Enhanced anti-tumor efficacy by sequential application of Wnt pathway antagonists in combination with taxanes

Wan-Ching Yen, Marcus Fischer, Belinda Cancilla, Fiore Cattaruzza, Tracy Tang, Pete Yeung, John Lewicki, Austin Gurney and Timothy Hoey

OncoMed Pharmaceuticals Inc., Redwood City, California, 94063

ABSTRACT

The Wnt/beta-catenin pathway, which signals through the Frizzled (FZD) receptor family and several co-receptors, has long been implicated in cancer. We have previously demonstrated that inhibition of Wnt/beta-catenin signaling by vantictumab (anti-Fzd7, OMP-18R5) or ipafricept (FZD8-Fc, OMP-54F28) inhibits tumor growth, decreases tumorigenicity and induces differentiation in solid tumors. The anti-tumor effect of our Wnt antagonists is most evident in combination with chemotherapeutic agents. We sought to determine if the anti-tumor effect of Wnt pathway inhibitors varied with different chemotherapeutic agents. We compared the growth inhibitory effect of vantictumab and ipafricept with either taxanes (paclitaxel and *nab*-paclitaxel) or with DNA synthesis inhibitors (gemcitabine and carboplatin) in patient-derived tumor xenografts. We observed enhanced anti-tumor activity when combining vantictumab or ipafricept with *nab*-paclitaxel or paclitaxel compared to the combination with gemcitabine or carboplatin in pancreatic ductal carcinoma and serous ovarian cancer xenograft models. Histologic analysis in a pancreatic ductal carcinoma indicated that *nab*-paclitaxel increased mitotic cells and beta-catenin levels. Importantly, the addition of vantictumab to *nab*-paclitaxel reversed the *nab*-paclitaxel-induced increase in mitotic cells and beta-catenin expression. A potential mechanism to account for these results involves the observation that Wnt/beta-catenin signaling is under cell cycle control and peaks at the G2/M phase. Taxanes inhibit microtubule function and block the cell cycle at G2/M. In contrast, other chemotherapeutic agents, such as platinum compounds and nucleoside analogs, inhibit DNA synthesis and block cell proliferation at S phase. Our findings suggest that combination of Wnt blockade with chemotherapeutic agents, such as taxanes, that induce G2/M arrest may resulted in enhanced anti-tumor activity. The optimal synergy of anti-Wnt plus taxane combination occurs when the antibody was applied prior to taxane. Further analyses in serous ovarian tumors reveal that pre-treatment with ipafricept resulted in dysregulated beta-catenin localization within giant multi-nucleated cells and up-regulation of genes associated with negative regulators of G1 progression. Our work provides evidence for the enhanced anti-tumor effect of Wnt pathway inhibitors in combination with taxanes and highlights the importance of preclinical examination to identify the most efficacious combination therapy regimens and the timing of antibody action for Wnt antagonists in combination with taxanes for optimal treatment efficacy.

BACKGROUND

- The Wnt/beta-catenin signaling pathway, which signals through the frizzled (FZD) receptor family and several co-receptors, plays an important role in controlling cell fate, self-renewal and maintenance of cancer stem cells. Dysregulation of this pathway has been implicated in cancer.
- We have developed two novel Wnt pathway antagonist antibodies:
 - ➢ Vantictumab (OMP-18R5), which blocks Wnt binding and canonical signalling induced by multiple Wnt family members (Gurney et al. PNAS 109:11717-22, 2012)
 - ➢ Ipafricept (OMP-54F28), consisting of a combination of a portion of the FZD8 receptor containing the extracellular ligand binding domain and a human IgG1 Fc fragment. This fusion protein blocks Wnt signaling induced by multiple Wnt family members by binding and sequestering Wnt.
- Preclinical studies show that vantictumab and ipafricept inhibit tumor growth, decrease tumorigenicity and induce differentiation in multiple patient-derived xenograft models. The anti-tumor effect of these antibodies is more evident in combination with chemotherapeutic agents.
- Recent work has demonstrated that Wnt/beta-catenin signaling is under cell cycle control and peak at G2/M phase (Trends in Cell Biol 20:453-60, 2010).
- Taxanes, which block cell cycle at G2/M and other DNA synthesis inhibitors are commonly used as standard-of-care chemotherapeutic agents in cancer treatment.
- *nab*-paclitaxel (Abraxane®), an albumin-stabilized paclitaxel, is effective in combination with gemcitabine for the treatment of pancreatic ductal adenocarcinoma (Von Hoff DD, et al. N Engl J Med 369:1691-1703, 2013) and has been approved by FDA as standard-of-care agent for pancreatic cancer treatment.
- We found that anti-WNT antibodies in combination with gemcitabine/*nab*-paclitaxel produced much greater anti-tumor effect than with gemcitabine in patient-derived pancreatic tumor models.
- The goal of this study is to examine whether different chemotherapeutic agents influence the anti-tumor effect of WNT pathway inhibitors in patient-derived xenograft tumor models.

MATERIALS and METHODS

- Patient-derived xenograft tumors were obtained from University, Molecular Response or NDRI. Both tumors were generated at OncoMed Pharmaceuticals, Inc. Tumors were passaged subcutaneously in NOD/SCID mice up to 4 passages.
- For efficacy studies, treatments were initiated when tumors reached 100 mm^3. Both antibodies and chemotherapeutic agents were administered intraperitonially.
- Histologic analysis used formalin-fixed, paraffin-embedded section or frozen sections. Slides were scanned using Imagescope (ScanScope AT, Aperio).
- For gene analysis, RNA was isolated from tumor tissues followed by c-DNA synthesis. The resulting c-DNA was analyzed by real-time PCR.
- Data are expressed as mean±S.E.M. Differences of $p<0.05$ are considered significantly different.

Comparison of Anti-WNT Antibodies in Combination with Gemcitabine vs. Gemcitabine/*nab*-Paclitaxel

OMP-PN25 Xenograft Tumors

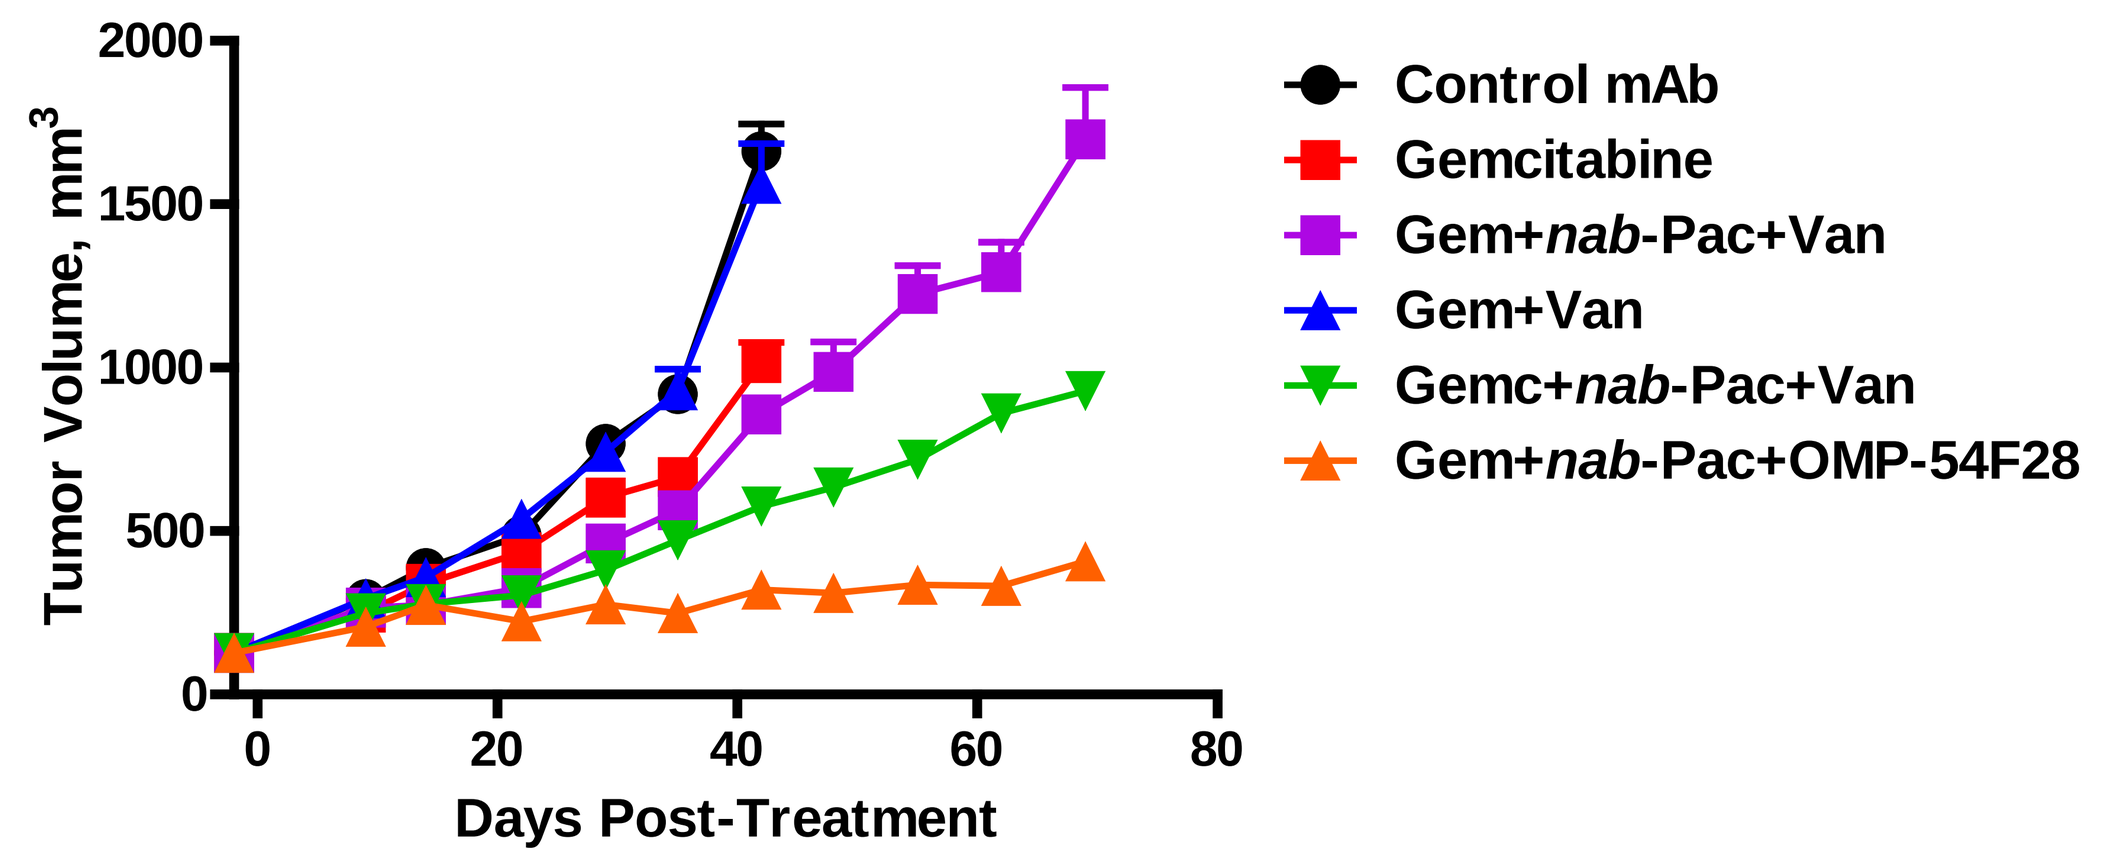
Control mAb
Gemcitabine
Gem+nab-Pac+Van
Gem+Van
Gemc+nab-Pac+Van
Gem+nab-Pac+OMP-54F28
Tumor Volume, mm3
Days Post-Treatment

Differential Anti-Tumor Effect of WNT Pathway Inhibitors in Combination with Taxanes

OMP-OV19 Ovarian Xenograft Tumors

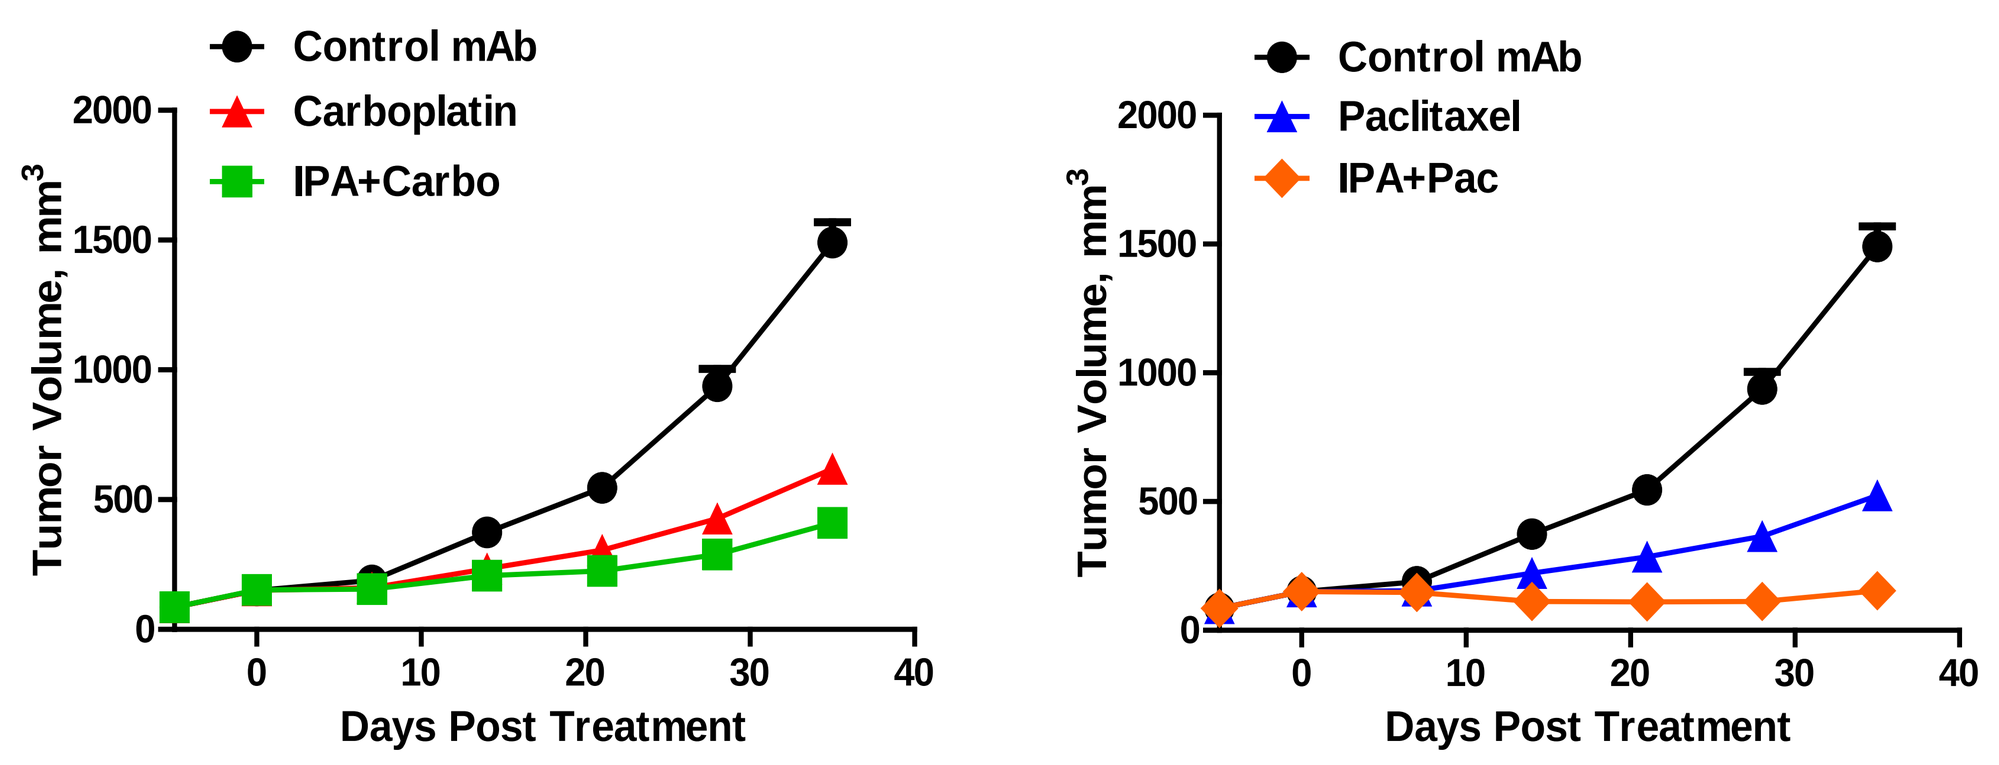
Control mAb
Carboplatin
IPA+Carbo
Control mAb
Paclitaxel
IPA+Pac
Tumor Volume, mm3
Days Post Treatment

OMP-PN13 Pancreatic Xenograft Tumors

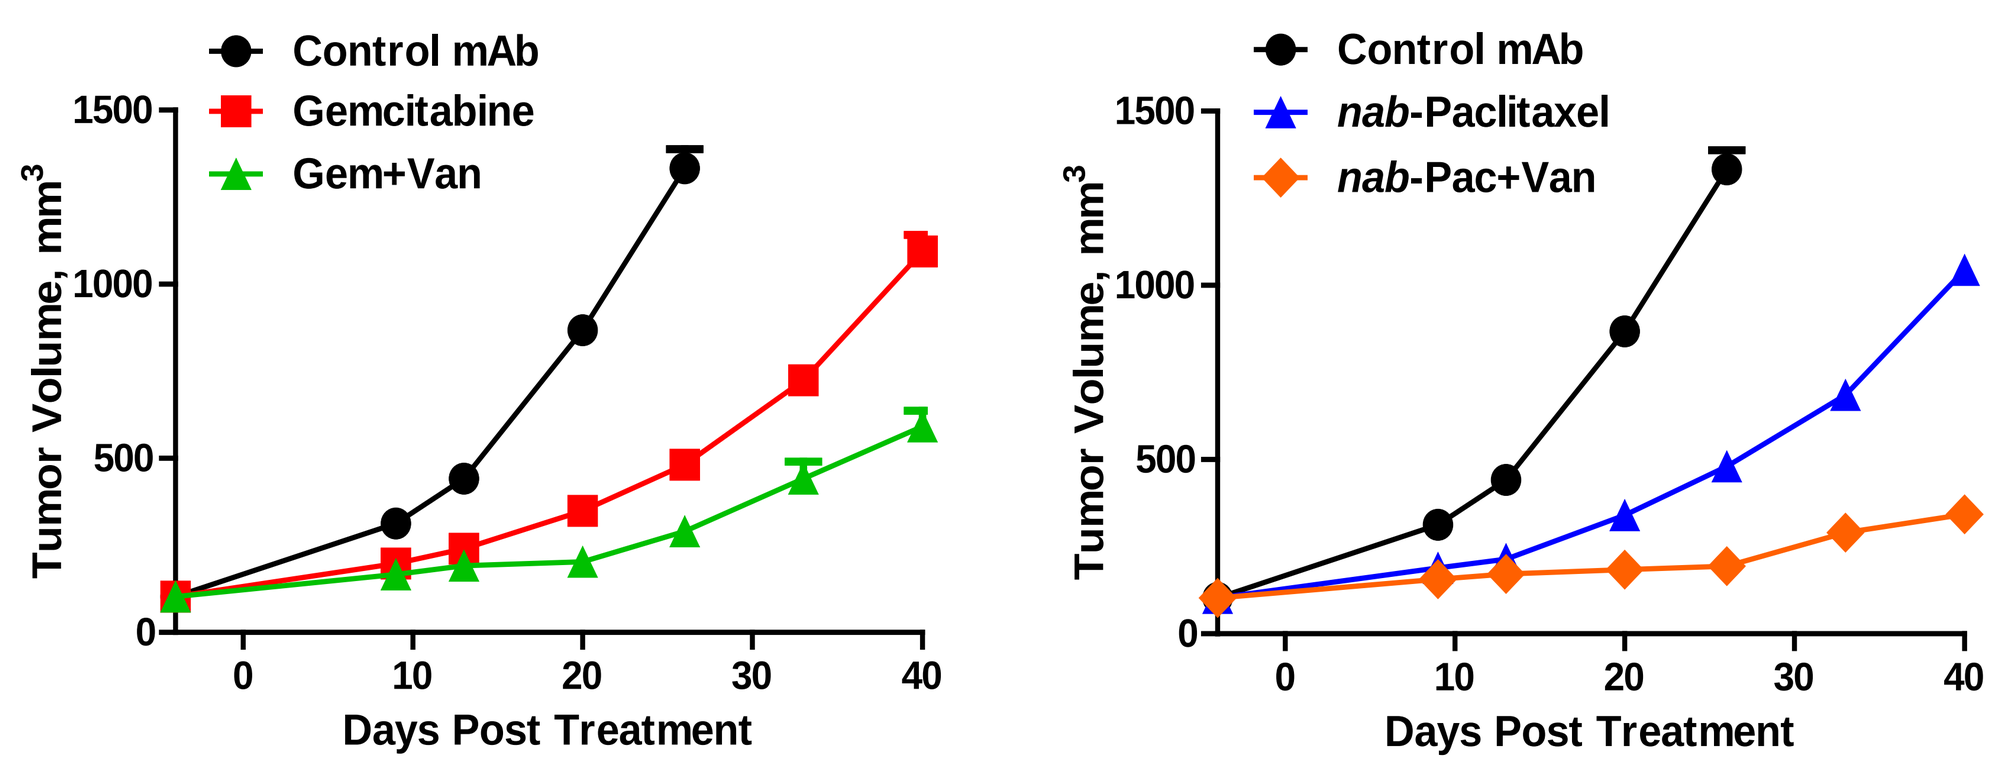
Control mAb
Gemcitabine
Gem+Van
Control mAb
nab-Paclitaxel
nab-Pac+Van
Tumor Volume, mm3
Days Post Treatment

Effect of Vantictumab Plus Gemcitabine or *nab*-Paclitaxel On Phospho-Histone H3 and β-Catenin Expression

Phospho-Histone H3

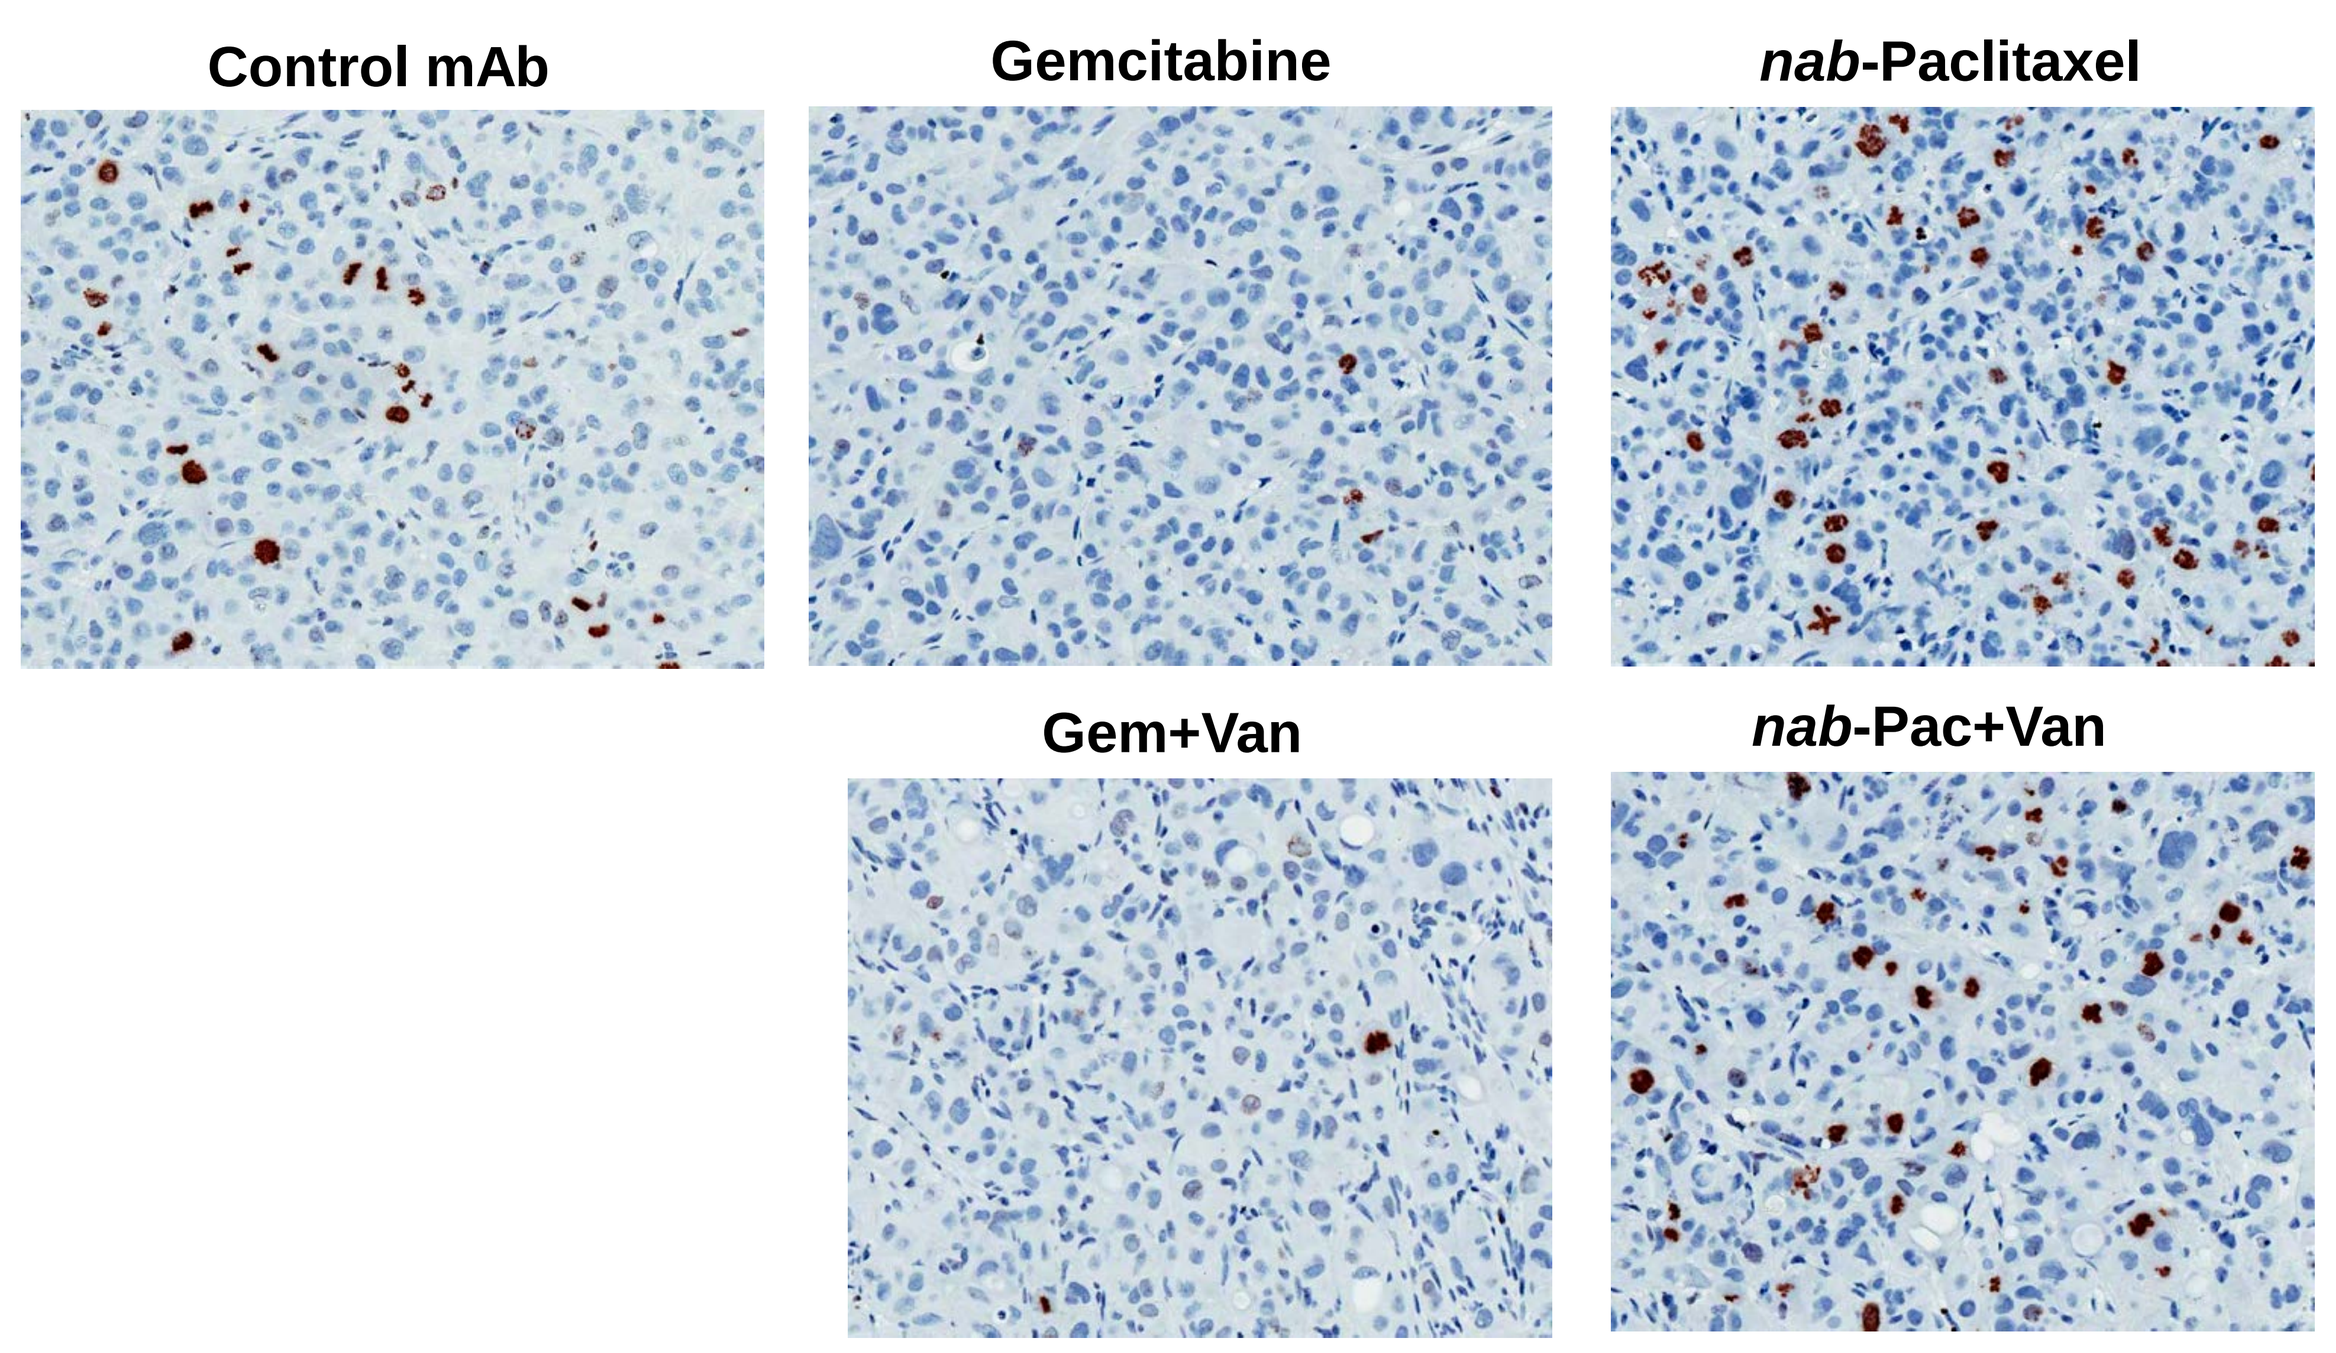
Control mAb
Gemcitabine
nab-Paclitaxel
Gem+Van
nab-Pac+Van

Total β-Catenin

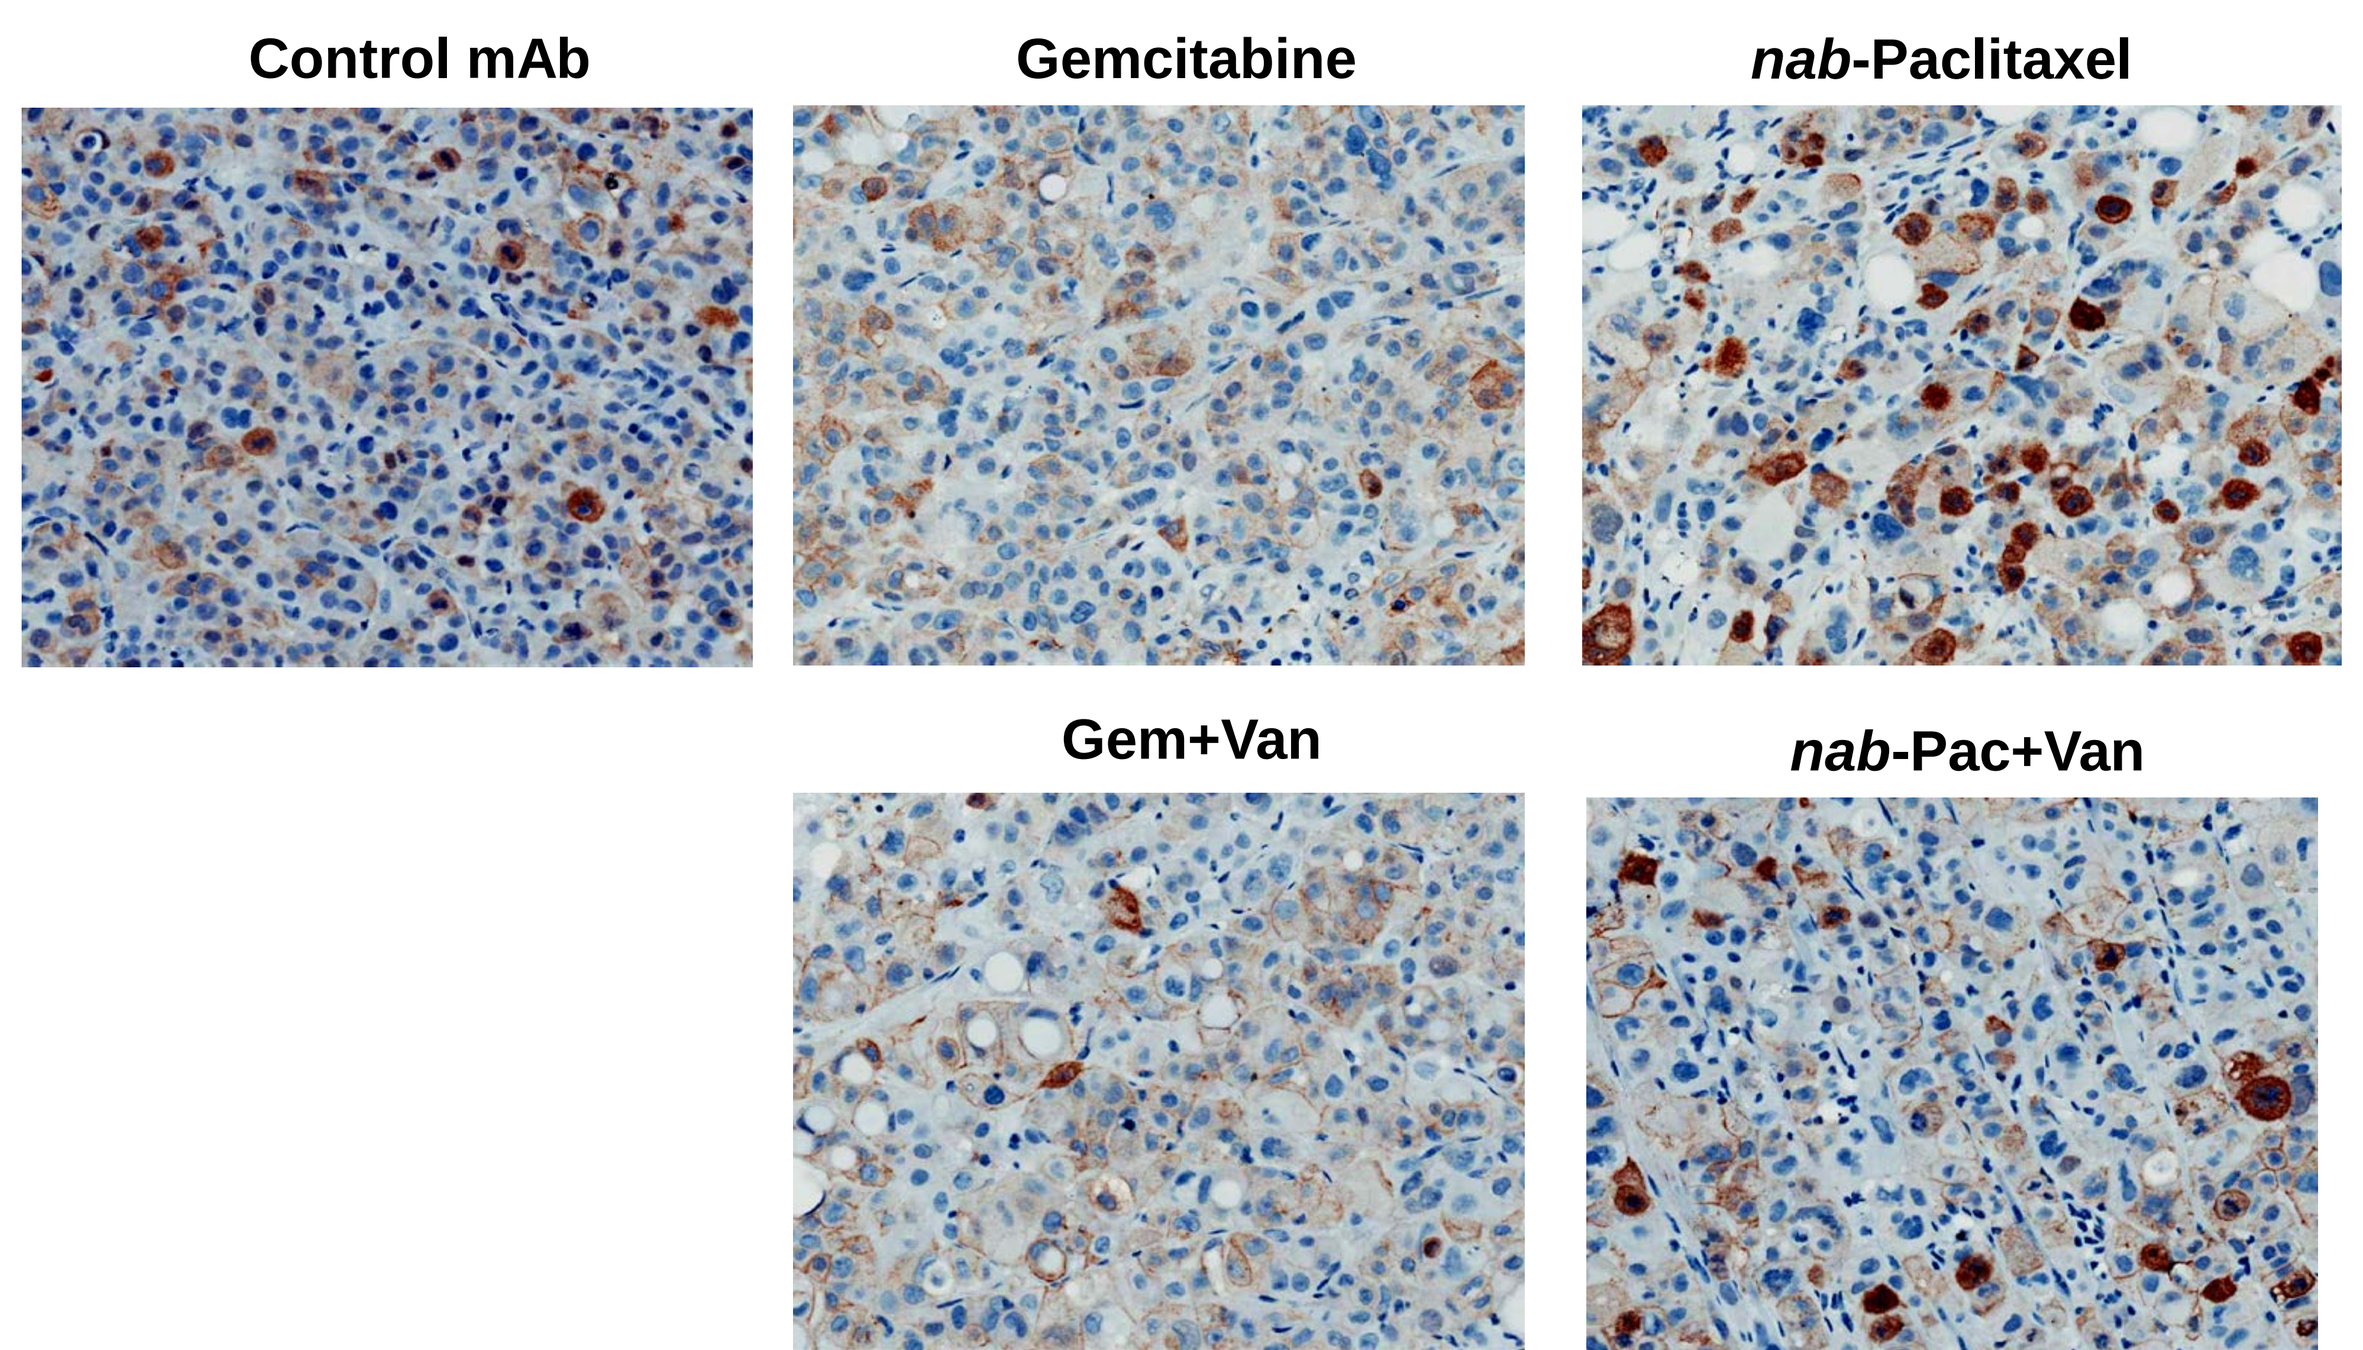
Control mAb
Gemcitabine
nab-Paclitaxel
Gem+Van
nab-Pac+Van

Intermittent Sequential Application of Vantictumab with Paclitaxel Reduces Cancer Initiating Cell Frequency

OMP-LU77 NSCLC Xenograft Tumors

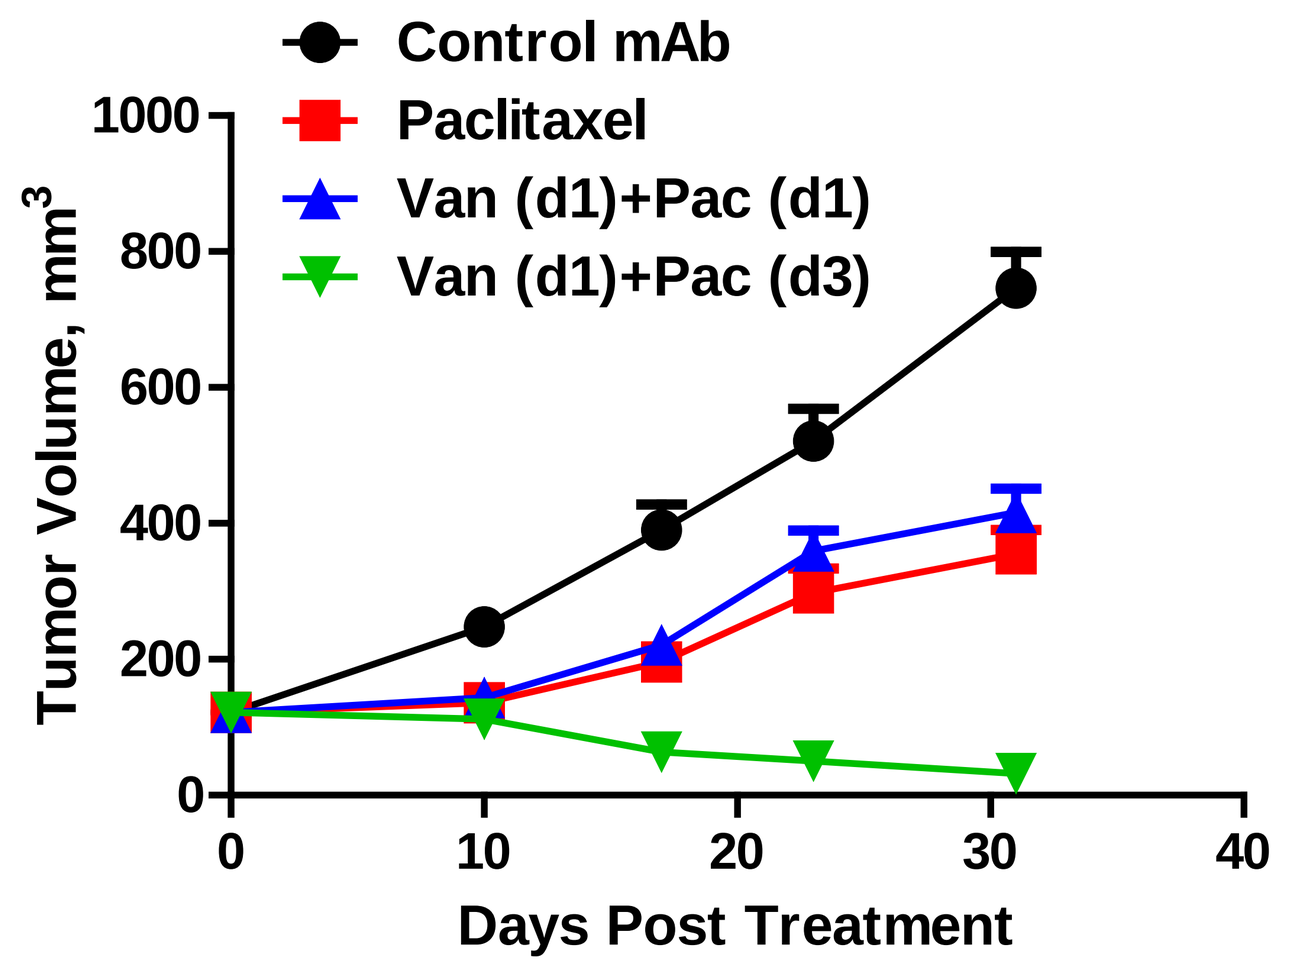
Control mAb
Paclitaxel
Van (d1)+Pac (d1)
Van (d1)+Pac (d3)
Tumor Volume, mm3
Days Post Treatment

Paclitaxel: 20 mg/kg Q3W
Vantictumab: 25 mg/kg Q3W

1. Isolate tumors post-treatment
2. Transplant 100, 1000 cells (n=10)
3. Grow 78 days without treatment
4. Calculate CSC Frequency based on tumor take rate

In Vivo Limiting Dilution Assay
79 Days Post Cell Injection

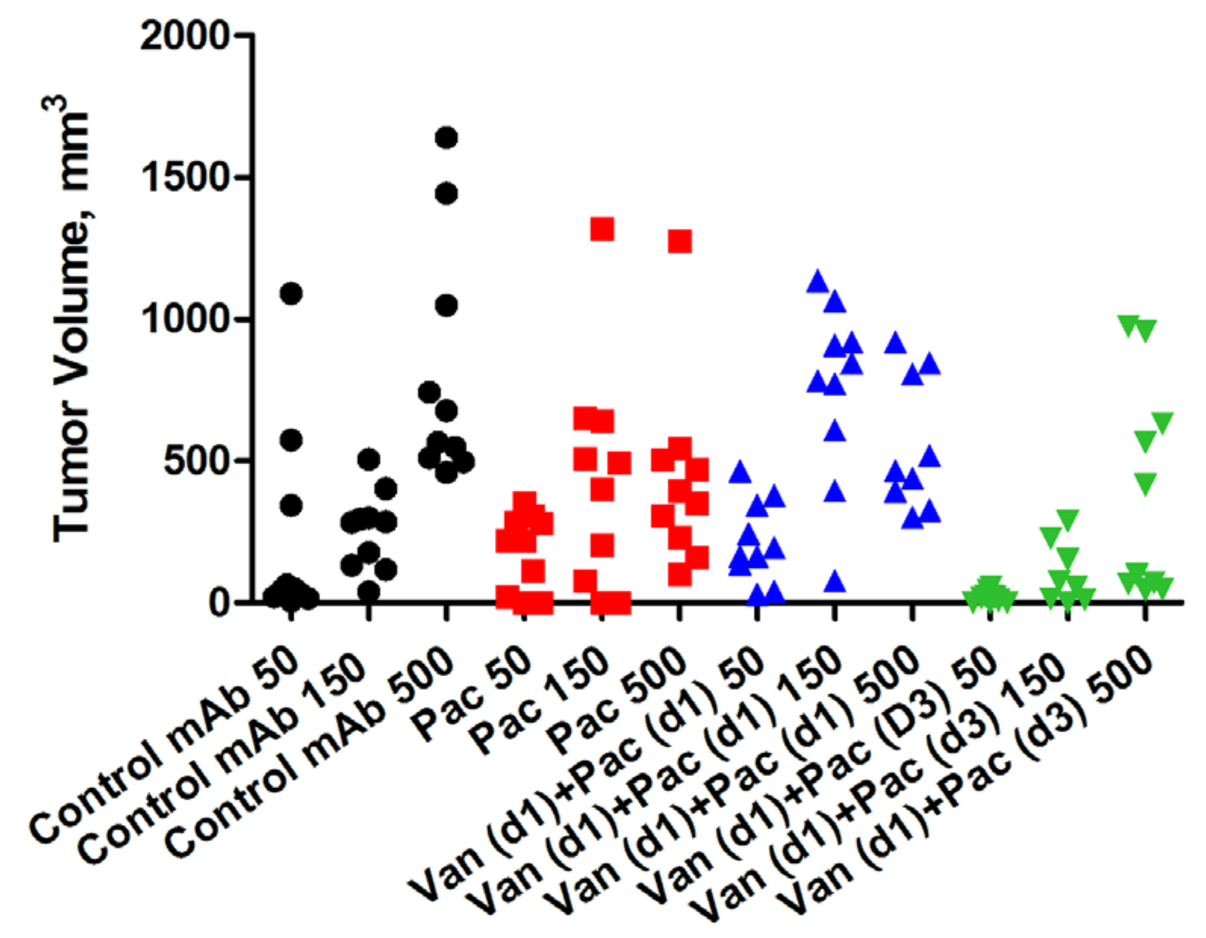
Tumor Volume, mm3
Control mAb 50
Control mAb 150
Control mAb 500
Pac 50
Pac 150
Pac 500
Van (d1)+Pac (d1) 50
Van (d1)+Pac (d1) 150
Van (d1)+Pac (d1) 500
Van (d1)+Pac (D3) 50
Van (d1)+Pac (d3) 150
Van (d1)+Pac (d3) 500

In Vivo CSC Frequency
Day 79 post-transplantation

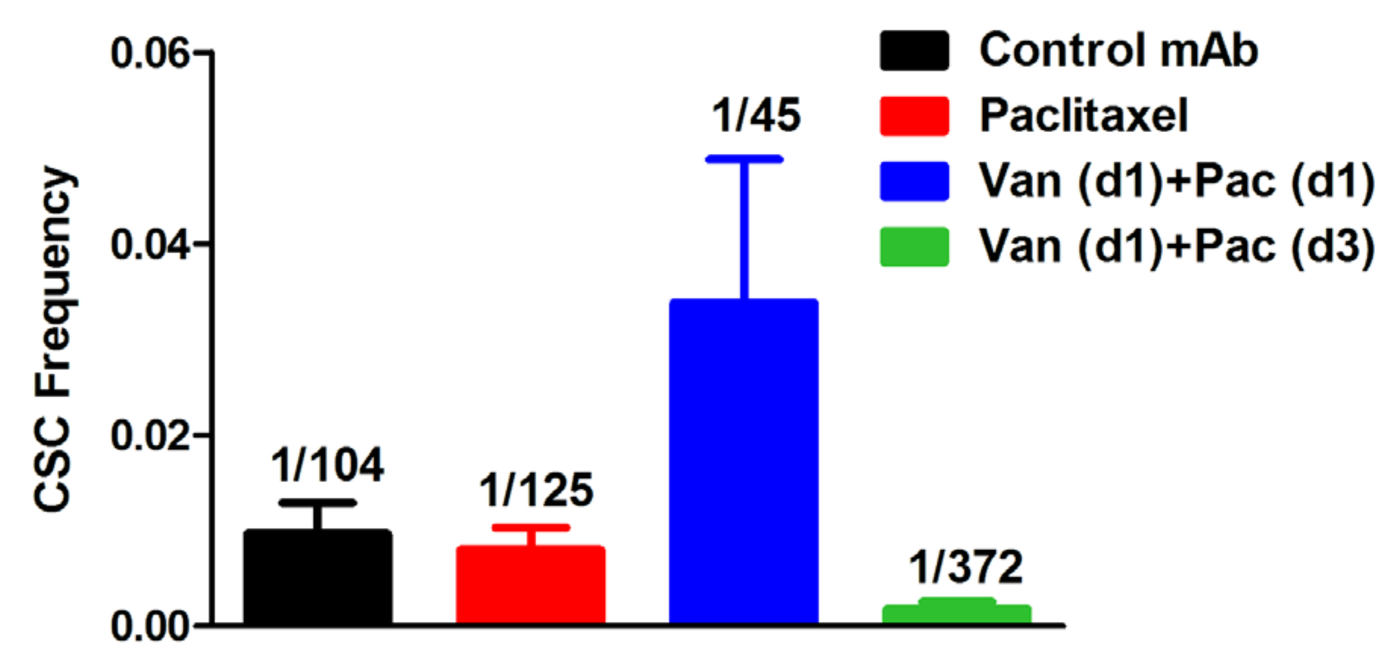
Control mAb
Paclitaxel
Van (d1)+Pac (d1)
Van (d1)+Pac (d3)
CSC Frequency
1/104
1/125
1/45
1/372

Intermittent Sequential Application of Ipafricept with Paclitaxel Induces Tumor Regression

OMP-OV19 Ovarian Xenograft Tumors

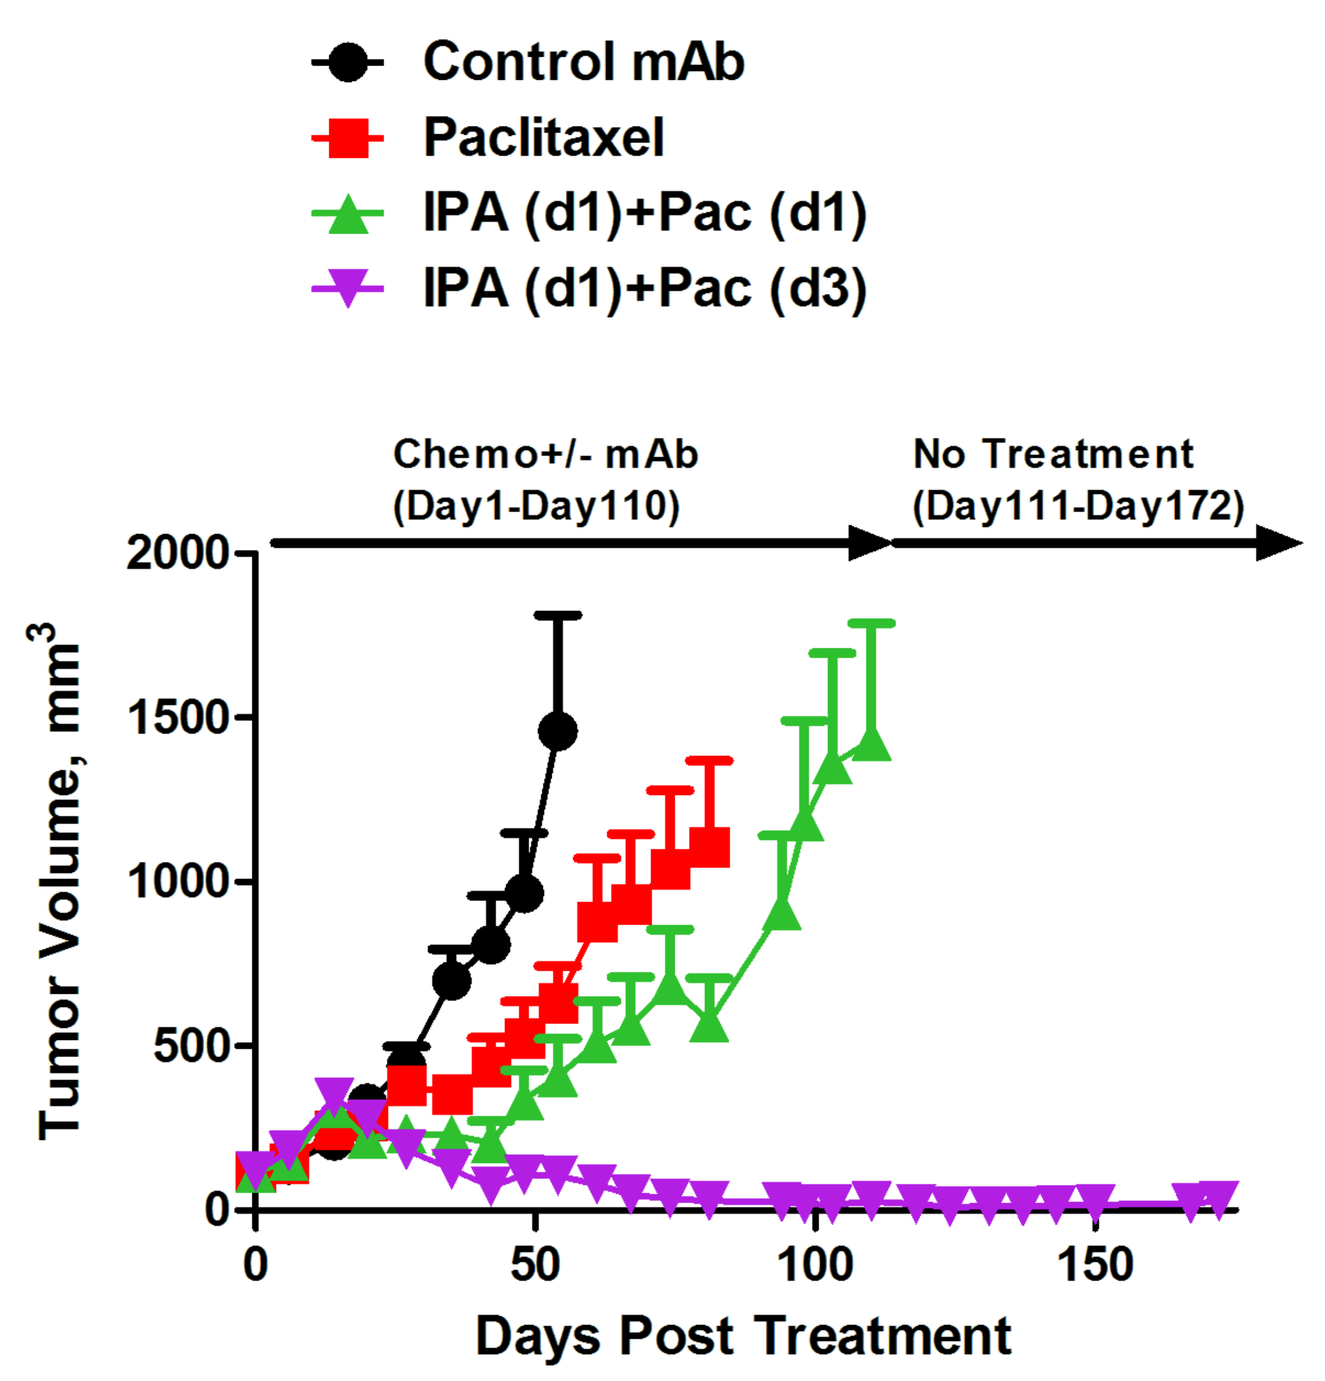
Control mAb
Paclitaxel
IPA (d1)+Pac (d1)
IPA (d1)+Pac (d3)
Chemo+/- mAb
(Day1-Day110)
No Treatment
(Day111-Day172)
Tumor Volume, mm3
Days Post Treatment

Paclitaxel: 20 mg/kg Q2W
Ipafricept: 20 mg/kg Q2W

IPA (d1)+Pac (d3)
90 Days Post Treatment Termination

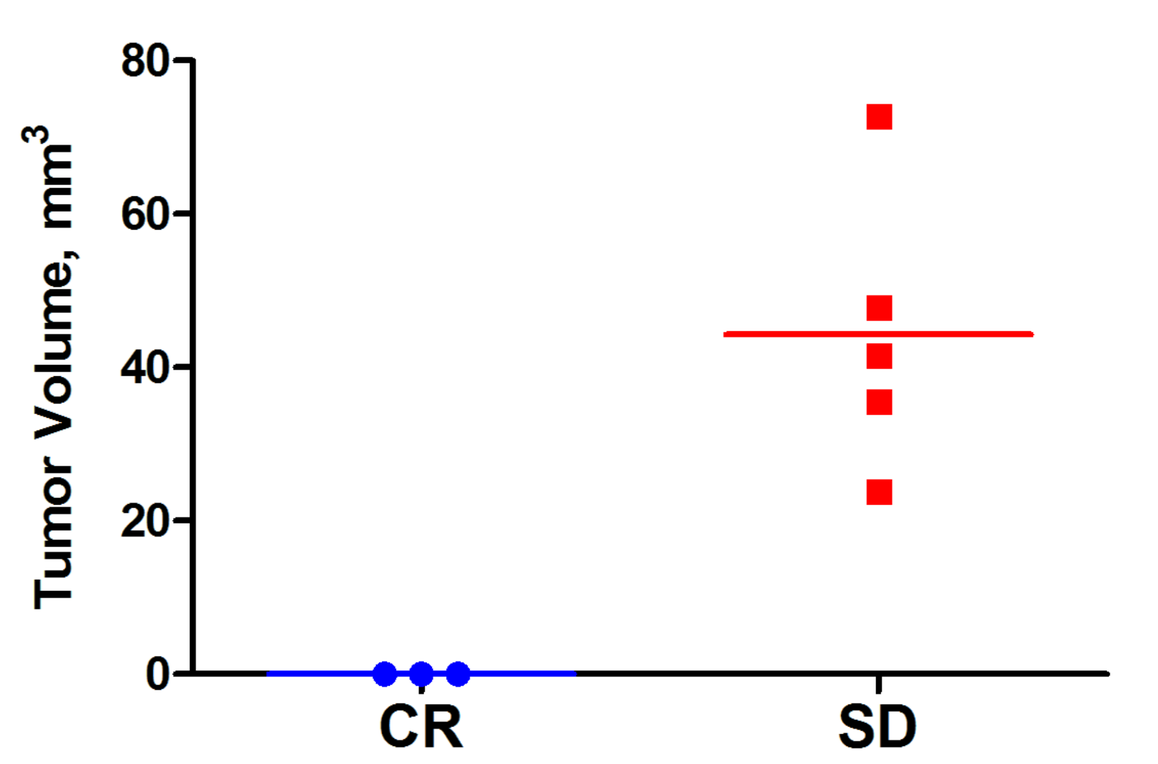
Tumor Volume, mm3
CR
SD

- Taxanes are more effective class of chemotherapeutic agents in combination with anti-WNT than non-taxane agents
- The enhanced combination effect is both taxane-specific and dose-dependent
- Anti-WNT plus taxane combination can be further improved by applying antibody prior to taxane
- Intermittent sequential application of anti-WNT plus taxane induces mitotic catastrophe and results in dysregulated beta-catenin localization within giant multi-nucleated cells
- The optimal synergy occurs when the antibody is applied 2-3 days prior to taxane

Intermittent Sequential Application of Ipafricept and Paclitaxel Enhance Anti-Tumor Activity, Induces Mitotic Catastrophe and Increases Expression of Genes Associated with Negative Regulators of G1/S Progression

OMP-OV38 Ovarian Xenograft Tumors

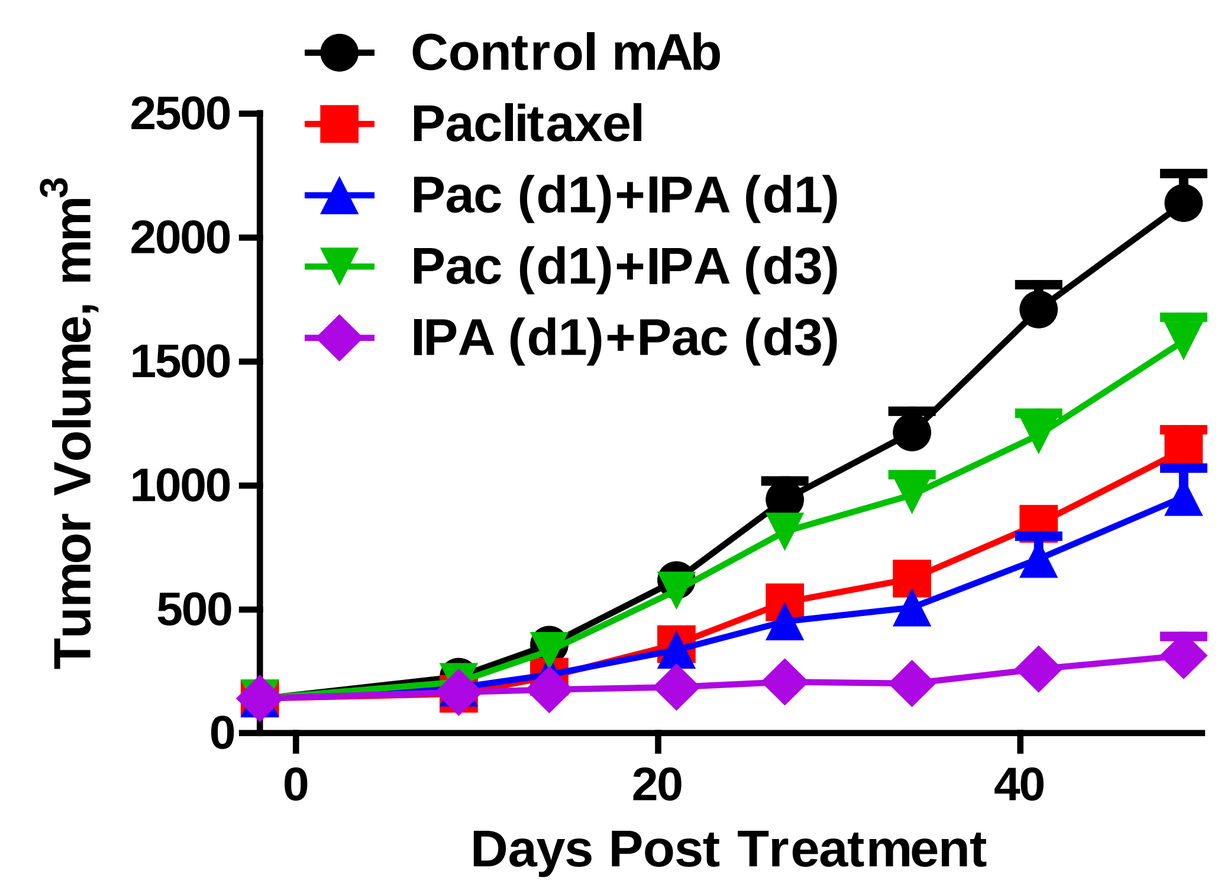
Control mAb
Paclitaxel
Pac (d1)+IPA (d1)
Pac (d1)+IPA (d3)
IPA (d1)+Pac (d3)
Tumor Volume, mm3
Days Post Treatment

Paclitaxell: 20 mg/kg Q2W
Ipafricept: 25 mg/kg Q2W

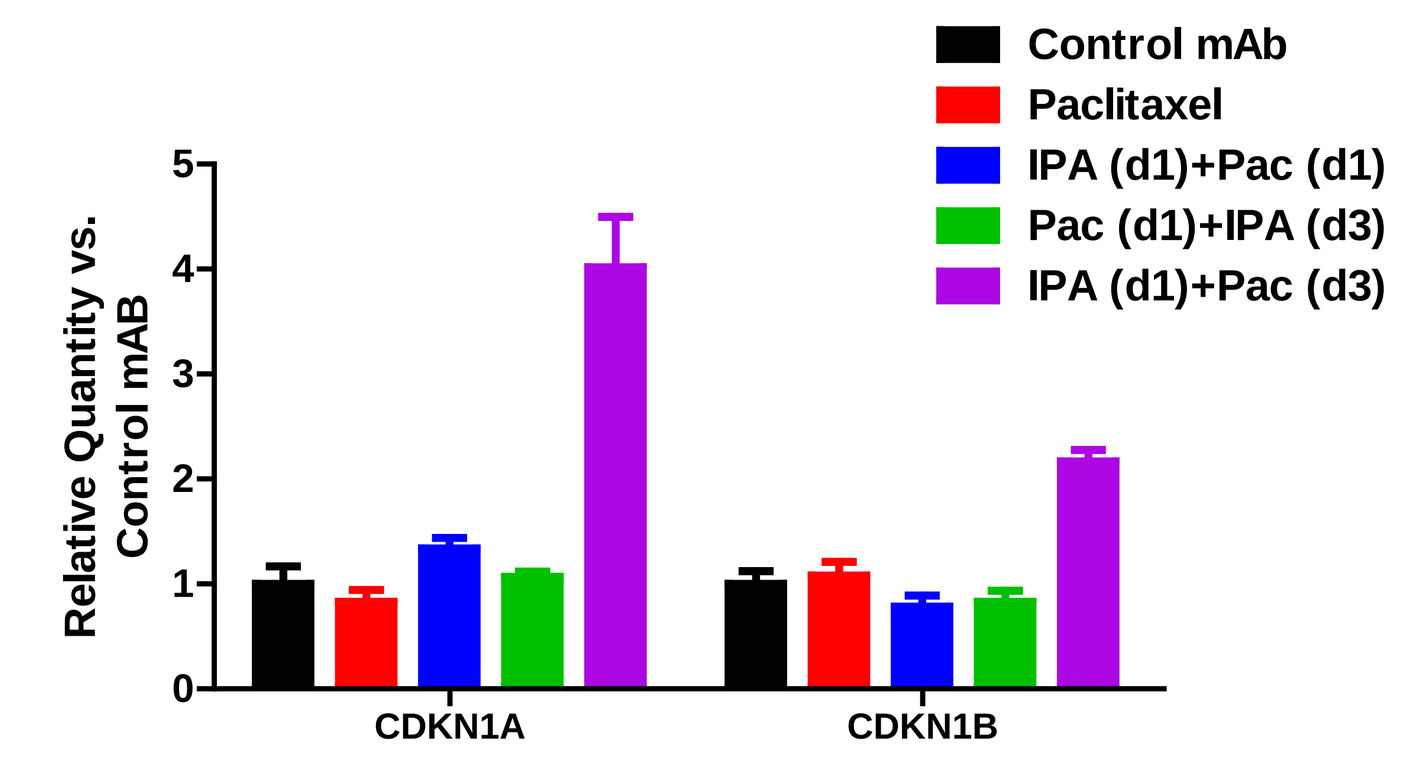
Control mAb
Paclitaxel
IPA (d1)+Pac (d1)
Pac (d1)+IPA (d3)
IPA (d1)+Pac (d3)
Relative Quantity vs. Control mAB
CDKN1A
CDKN1B

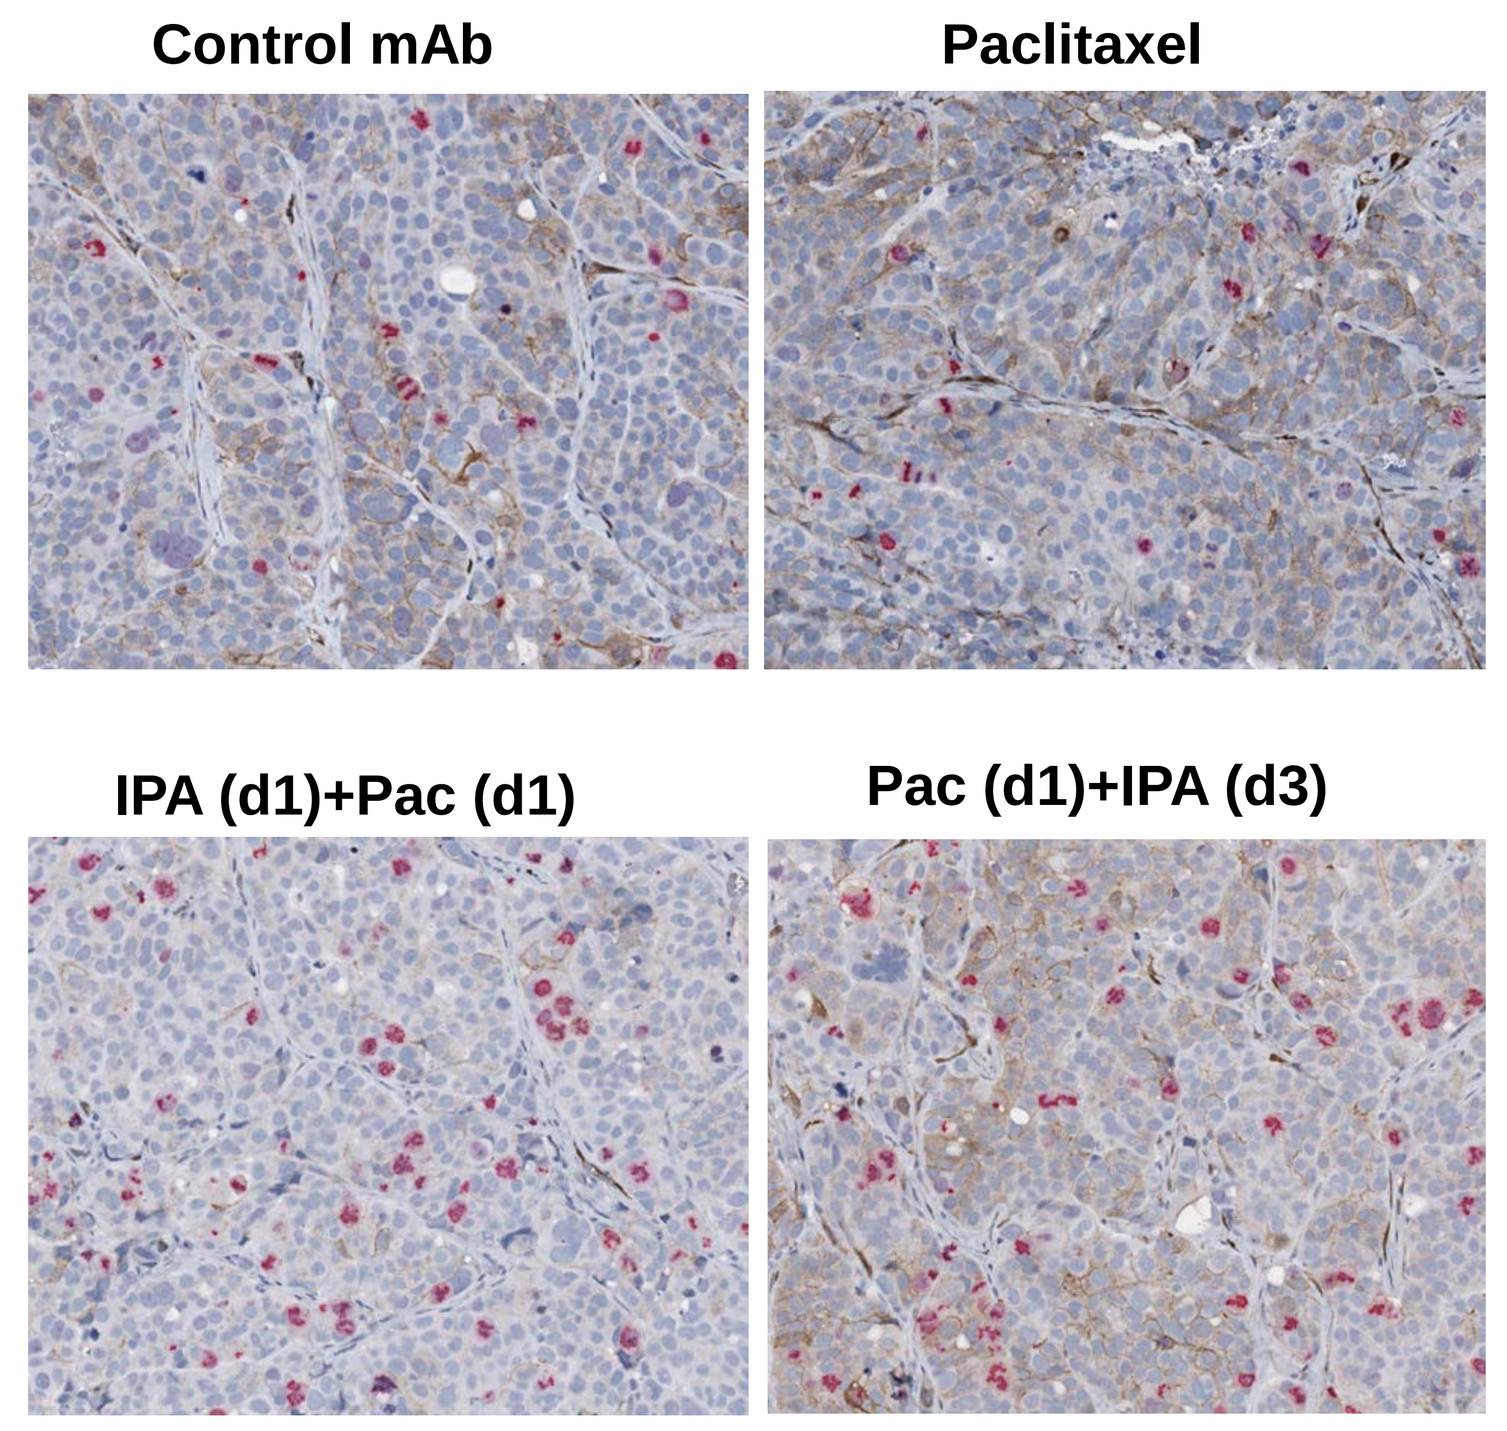
Control mAb
Paclitaxel
IPA (d1)+Pac (d1)
Pac (d1)+IPA (d3)

IPA (d1)+Pac (d3)

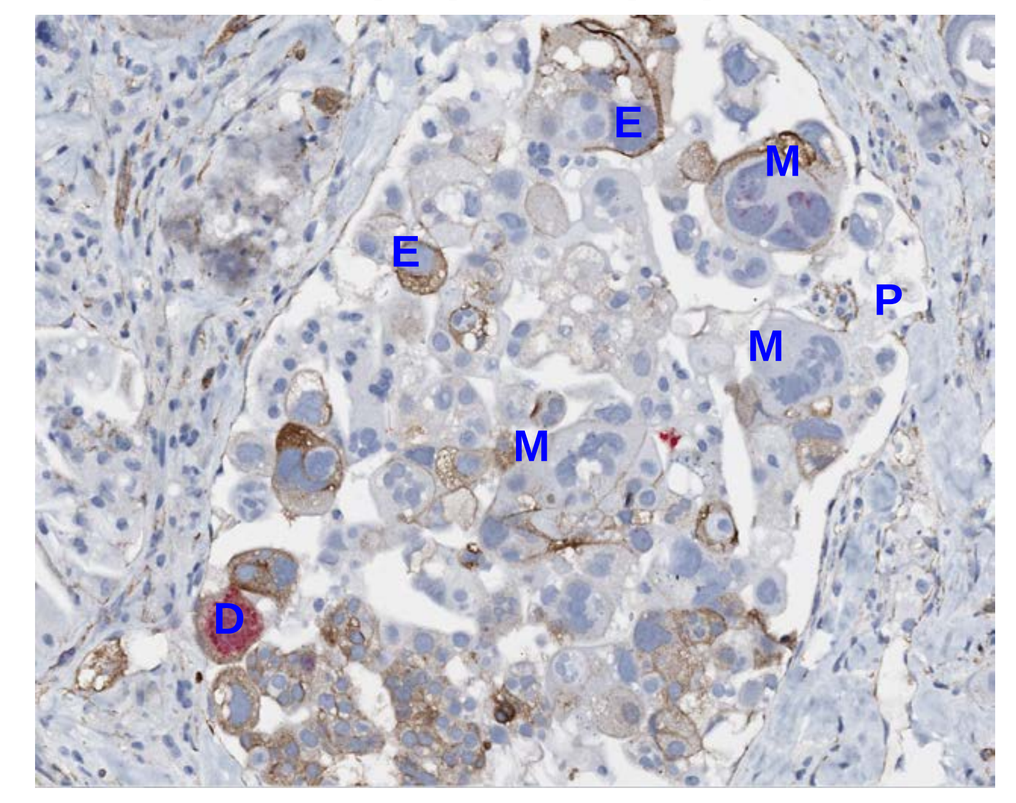
E
M
P
D

E: Enlarged cells with giant nuclei
M: Multinucleated cells
P: Pyknotic nuclei
D: Dividing cells

β-catenin: Brown
Phospho-Histone H3: Red

Optimal Synergy of Sequential Application of Anti-WNT Antagonists Plus Taxane Combination Occurs When the Antibody is Applied 2-3 Days Prior to Taxane

OMP-OV38 Ovarian Xenograft Tumors

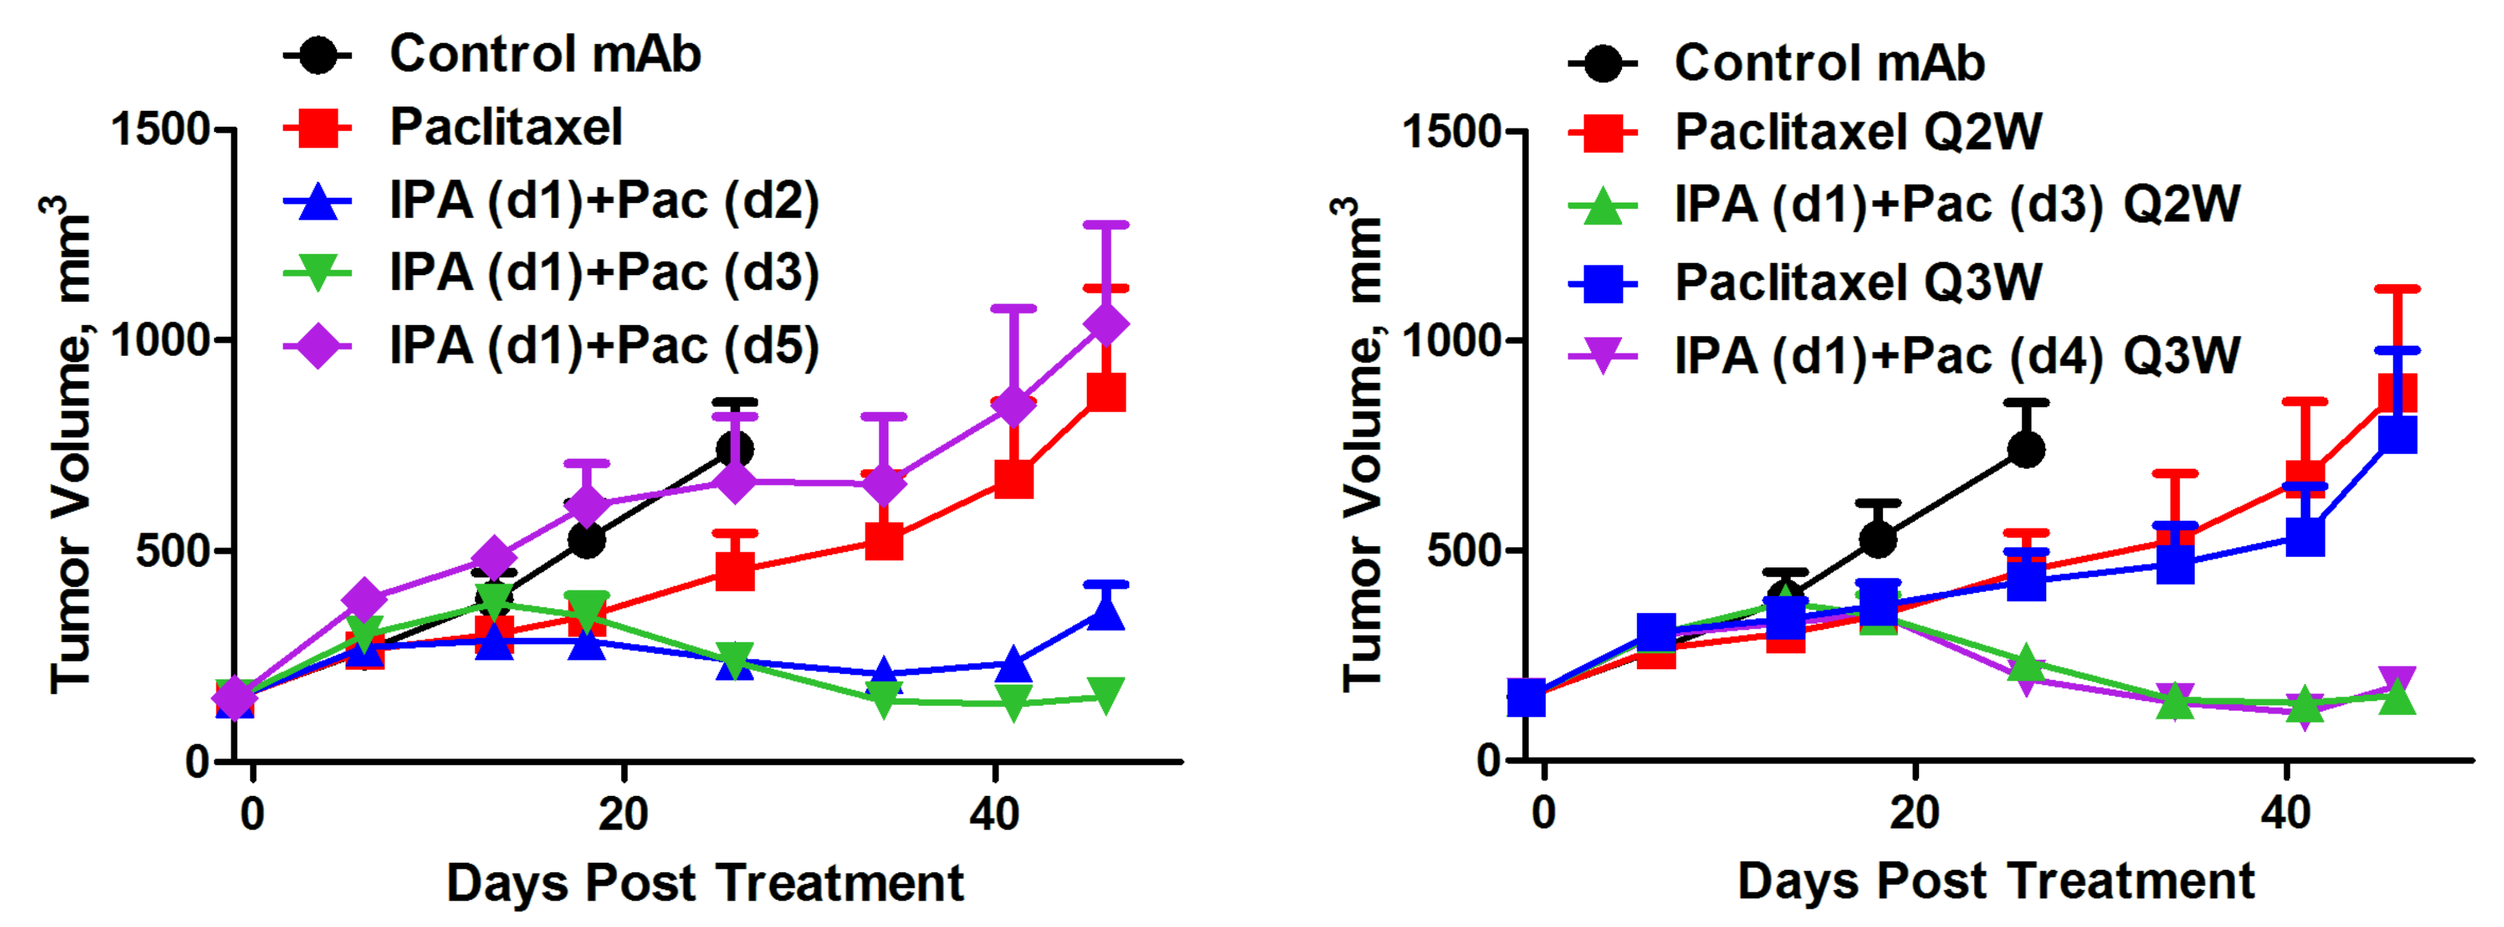
Control mAb
Paclitaxel
IPA (d1)+Pac (d2)
IPA (d1)+Pac (d3)
IPA (d1)+Pac (d5)
Control mAb
Paclitaxel Q2W
IPA (d1)+Pac (d3) Q2W
Paclitaxel Q3W
IPA (d1)+Pac (d4) Q3W
Tumor Volume, mm3
Days Post Treatment

Paclitaxel: 20 mg/kg Q2W
Ipafricept: 25 mg/kg Q2W

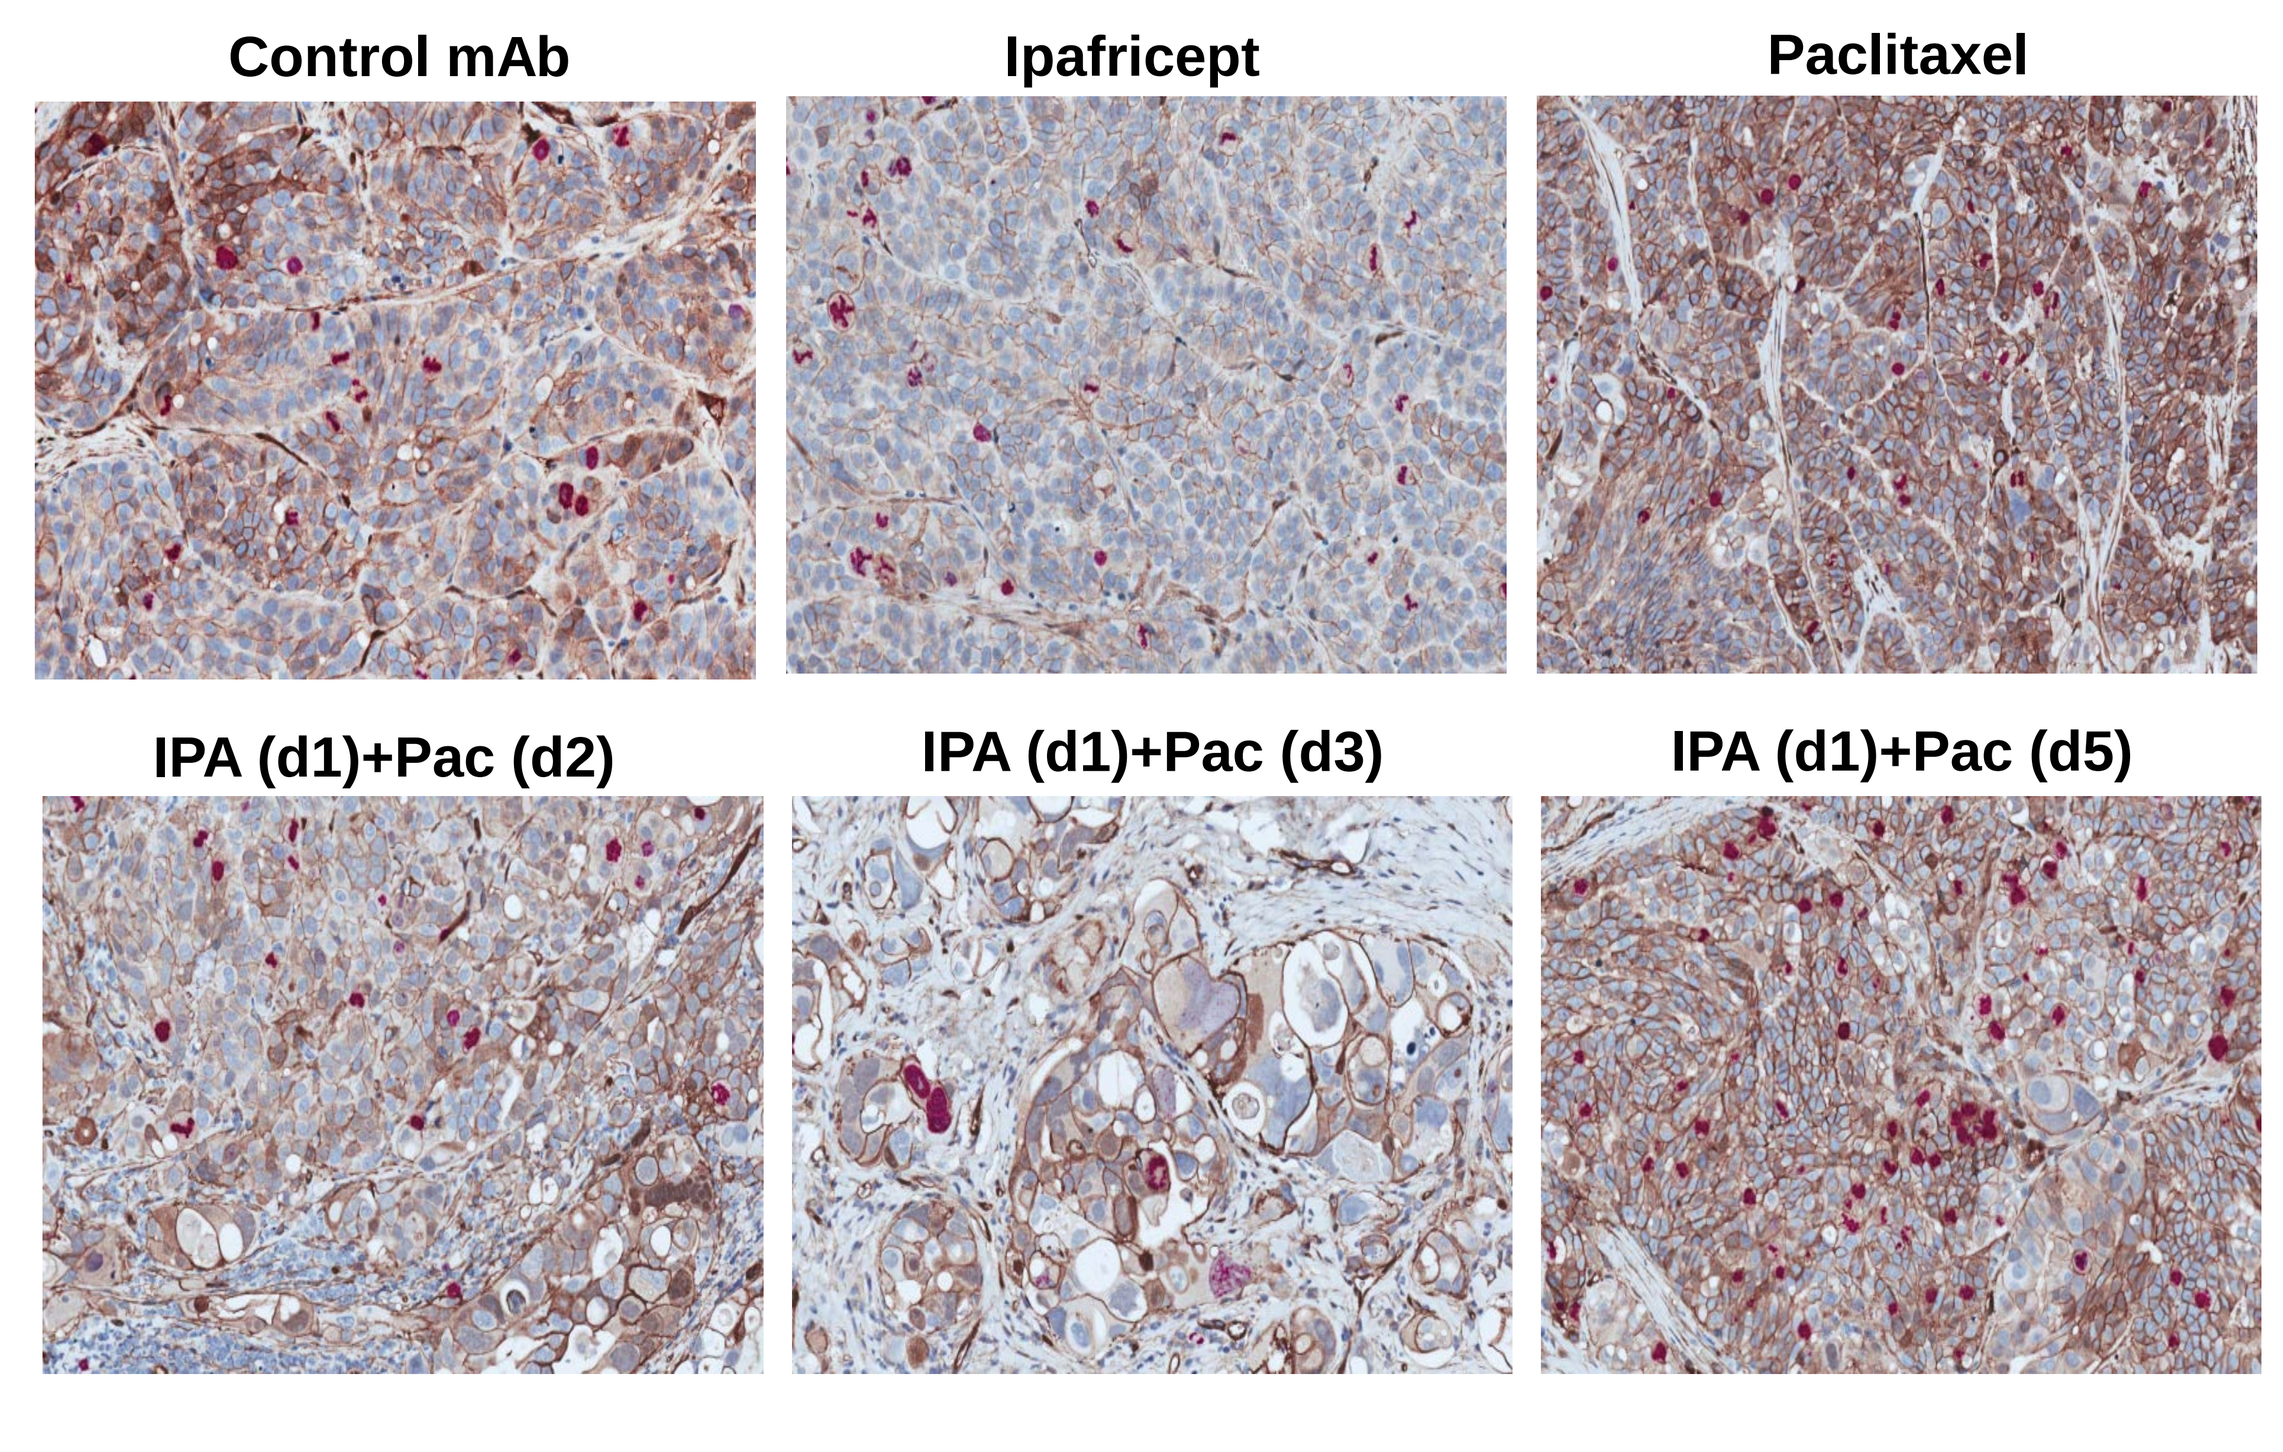
Control mAb
Ipafricept
Paclitaxel
IPA (d1)+Pac (d2)
IPA (d1)+Pac (d3)
IPA (d1)+Pac (d5)