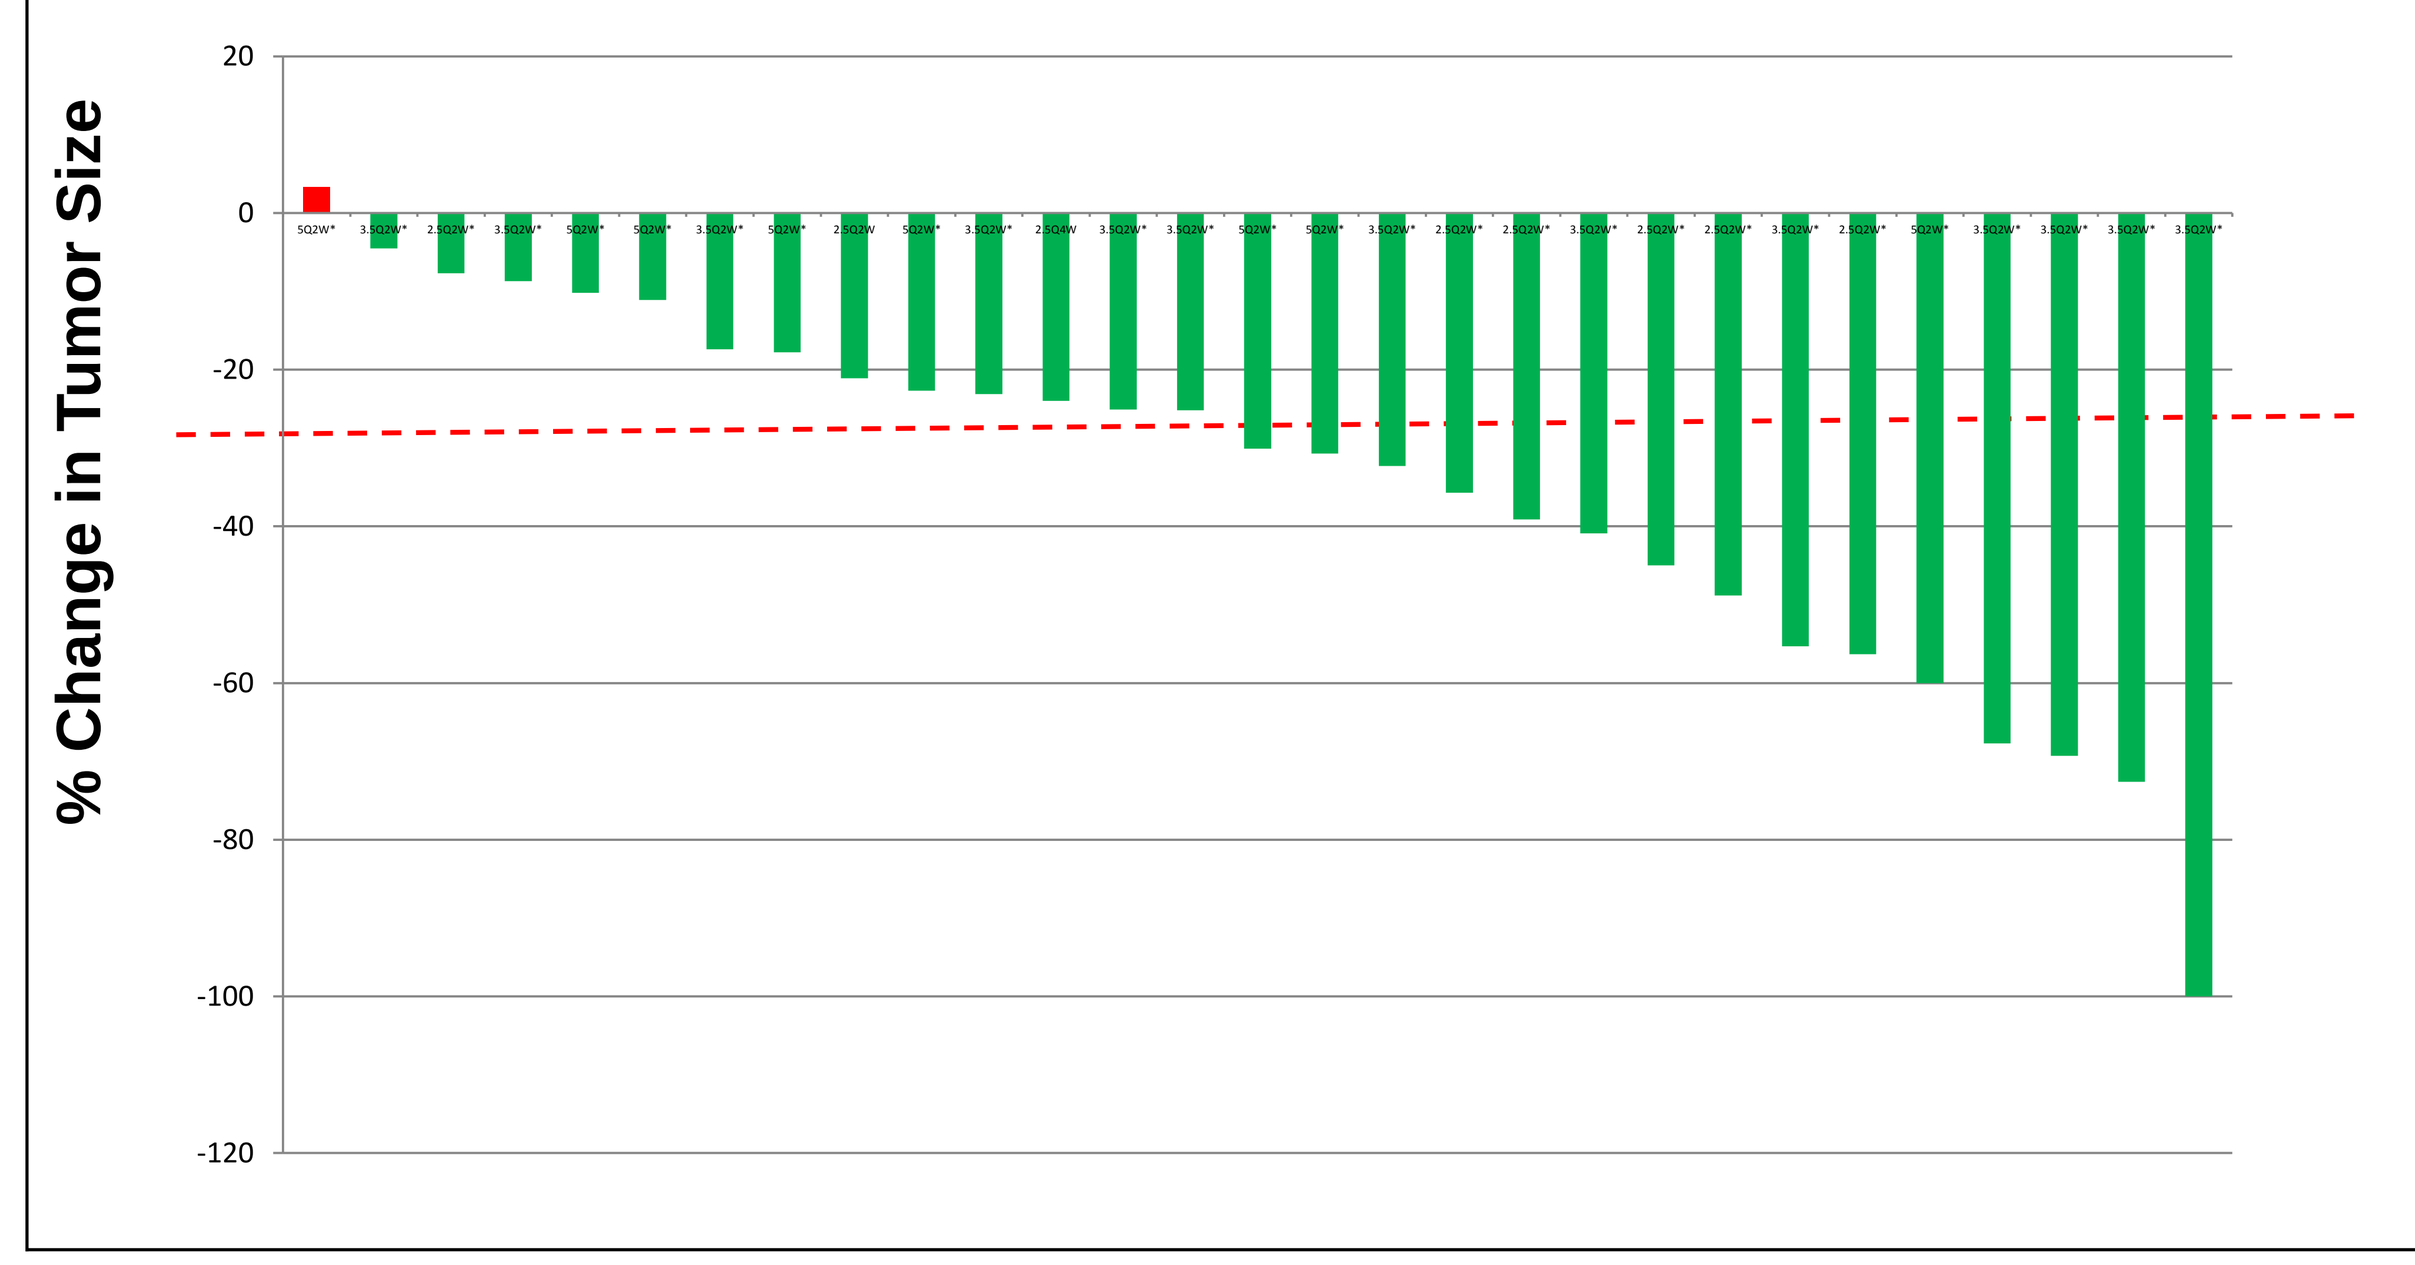
% Change in Tumor Size

Progression-Free Survival

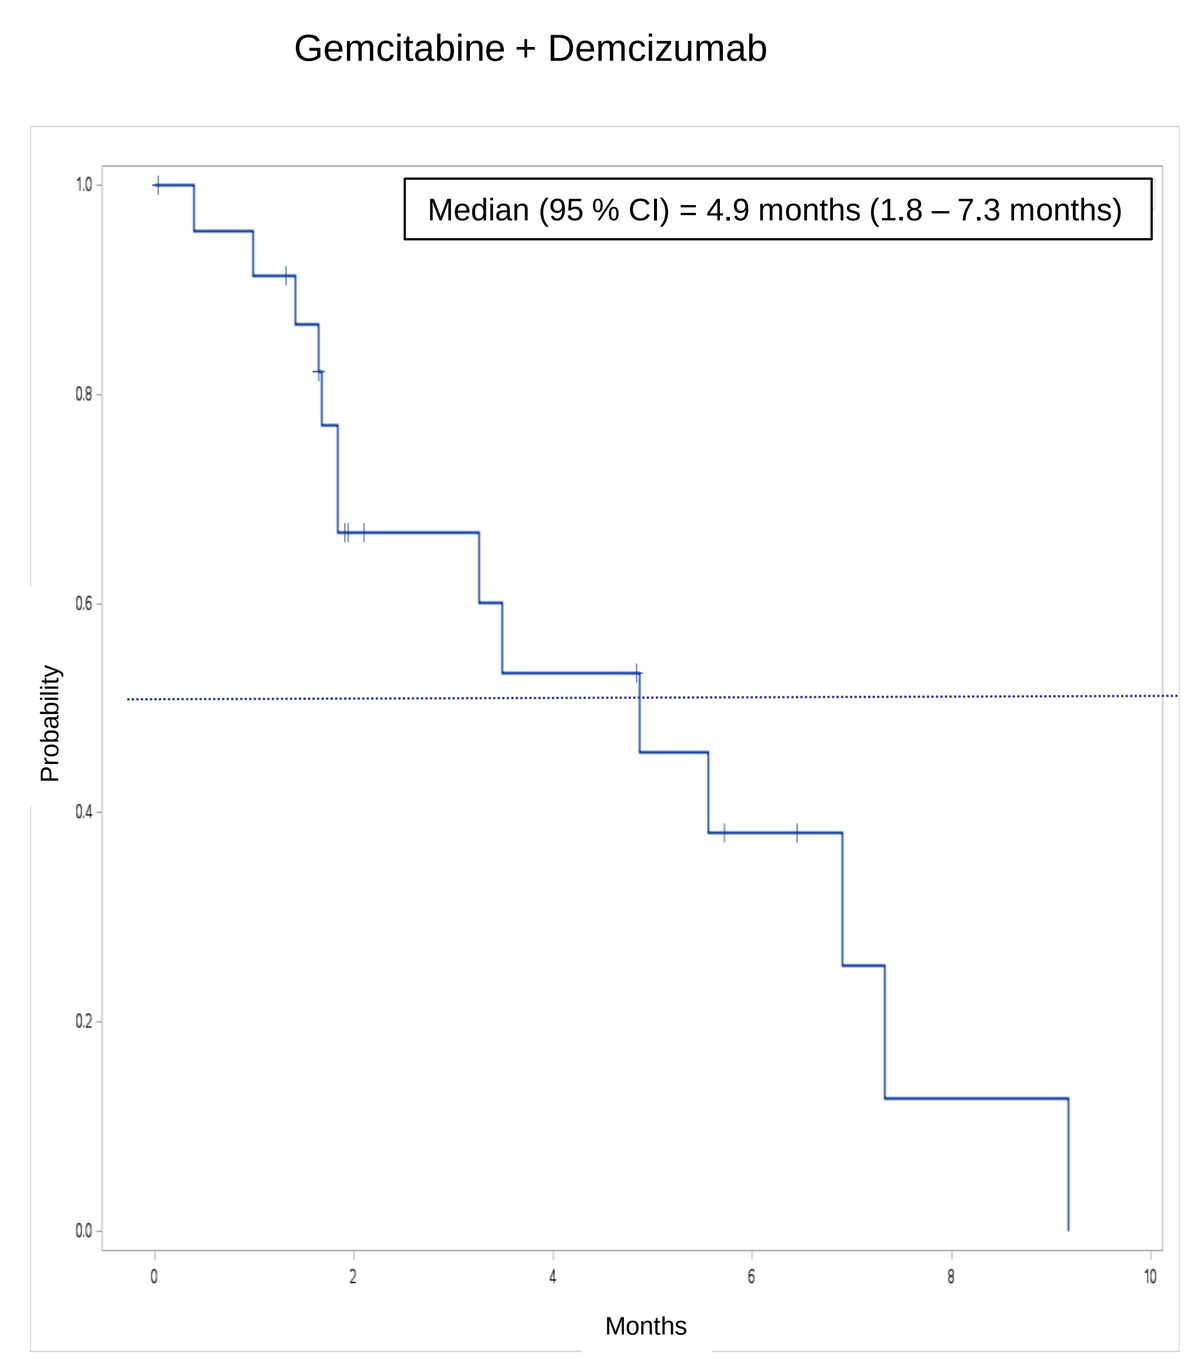
Gemcitabine + Demcizumab
Median (95 % CI) = 4.9 months (1.8 – 7.3 months)
Probability
Months

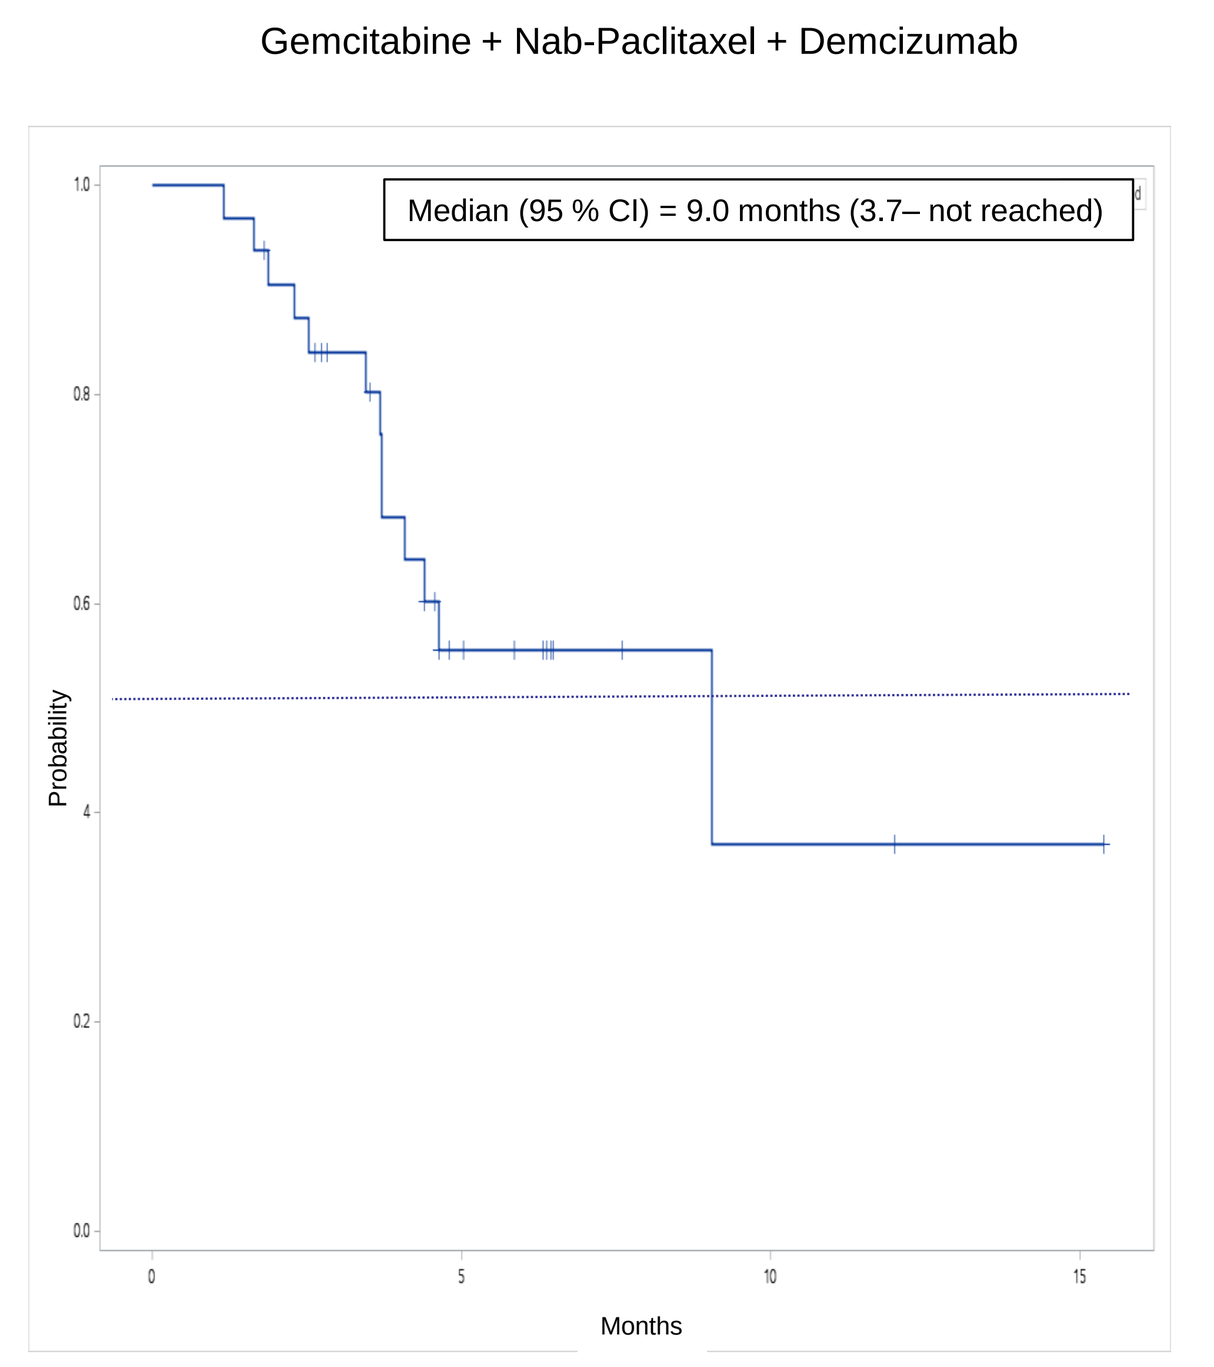
Gemcitabine + Nab-Paclitaxel + Demcizumab
Median (95 % CI) = 9.0 months (3.7– not reached)
Probability
Months

Survival DEM/GEM/Nab-Paclitaxel Patients

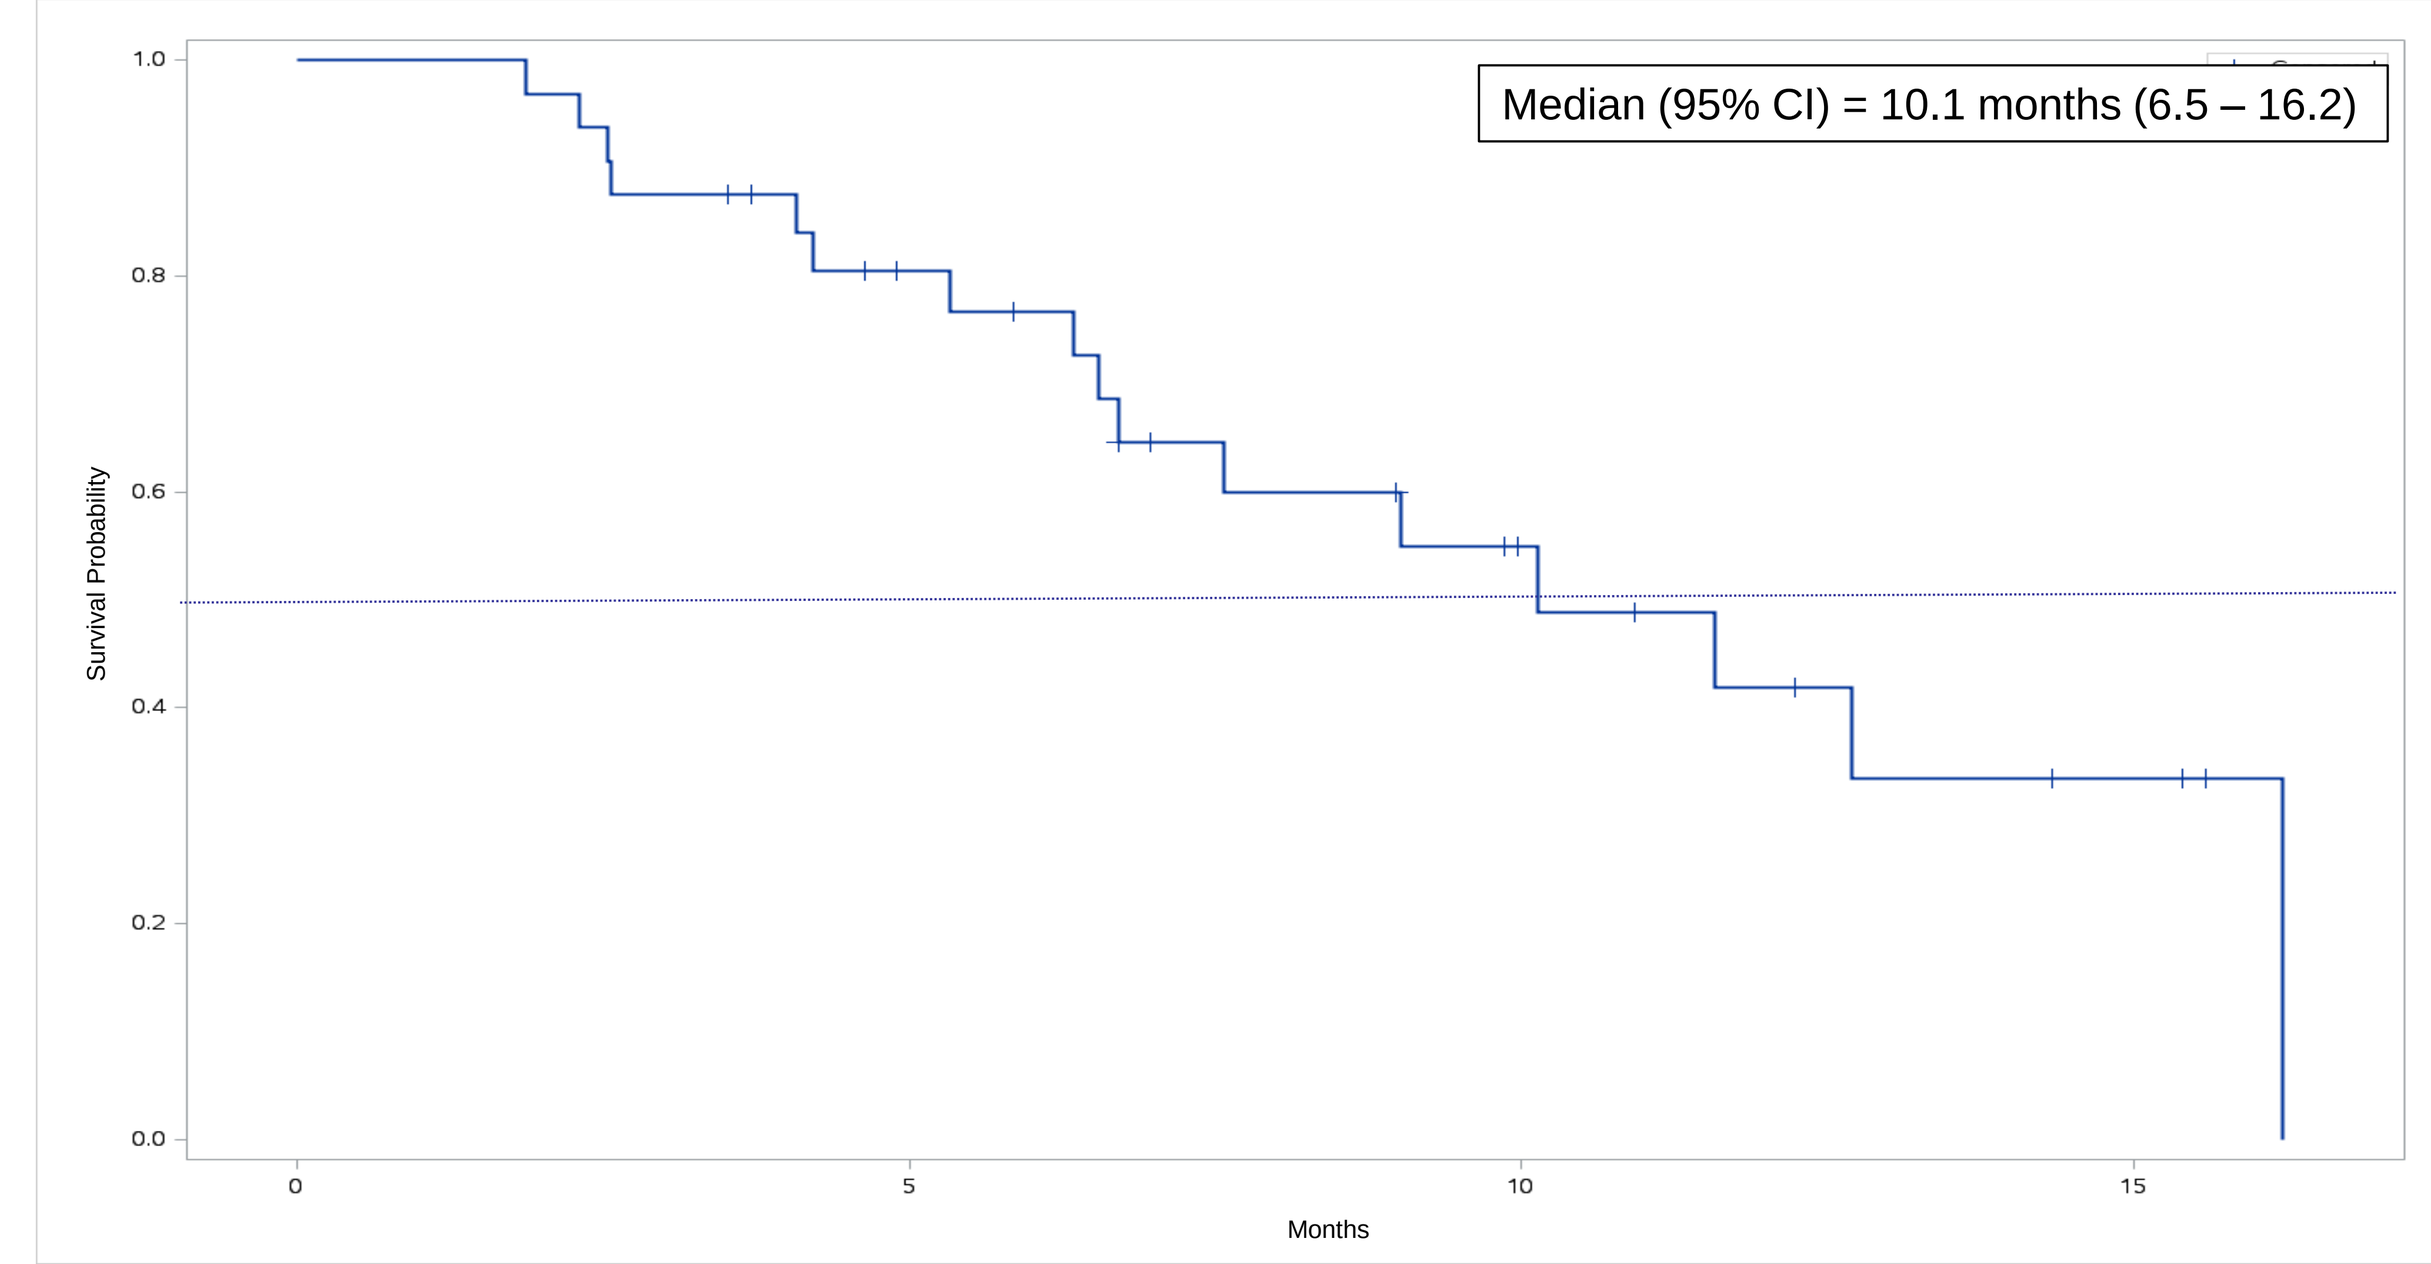
Median (95% CI) = 10.1 months (6.5 – 16.2)
Survival Probability
Months

Survival not collected in the 1st few dose cohorts beyond treatment termination. Thus, survival data not presented for the gemcitabine + demcizumab patients.

Exploratory DLL4 Expression Biomarker Analyses Truncated Patients (n = 15)

DLL4 IHC

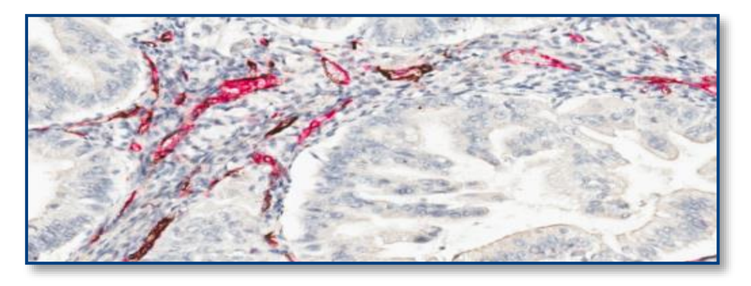

- Intra-tumoral stained vessels evaluated for CD31 & DLL4 by IHC
 - % DLL4 positive vessels/ total vessels (CD31 positive plus DLL4 positive vessels)
 - Scoring performed by a board-certified clinical anatomic pathologist

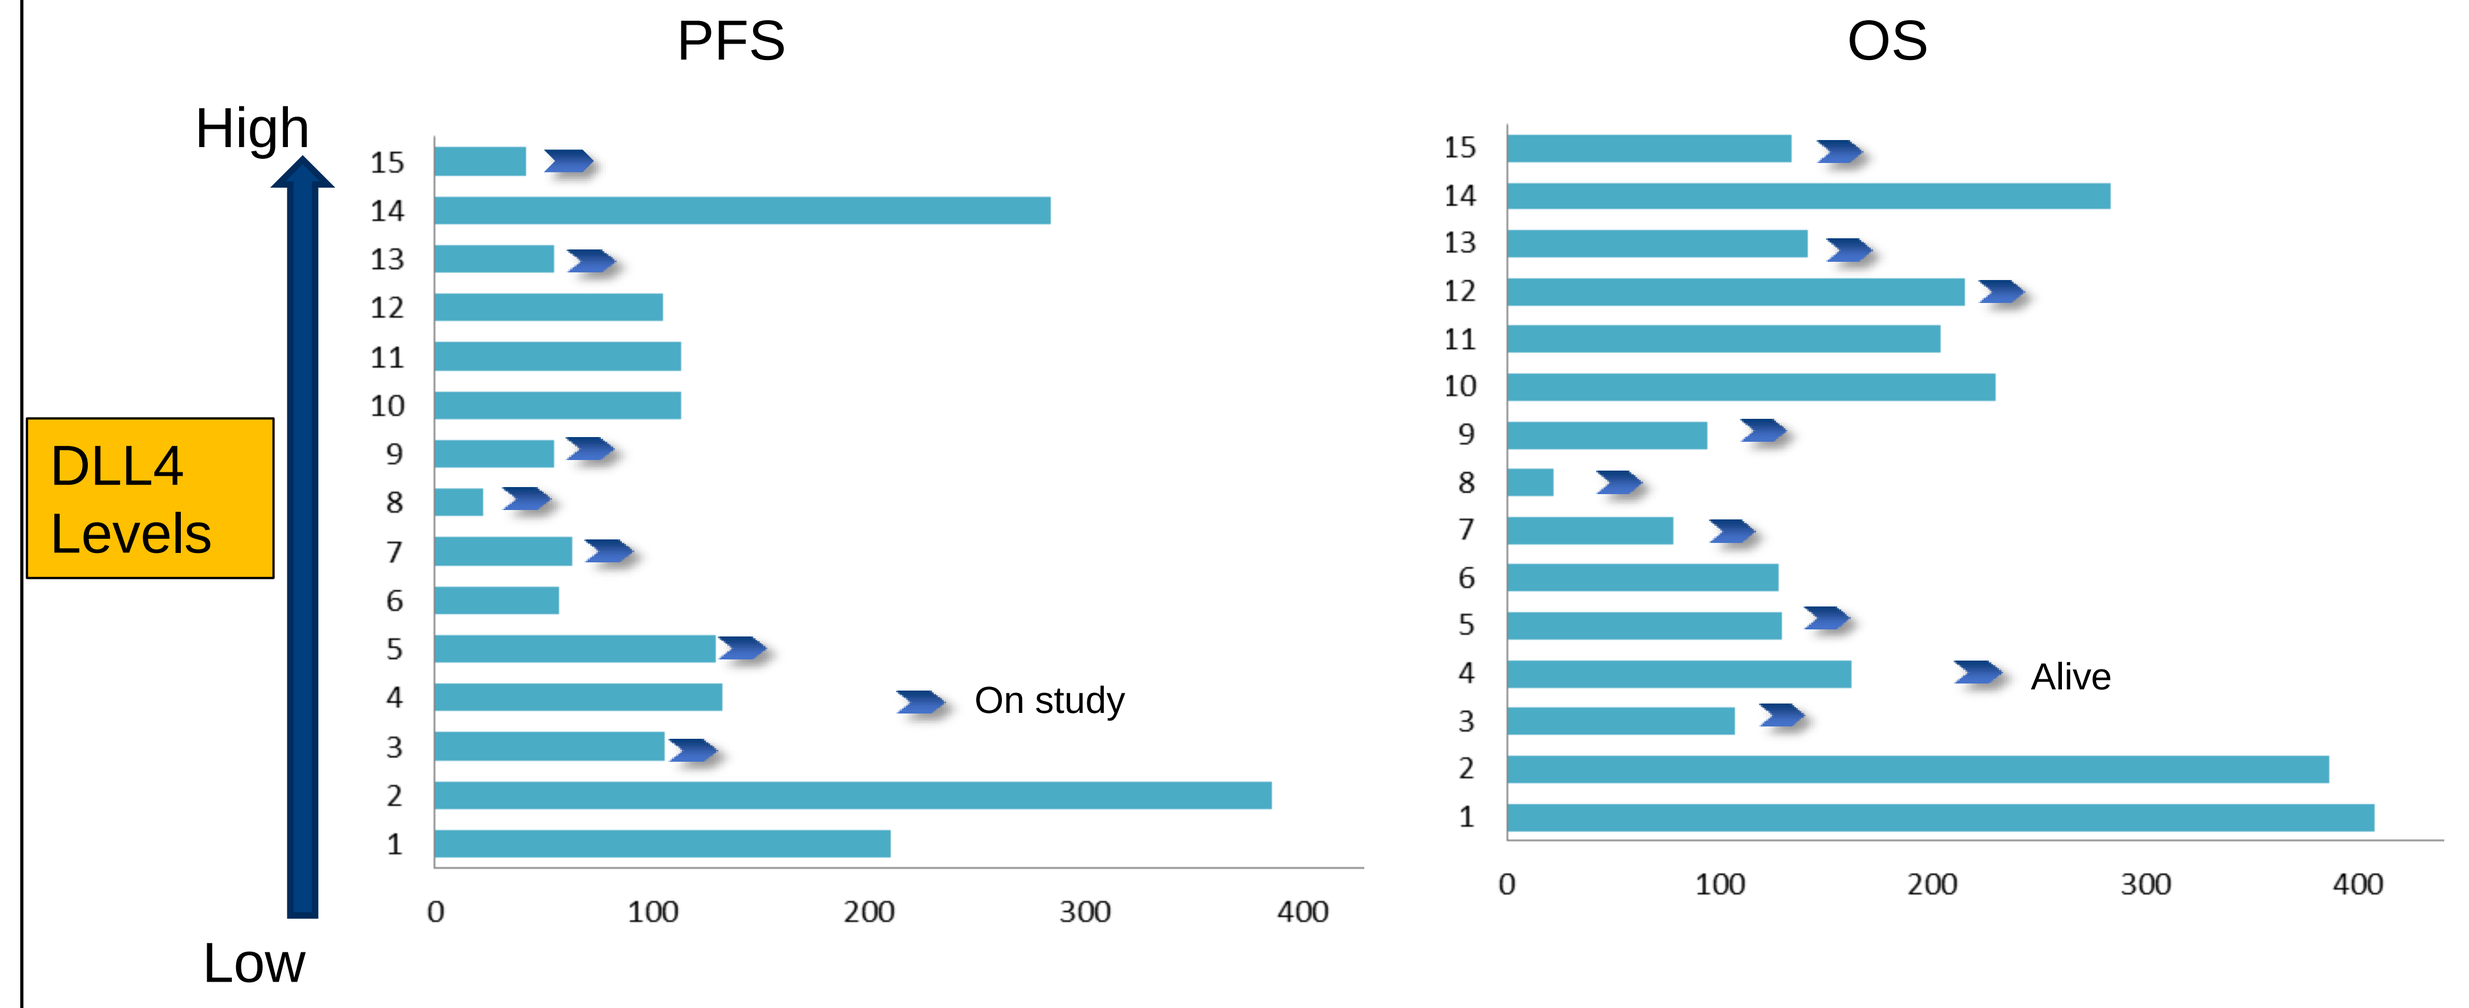
PFS
OS
High
Low
DLL4 Levels
On study
Alive

Overall Survival Exploratory Biomarker Analyses Truncated Patients (n = 15)

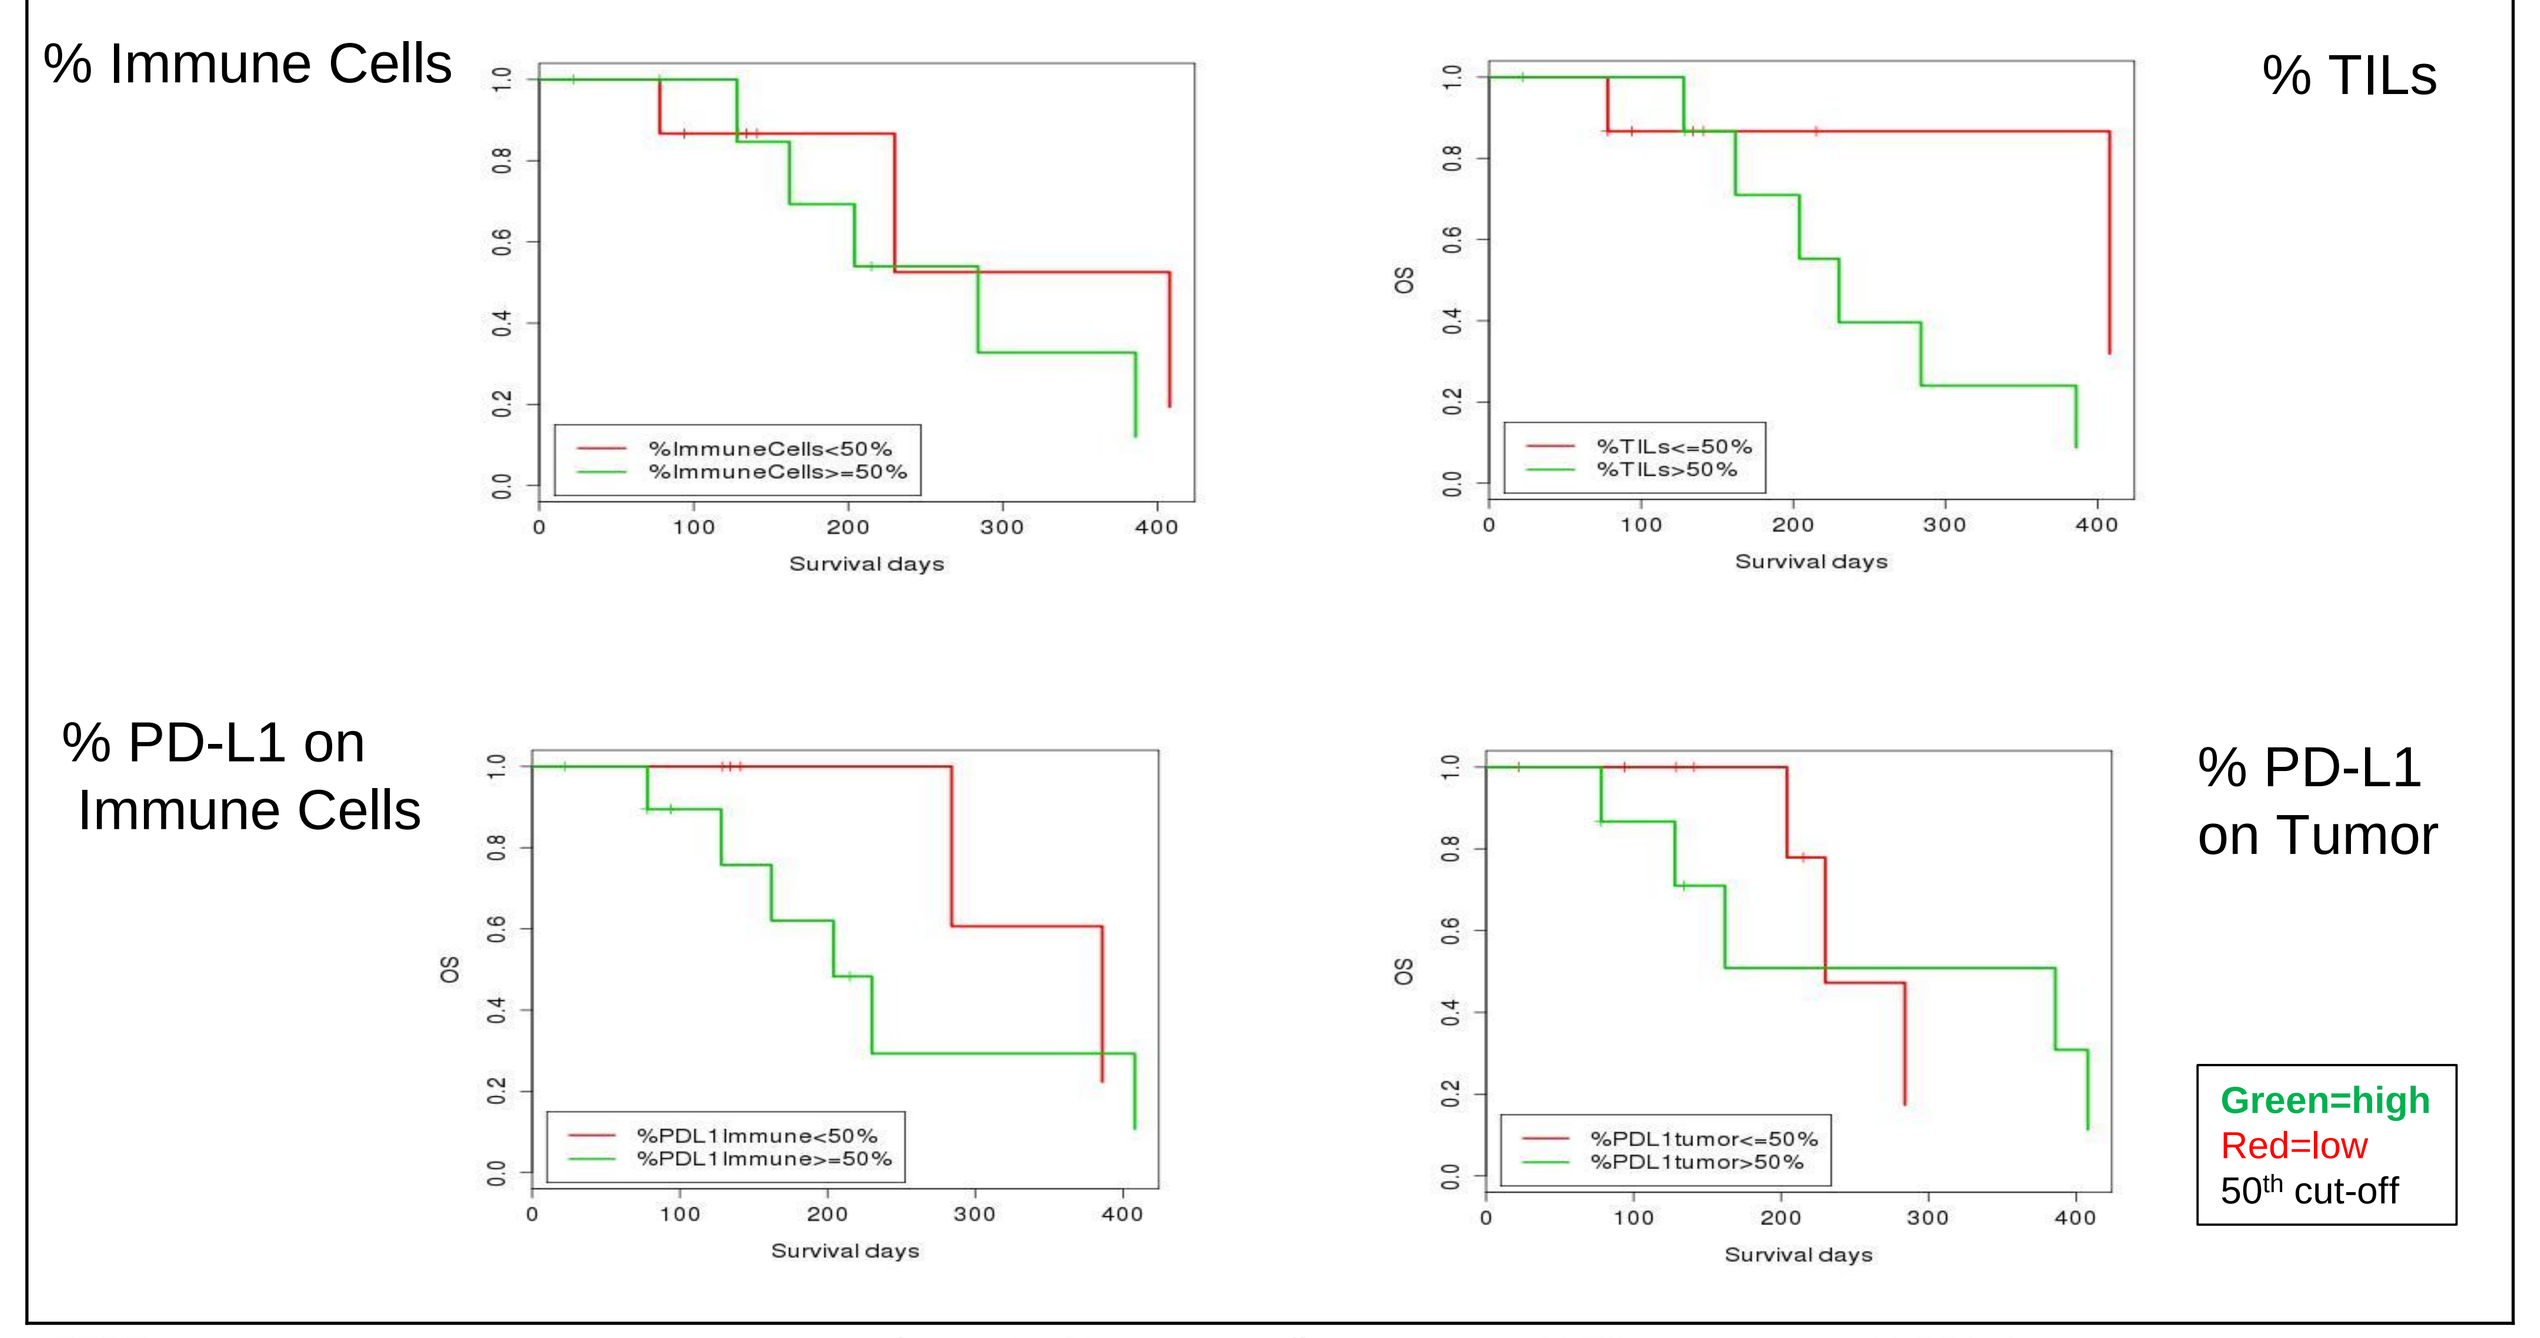
% Immune Cells
% TILs
% PD-L1 on Immune Cells
% PD-L1 on Tumor
%ImmuneCells<50%
%ImmuneCells>=50%
%TILs<=50%
%TILs>50%
%PDL1Immune<50%
%PDL1Immune>=50%
%PDL1tumor<=50%
%PDL1tumor>50%
OS
Survival days
Green=high
Red=low
50th cut-off

FFPE specimens from truncated patients were evaluated for % immune cells/tumor area, % TILs/tumor area, % PDL-1 positivity (membrane plus cytoplasm) on infiltrating immune cells and % PDL-1 positivity on tumor cells.

Summary

- This is an ongoing Phase 1b dose escalation study of demcizumab, a cancer stem cell targeting monoclonal antibody (targeting the DLL4 ligand in the Notch pathway) plus gemcitabine with or without nab-paclitaxel in 1st line pancreatic cancer patients.
- Demcizumab and gemcitabine with or without nab-paclitaxel were generally well tolerated with fatigue, nausea and vomiting being the most common drug related toxicities. The hypertension was managed with anti-hypertensives. Grade 2-3 pulmonary hypertension occurred in 2 patients and Grade 2 heart failure occurred in one patient receiving demcizumab for greater than 100 days, but none of the patients treated with truncated demcizumab developed pulmonary hypertension or heart failure.
- Patients are being followed with cardiac monitoring using B-type natriuretic peptide (BNP) and echocardiography. BNP appears to be an early indicator of cardiotoxicity. In addition, a cardioprotective medication (i,e, an angiotensin-converting enzyme inhibitor or carvedilol) was administered to patients with rising BNPs and this strategy appears to prevent cardiotoxicity.
- Truncated demcizumab therapy (i.e. 70 days of therapy) appears to prevent the onset of late cardiopulmonary toxicity, as none of the 32 patients treated in this manner developed heart failure or pulmonary hypertension.
- Fourteen of the 28 (50%) evaluable patients who received DEM/GEM/nab-paclitaxel had a RECIST partial response and 11 had stable disease resulting in a clinical benefit rate of 89%. The Kaplan-Meier estimated median progression free survival was 9.0 months (4.4 – not reached) and the Kaplan Meier estimated overall survival was 10.1 months (6.5 – 16.2) for the patients who received DEM/GEM/nab-paclitaxel.
- A randomized Phase 2 trial (***YOSEMITE***) in 1st line pancreatic cancer is ongoing. The truncated dose of demcizumab for the Phase 2 study is 3.5 mg/kg once every 2 weeks.